compugen FROM CODE TO CURE™

Poster #422

PHASE 1 STUDY OF COM701 MONOTHERAPY AND IN COMBINATION WITH NIVOLUMAB IN PATIENTS WITH ADVANCED SOLID TUMORS.

E. Ileana Dumbrava[1], G. Fleming[2], E. Hamilton[3], R. Sullivan[4], A Patnaik[5], K. Papadopoulos[5], A. ElNaggar[6], J. Hunter[7], J. Olweny[7], A. Adewoye[7], B. Chmielowski[8], D. Shepard[9], M. Sharma[10], E. Lim[11], D. Vaena[6], D. Rasco[5].

[1]Investigational Cancer Therapeutics, The University of Texas MD Anderson Cancer Center, Houston, TX, [2]The University of Chicago Medicine, Chicago, IL, [3]Drug Development Unit, Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, [4]Medical Oncology, Massachusetts General Hospital, Harvard Medical School, Boston, MA, [5]Clinical Research, The START Center for Cancer Care, San Antonio, TX, [6]West Cancer Center and Research Institute, Memphis, TN, [7]Clinical Development., Compugen USA Inc., South San Francisco, CA, [8]Division of Hematology – Medical Oncology, University of California Los Angeles, Los Angeles, CA, [9]Hematology and Medical Oncology, Cleveland Clinic, Cleveland, OH, [10]Clinical Research, START Midwest, Grand Rapids, MI, [11]Medical Oncology, Columbia University Medical Center, New York, NY.

BACKGROUND

- COM701 is a novel first-in-class humanized IgG4 monoclonal antibody that binds with high affinity to poliovirus receptor related immunoglobulin domain containing (PVRIG) blocking its interaction with its ligand, PVRL2 [1]
- Nivolumab is an anti-PD-1 antibody approved in patients with several malignancies [2].
- PD-1 inhibitors play an important role in this axis by modulating DNAM activation [3]
- In preclinical experiments we have demonstrated that PVRIG inhibition alone and in combination with anti-PD-1 leads to activation of T cells in the tumor microenvironment generating an anti-tumor immune response and tumor growth inhibition [1]
- Although ICI revolutionized cancer treatment there is an urgent need to develop treatments for patients who are refractory or relapse after treatment with ICI.
- We hypothesize that COM701 will be safe and tolerable and demonstrate antitumor activity in pts with R/R solid tumors

PVRIG IS A Novel Checkpoint in the TIGIT/DNAM-1 AXIS

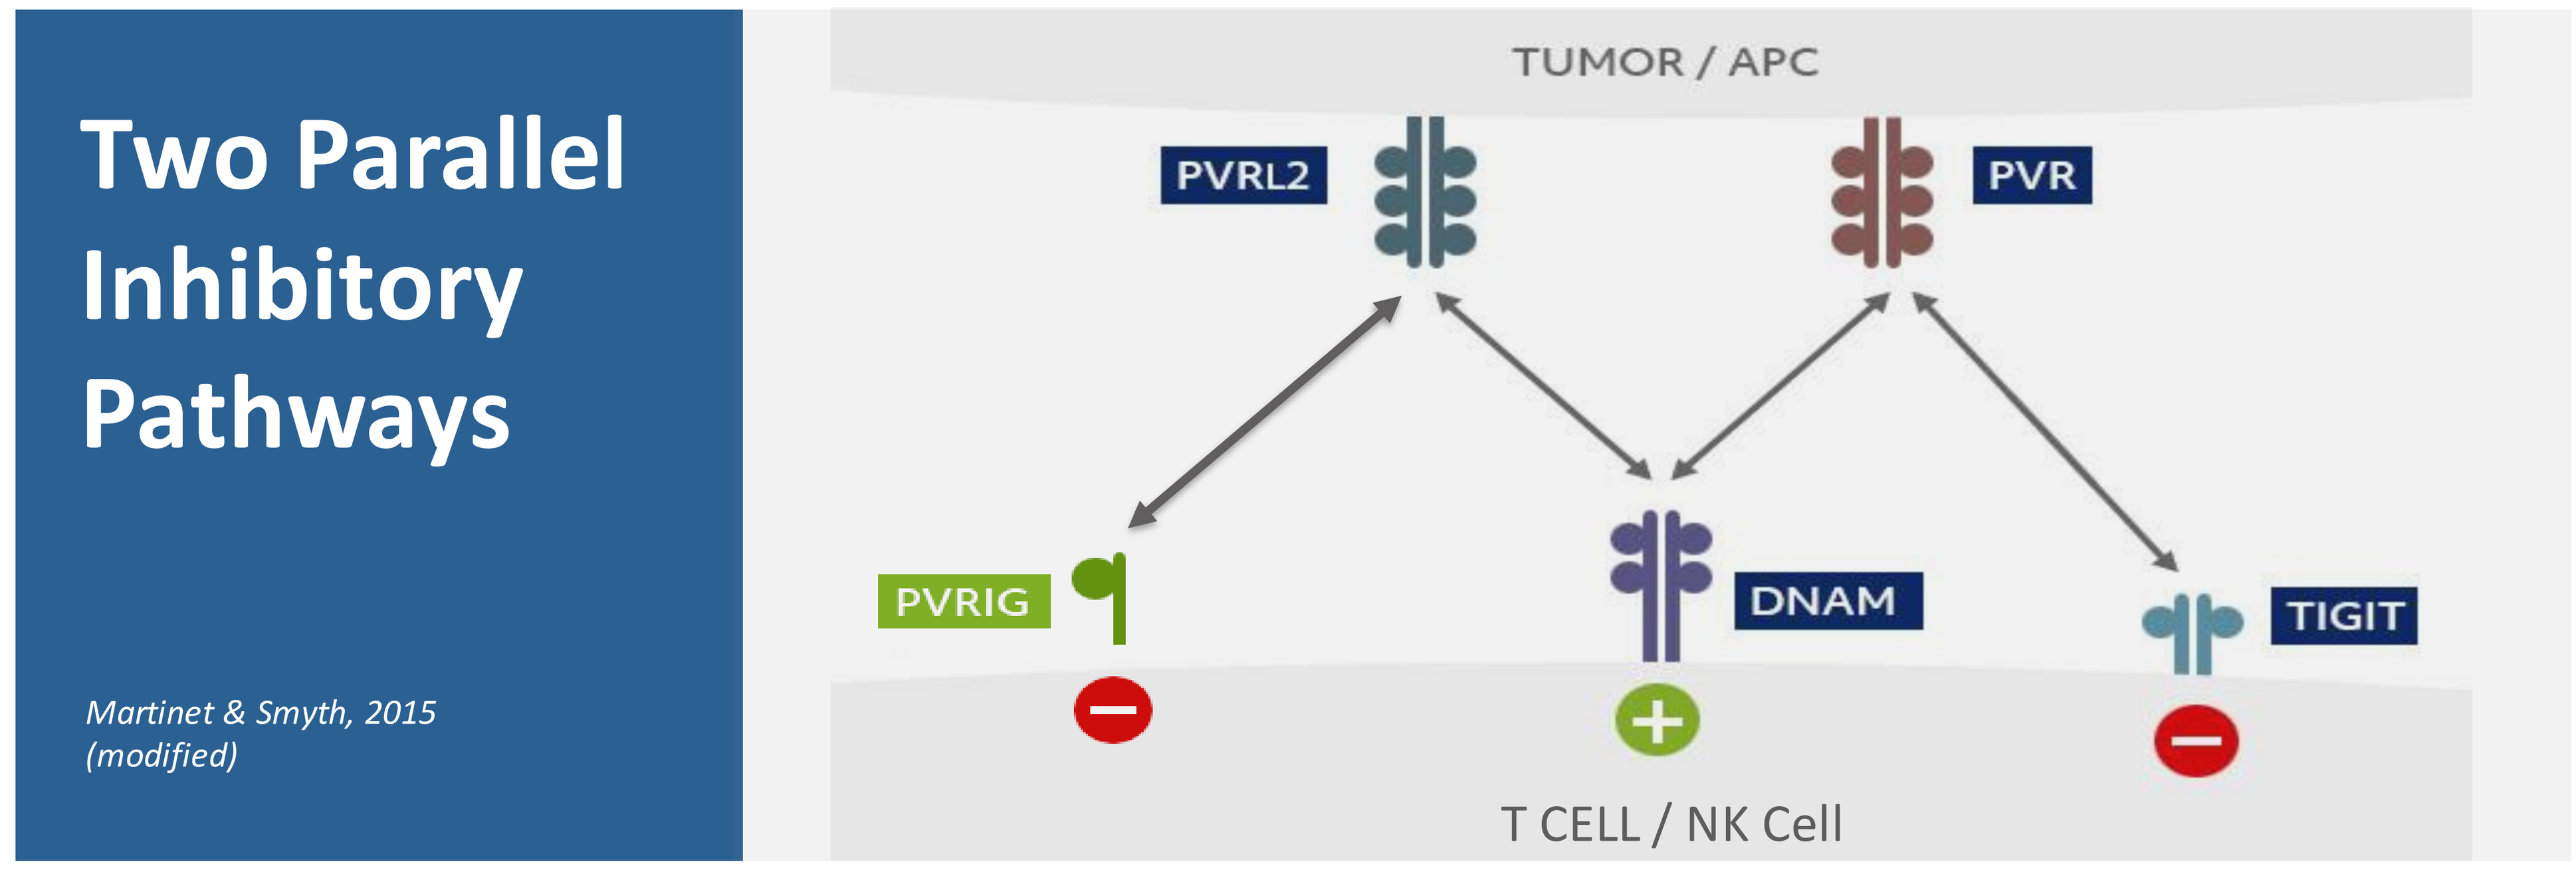

Two Parallel Inhibitory Pathways
Martinet & Smyth, 2015 (modified)
TUMOR / APC
PVRL2
PVR
PVRIG
DNAM
TIGIT
T CELL / NK Cell

PVRIG INHIBITION REDUCES TUMOR GROWTH IN MOUSE CANCER MODELS[1]

PVRIG KO MICE (MC38)

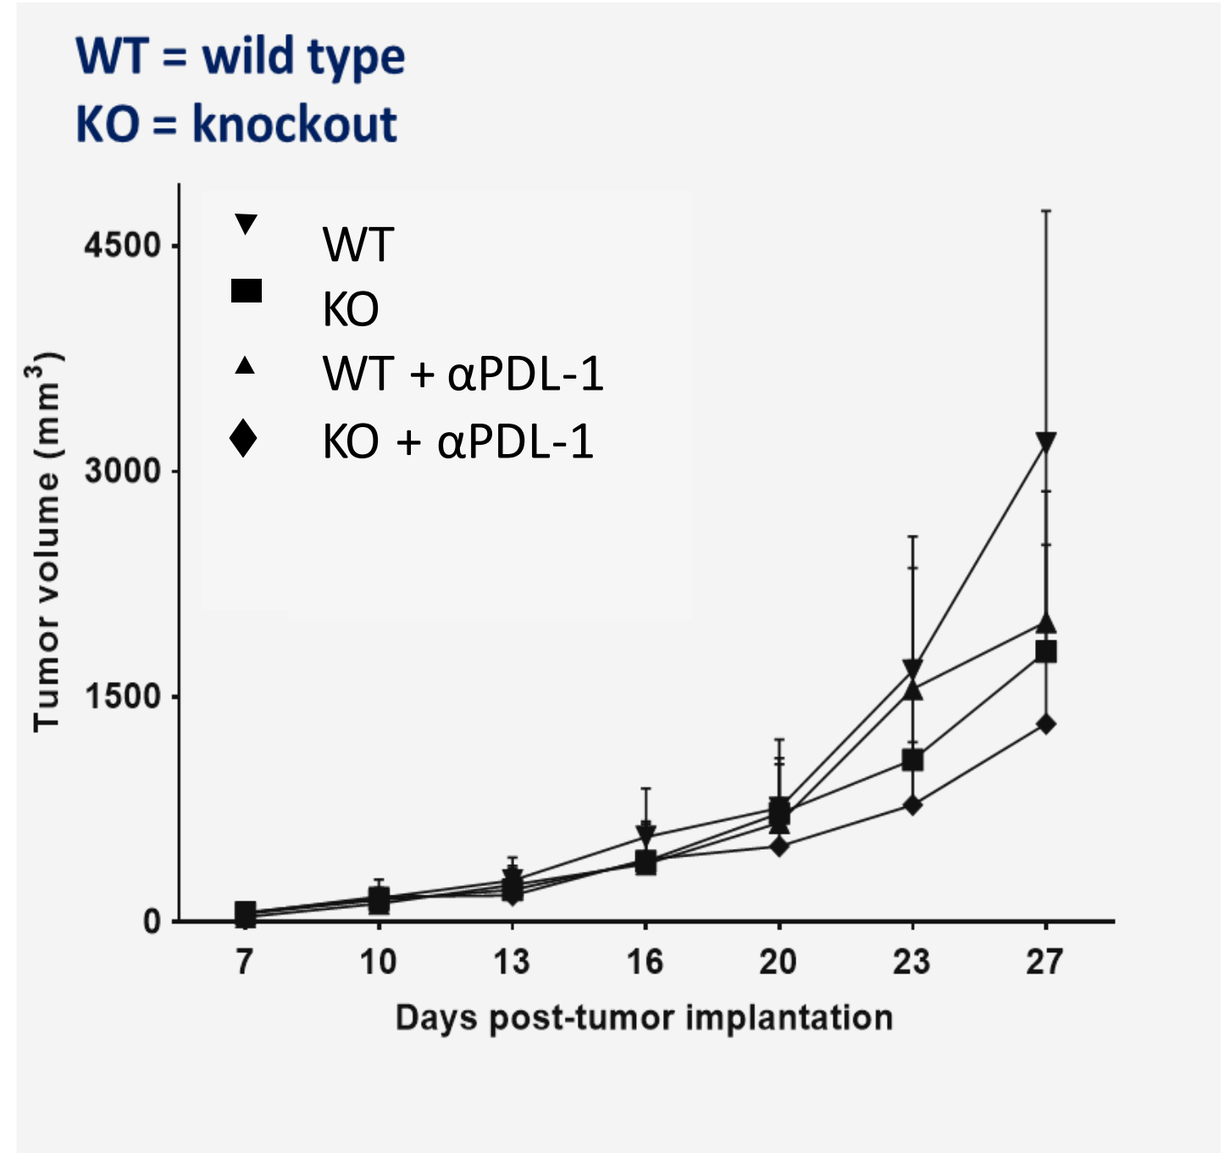

WT = wild type
KO = knockout
WT
KO
WT + αPDL-1
KO + αPDL-1
Tumor volume (mm³)
Days post-tumor implantation

Reduced tumor growth in KO mice

anti-PVRIG + anti-PDL-1 (CT26)

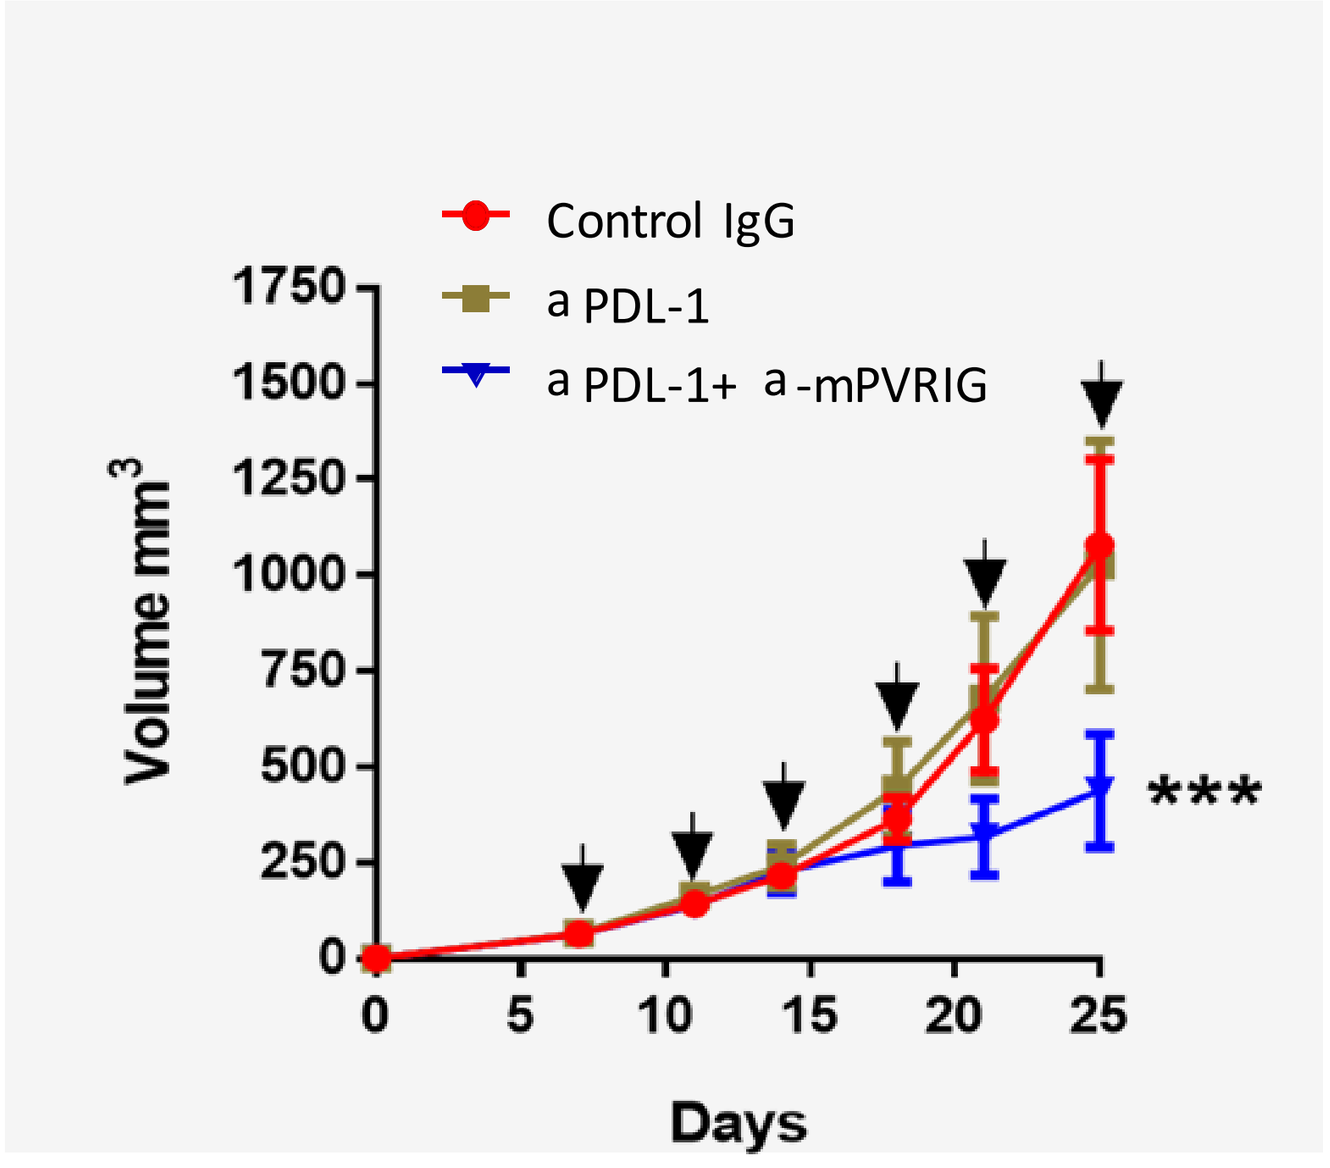

Control IgG
a PDL-1
a PDL-1+ a -mPVRIG
Volume mm³
Days

Synergistic tumor growth inhibition with anti-PD1

anti-PVRIG + TIGIT KO (B16)

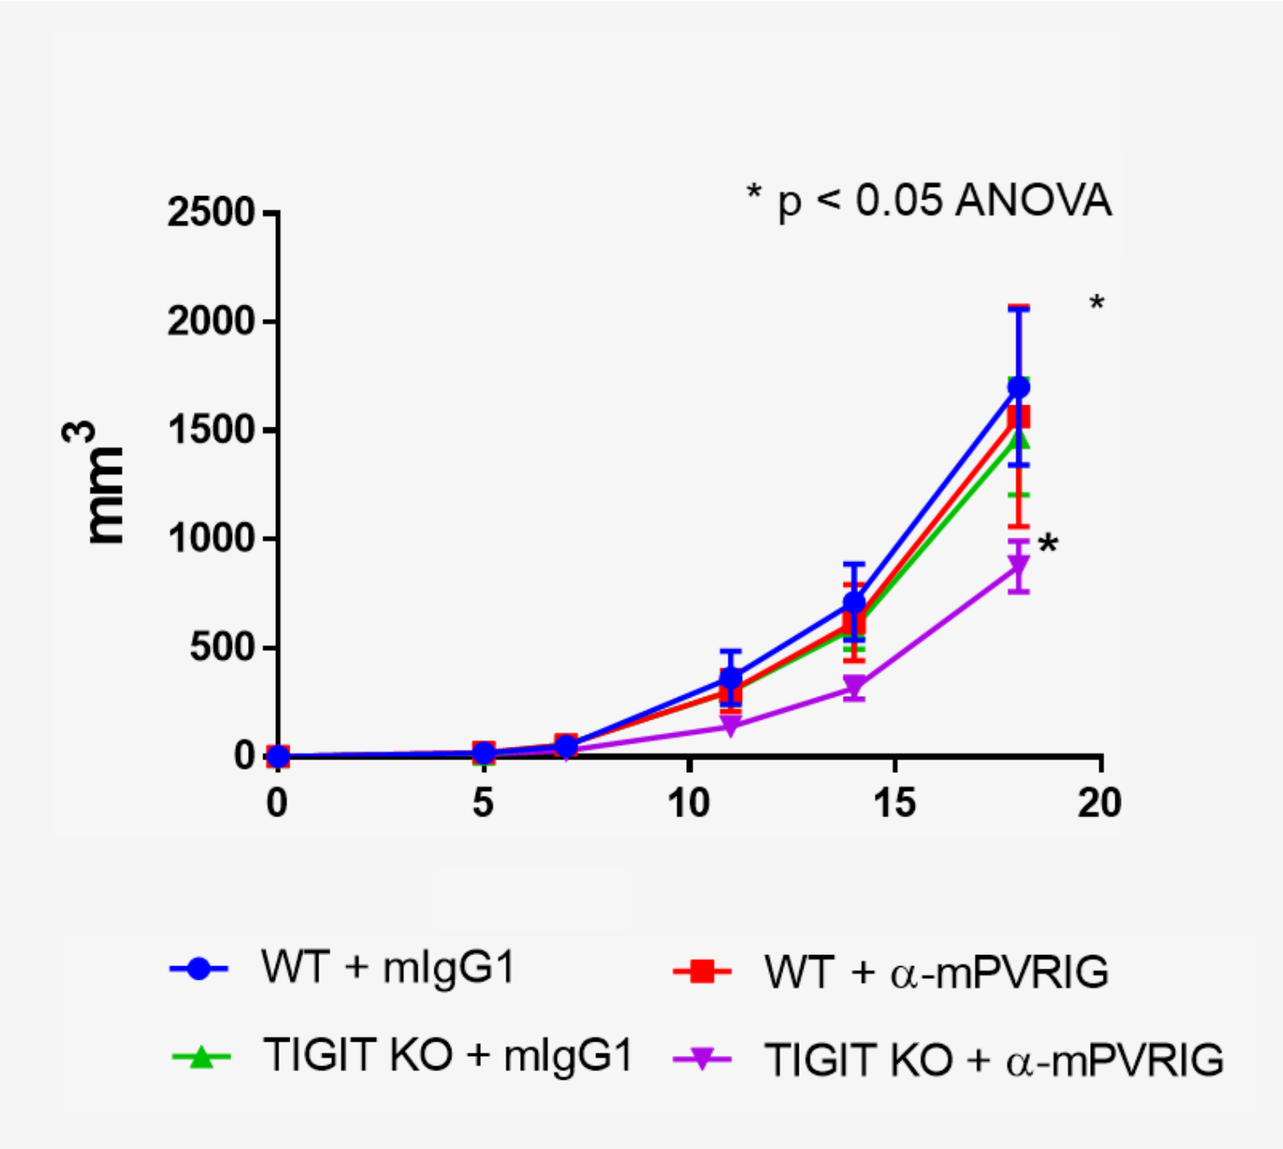

* p < 0.05 ANOVA
mm³
WT + mIgG1
WT + α-mPVRIG
TIGIT KO + mIgG1
TIGIT KO + α-mPVRIG

PVRIG inhibition required for tumor growth inhibition in TIGIT KO mice

METHODS

- NCT03667716 is an ongoing open-label first-in-human phase 1 study in pts with R/R solid tumors
- We report on the initial part of this study evaluating the safety and tolerability of escalating doses of COM701 monotherapy IV Q3 weeks and in combination with nivolumab 360mg IV Q3 weeks (red and green boxes respectively)

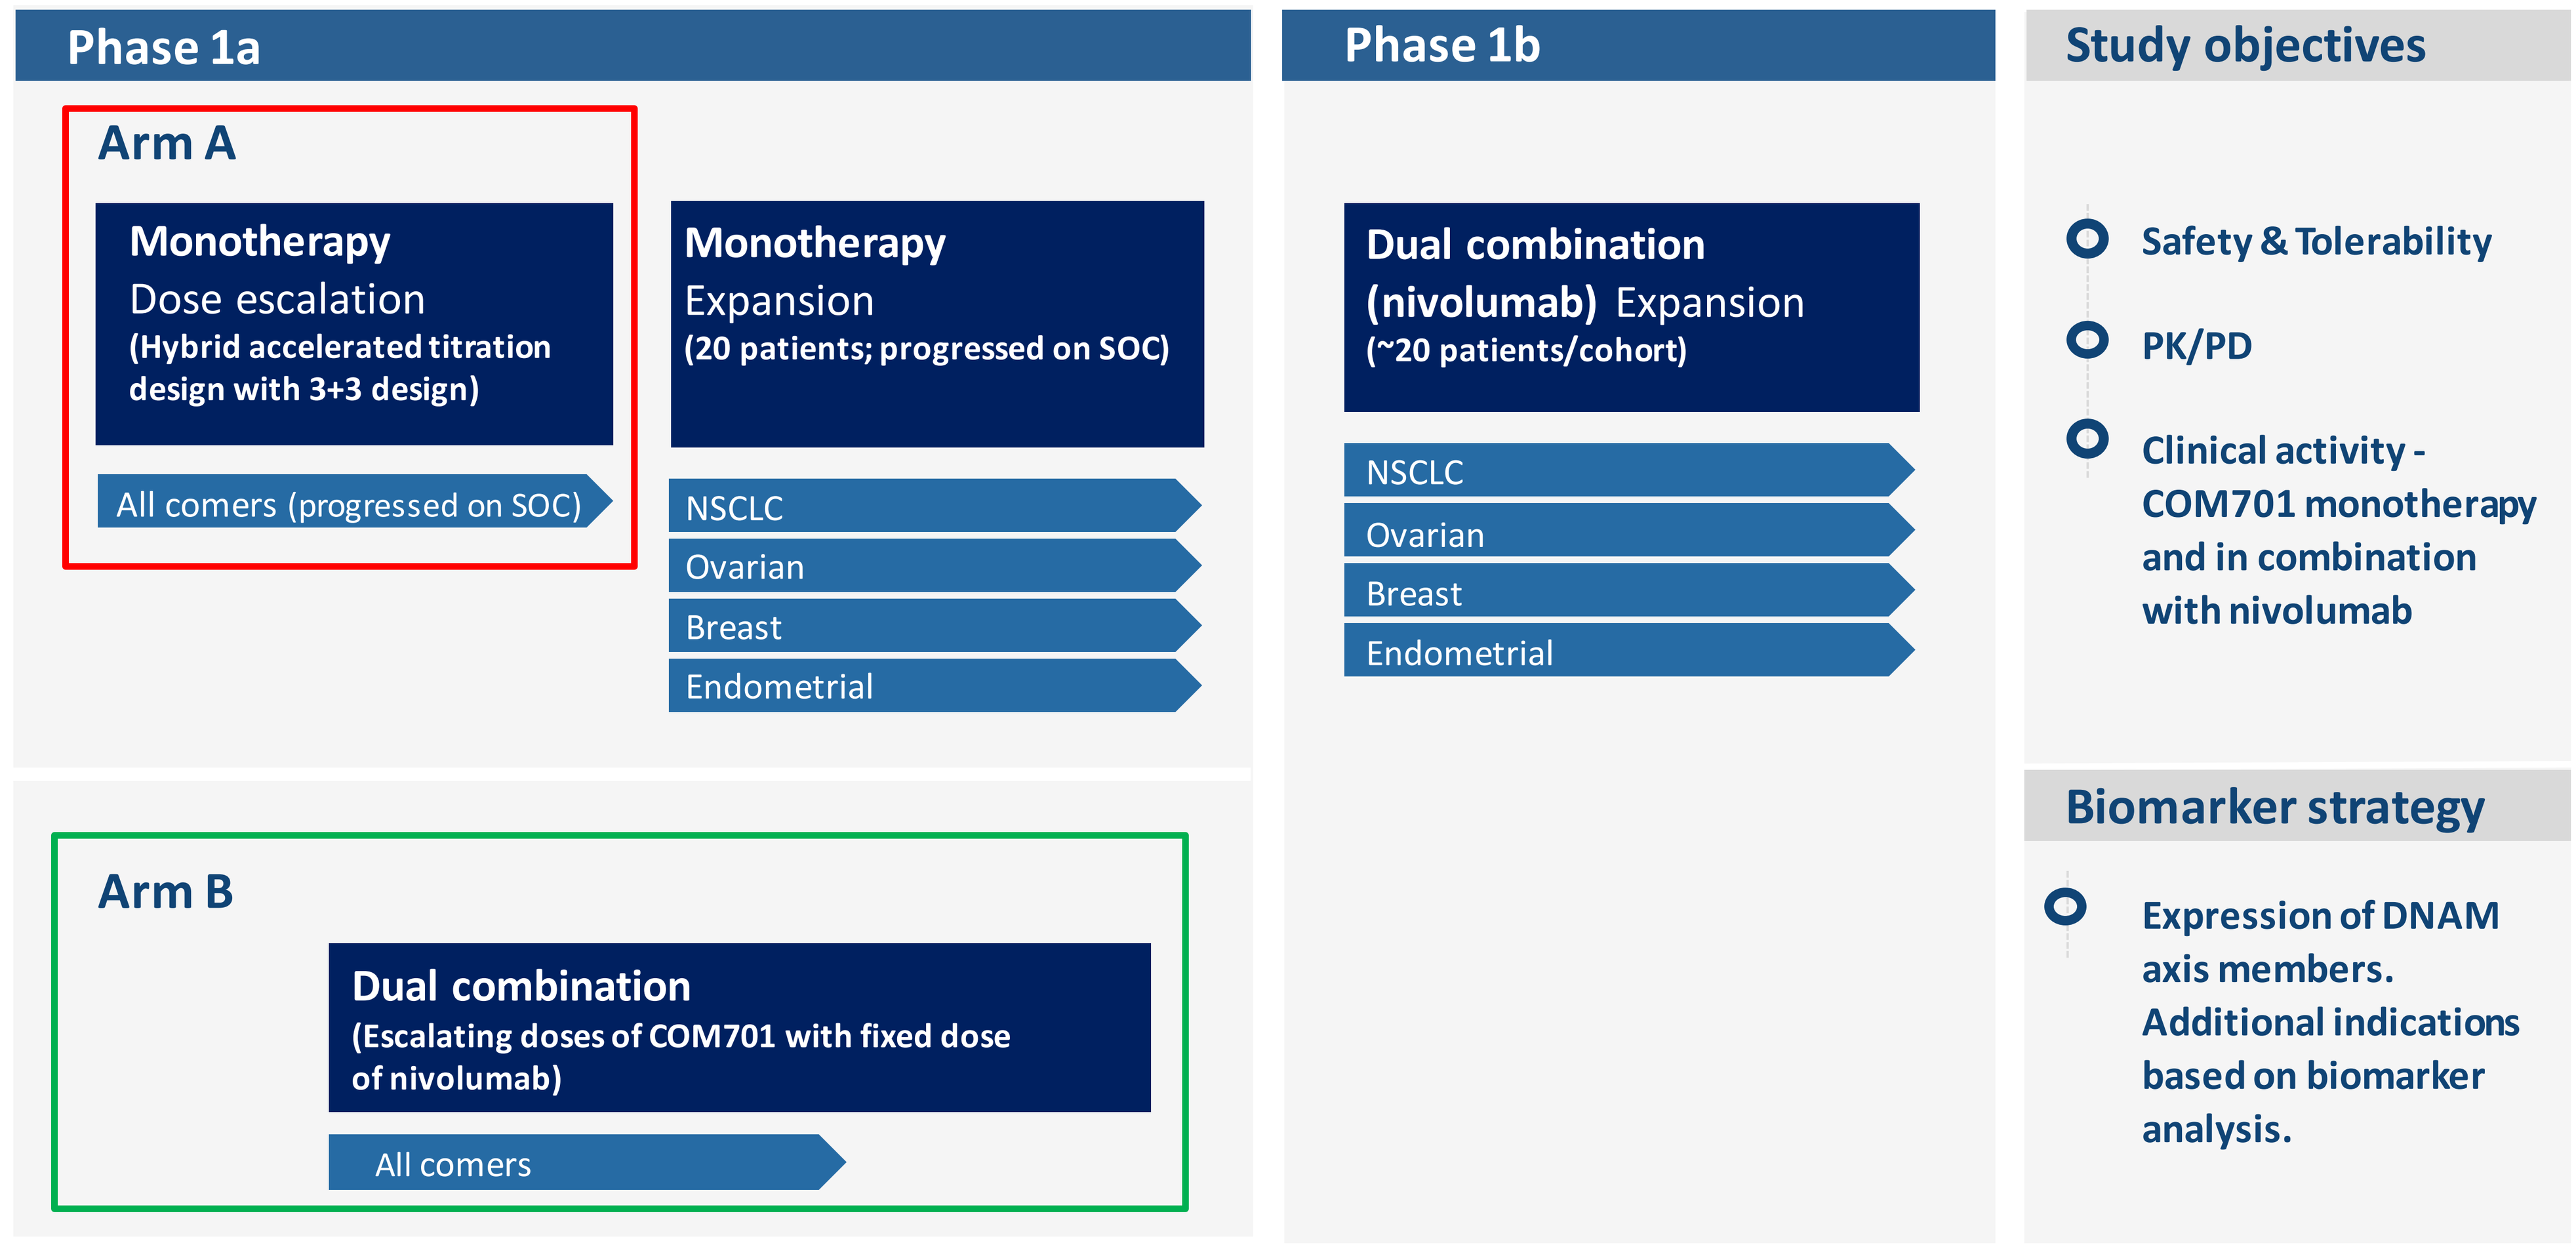

Phase 1a
Arm A
Monotherapy
Dose escalation
(Hybrid accelerated titration design with 3+3 design)
All comers (progressed on SOC)
Monotherapy
Expansion
(20 patients; progressed on SOC)
NSCLC
Ovarian
Breast
Endometrial
Arm B
Dual combination
(Escalating doses of COM701 with fixed dose of nivolumab)
All comers
Phase 1b
Dual combination (nivolumab) Expansion
(~20 patients/cohort)
NSCLC
Ovarian
Breast
Endometrial
Study objectives
Safety & Tolerability
PK/PD
Clinical activity - COM701 monotherapy and in combination with nivolumab
Biomarker strategy
Expression of DNAM axis members. Additional indications based on biomarker analysis.

PRIMARY OUTCOME MEASURES

- To evaluate the safety profile of COM701 as monotherapy and in combination with nivolumab in patients with advanced solid tumors
- The incidence of adverse events and dose-limiting toxicities (21-day DLT window) graded as per CTCAE v4.03
- To identify the maximum tolerated dose and/or the recommended dose for expansion
- To characterize the PK profile of COM701 as monotherapy and in combination with nivolumab

SECONDARY OUTCOME MEASURES

- To characterize the immunogenicity of COM701 alone and in combination with nivolumab
- To evaluate preliminary antitumor activity of COM701 in combination with nivolumab (Phase 1b only) responses as per RECIST v1.1

EXPLORATORY OUTCOME MEASURES

- To evaluate preliminary antitumor activity of COM701 as monotherapy
- To assess any association of DNAM axis members with clinical outcome
- To explore evidence of COM701-mediated PD effect in blood as monotherapy as well as in combination with nivolumab

KEY INCLUSION CRITERIA

- Age ≥18 yrs
- Histologically or cytologically confirmed, locally advanced or metastatic solid malignancy and has exhausted all the available standard therapy or is not a candidate for the available standard therapy
- ECOG performance status 0-1
- Prior anti-PD-1, anti-PD-L1, anti-CTLA-4, OX-40, CD137 permissible
- Adequate hematological, hepatic and renal function

KEY EXCLUSION CRITERIA

- Active autoimmune disease requiring systemic therapy in the last 2 years prior to the first dose of COM701
- Symptomatic interstitial lung disease or inflammatory pneumonitis
- Untreated or symptomatic central nervous system metastases
- History of immune-related events that lead to immunotherapy treatment discontinuation

ACCRUAL INFORMATION

- No dose-limiting toxicities have been observed in the 7th COM701 monotherapy dose level and earlier dose levels (red box)
- No dose-limiting toxicities have been observed in the 3rd COM701 + nivolumab dose level and earlier dose levels (green box)
- As of the date of this presentation the 8th COM701 mono dose and 4th COM701 + nivolumab dose levels are open to enrollment at IV Q4 weeks schedule

ACKNOWLEDGMENT

- We thank the patients for participating in this clinical trial and their families
- The investigators and clinical trial sites
- Study NCT03667716 is in collaboration with Bristol-Myers Squibb

REFERENCE

1. Spencer L, Ofer L et al, Discovery of COM701, a therapeutic antibody targeting the novel immune checkpoint PVRIG, for the treatment of cancer. J Clin Oncol. 2017; (suppl; abstr 3074)
2. Nivolumab package insert. http://packageinserts.bms.com/pi/pi_opdivo.pdf. Accessed 07/22/2019.
3. Wang B, Zhang W et al., Combination cancer immunotherapy targeting PD-1 and GITR can rescue CD8+ T cell dysfunction and maintain memory phenotype. Sci. Immunol. 2018; Nov 2:3(29).

Study sponsored by Compugen Ltd. For more information: www.cgen.com

Presented at SITC Annual Meeting, National Harbor, Maryland. November 6 - 10, 2019