Novel DNAM-1 Axis Member, PVRIG, is Potentially a Dominant Checkpoint Involved in Stem-Like Memory T Cell – Dendritic Cell Interaction

Zoya Alteber, Gady Cojocaru, Masha Frenkel, Emmanuel Weyl, Lital Sever, Nadav Cohen, Niv Sabath, Assaf Wool, Amit Novik, Yossef Kliger, Zurit Levine, Eran Ophir | Compugen Ltd, Holon, Israel

compugen
FROM CODE TO CURE®

BACKGROUND

T cell accumulation in tumors is a prerequisite for response to cancer immunotherapy. Recent studies highlighted the importance of an early-memory (stem-like) T cell sub-population, that can self-renew and differentiate into effector cells, and of dendritic cells (DCs), which are essential for T cell expansion following checkpoint blockade. PVRIG is a novel inhibitory receptor that competes with the co-activating receptor DNAM-1, for the binding of a shared ligand, PVRL2. PVRIG expression is induced on T and NK tumor infiltrating cells, whereas PVRL2 is expressed on tumor, endothelial and myeloid cells in the tumor micro-environment (TME). We investigated the expression of PVRIG and PVRL2 across TME immune subpopulations.
Methods: Publicly available TME scRNA sequencing datasets were analyzed for expression of PVRIG and PVRL2 across immune subsets. Gene co-expression was analyzed using either principal component analysis (PCA) or Pearson's correlation. Observations were validated by flow cytometry and immunohistochemistry analysis across a variety of tumor indications. Proximity Extension Assay (PEA, Olink) was conducted using serums collected at several time points from COM701- (anti-PVRIG antibody) and nivolumab-treated patients in a Phase-1 study (NCT03667716).

PVRIG PROTEIN HAS HIGHER EXRESSION ON EARLY MEMORY CD8+ T CELLS IN THE TME

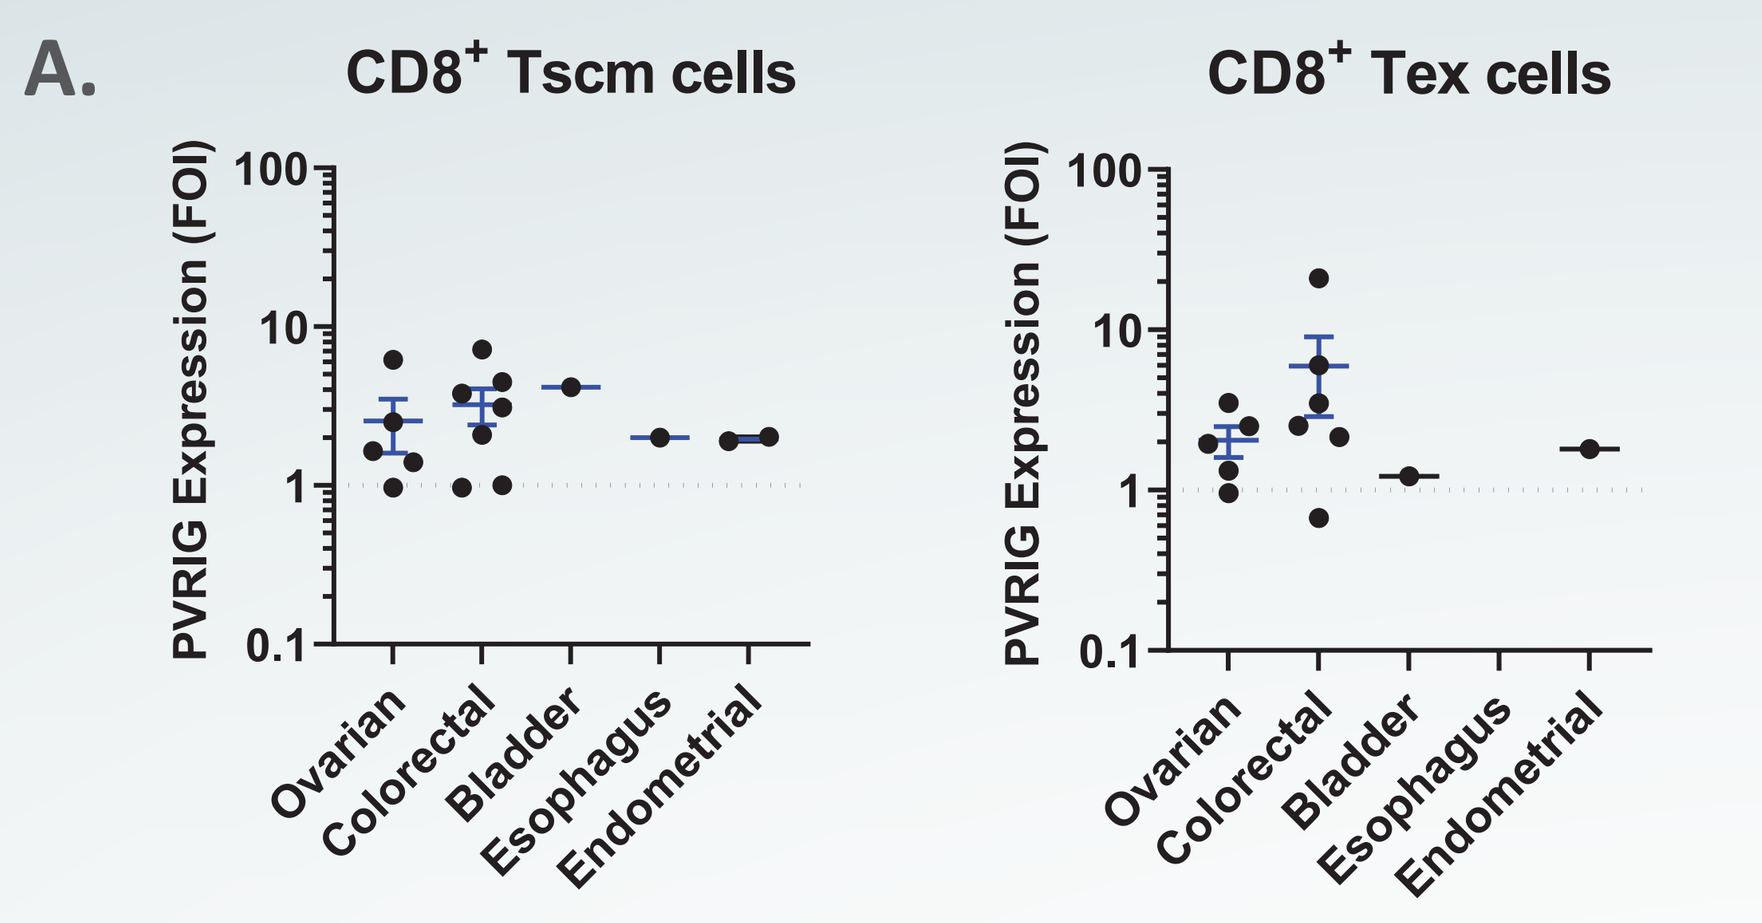
A.
CD8+ Tscm cells
CD8+ Tex cells
PVRIG Expression (FOI)
100
10
1
0.1
Ovarian
Colorectal
Bladder
Esophagus
Endometrial

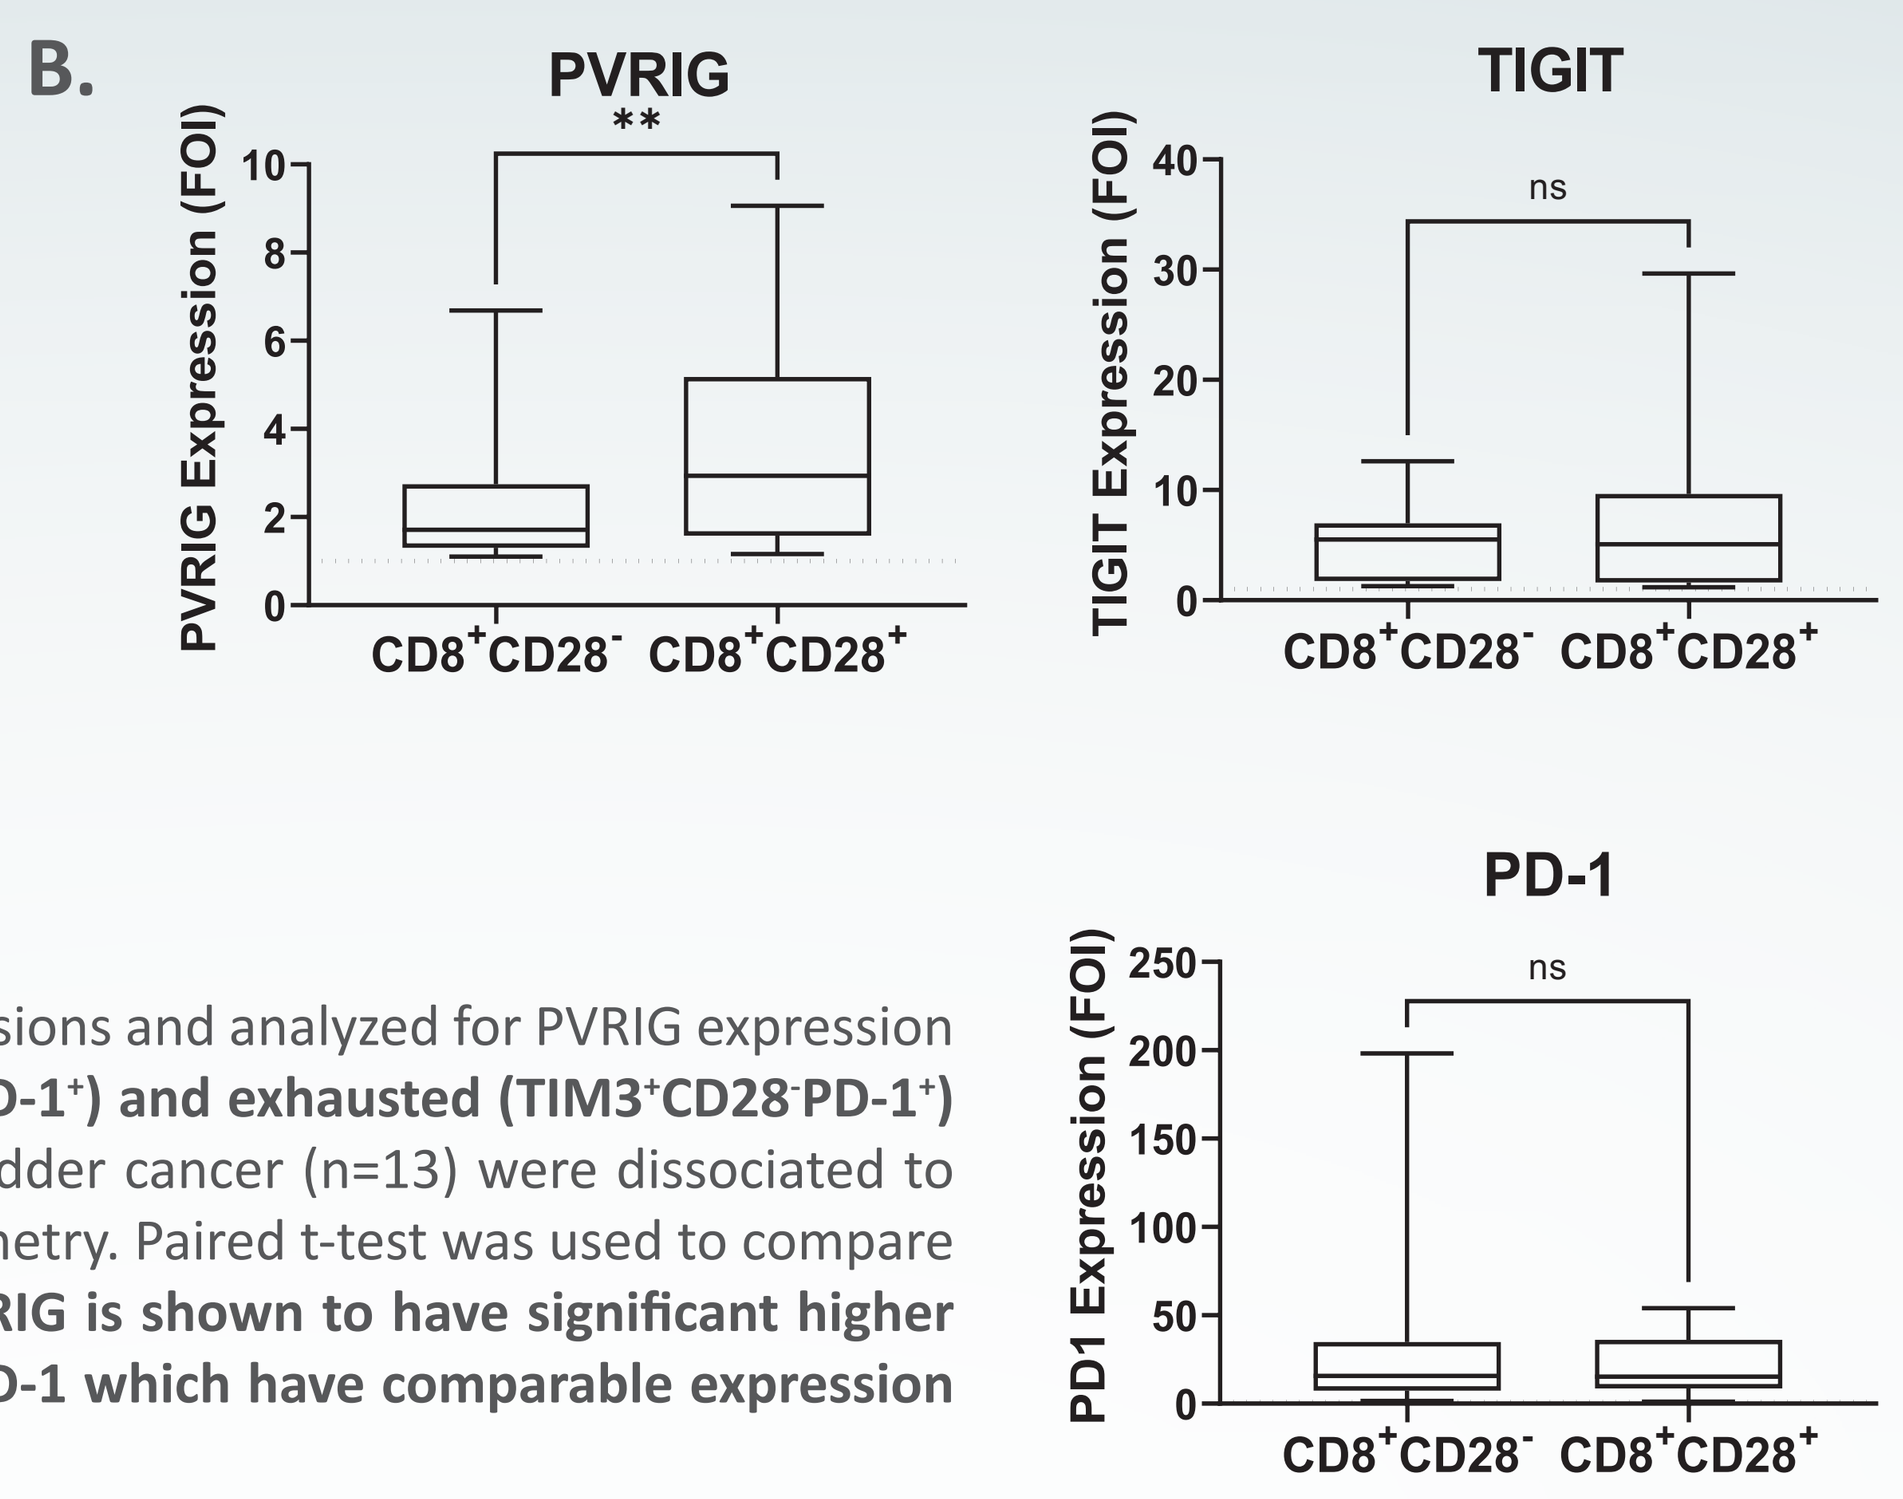
B.
PVRIG
**
PVRIG Expression (FOI)
TIGIT
ns
TIGIT Expression (FOI)
PD-1
ns
PD1 Expression (FOI)
CD8+CD28-
CD8+CD28+

A. Indicated cancer samples (n=16) were dissociated to single cell suspensions and analyzed for PVRIG expression by flow-cytometry. **PVRIG is expressed by both stem-like (TIM3-CD28+PD-1+) and exhausted (TIM3+CD28-PD-1+) CD8+ T cells B.** Samples of CRC, stomach, ovarian, endometrial and bladder cancer (n=13) were dissociated to single cell suspensions and analyzed for protein expression by flow-cytometry. Paired t-test was used to compare between PVRIG, TIGIT and PD-1 expression among cell populations. **PVRIG is shown to have significant higher expression on early memory (CD28+) T cells, in contrast to TIGIT and PD-1 which have comparable expression on CD28+ and CD28- T cells**

PVRL2 IS EXPRESSED IN TERTIARY LYMPHOID STRUCTURES IN THE TUMOR BED

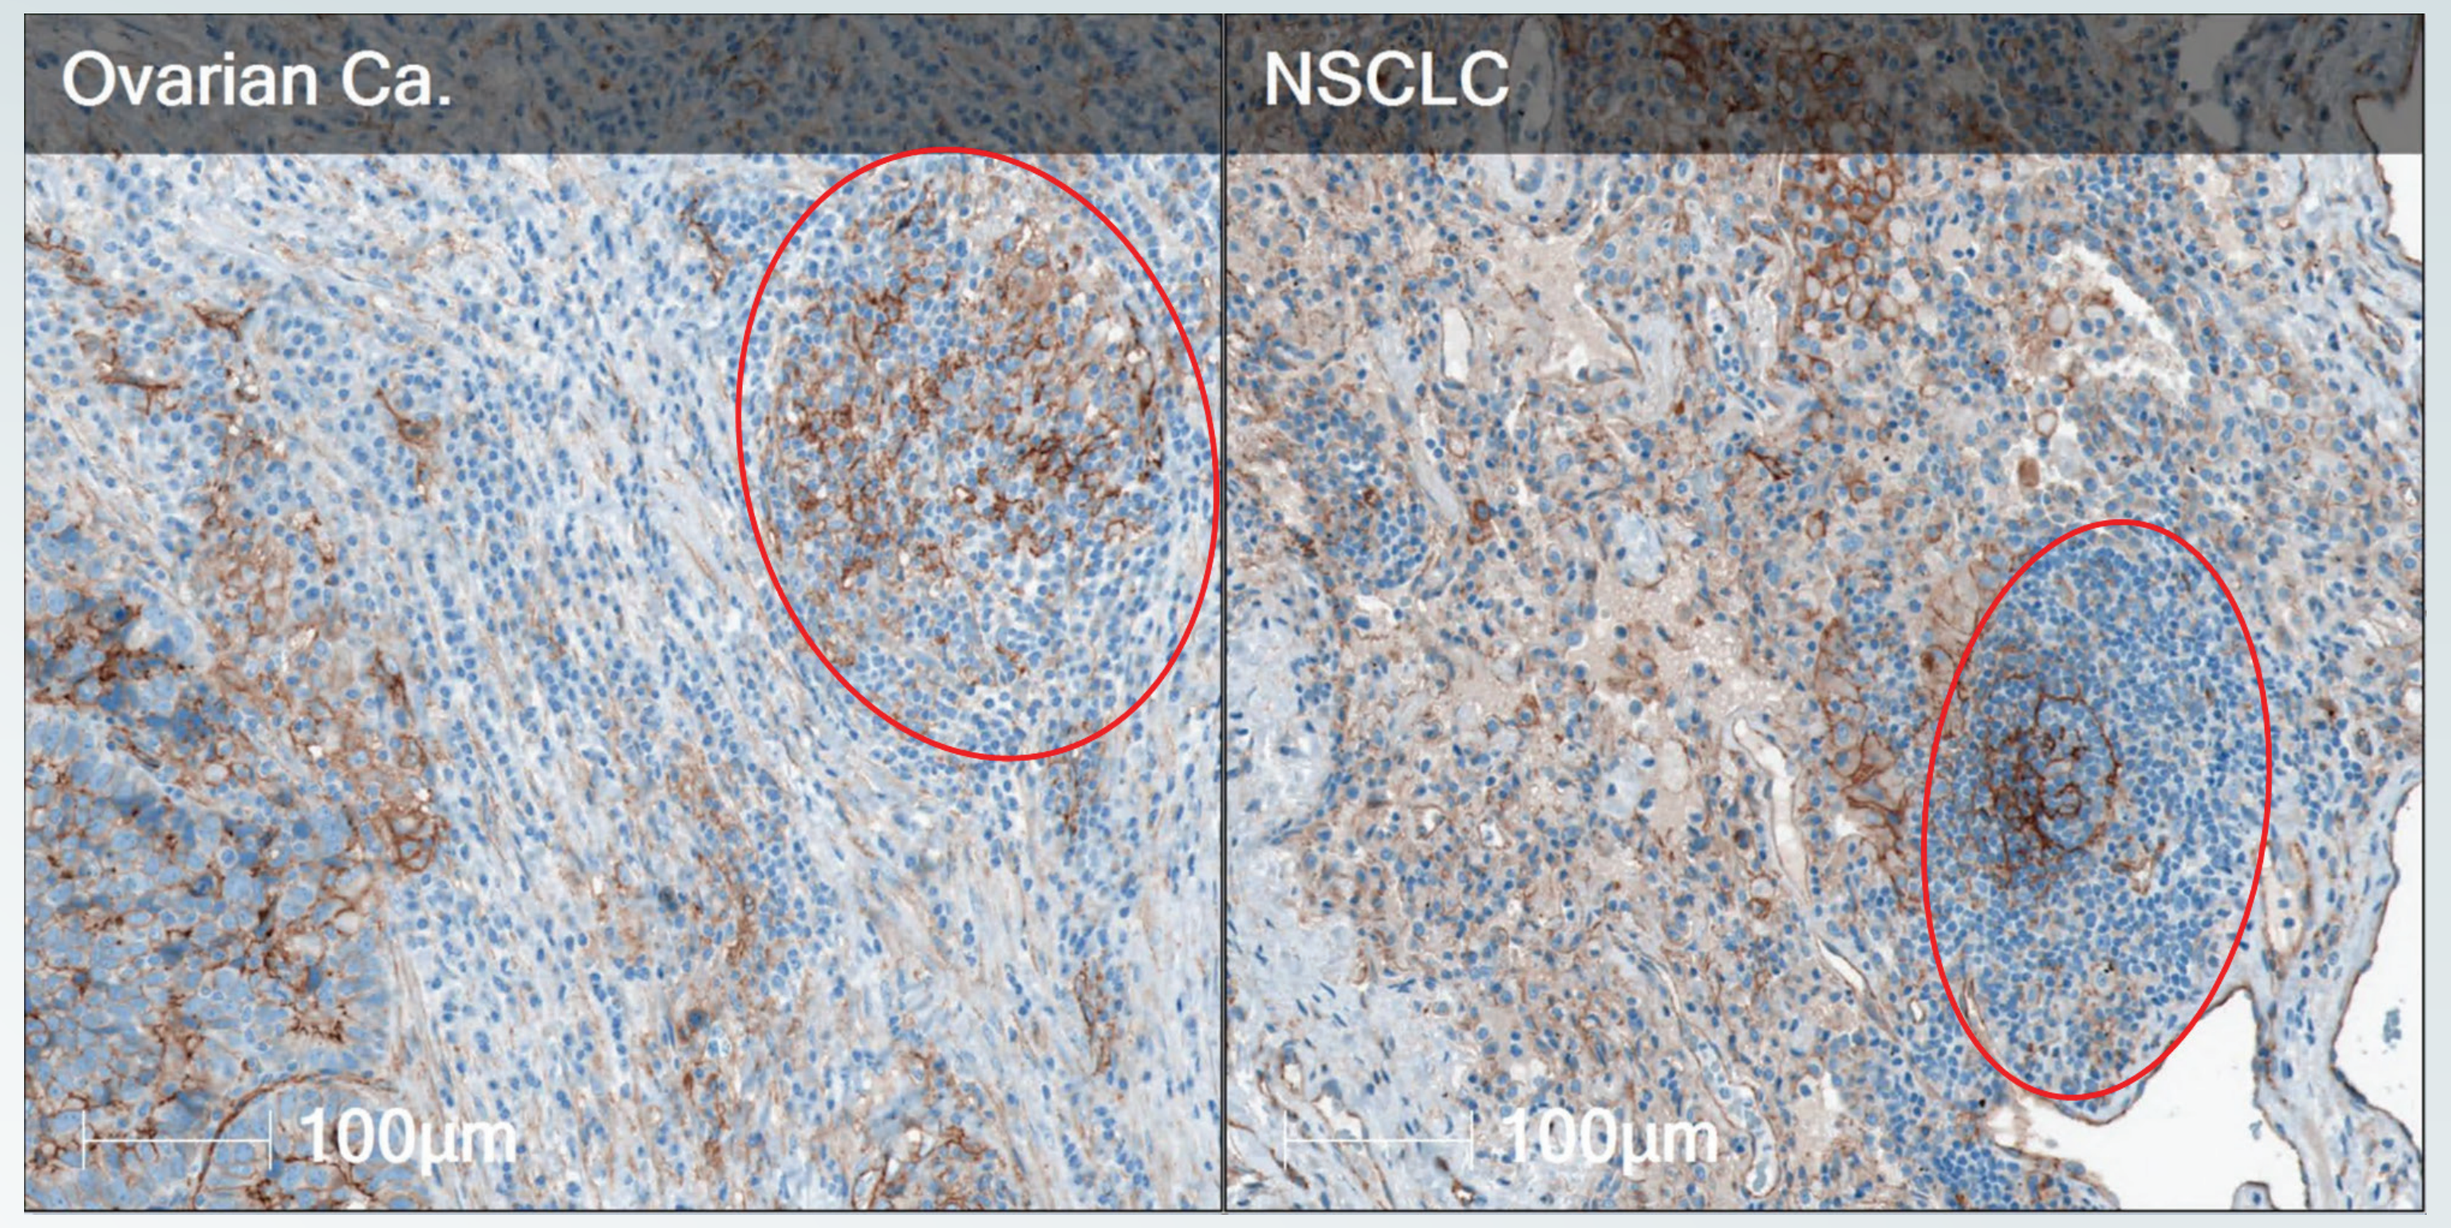
Ovarian Ca.
NSCLC
100µm
100µm

Tertiary Lymphoid Structures (TLSs) were identified in subsets of samples across all tested tumors (NSCLC, CRC primary and metastasis, ovarian cancer, endometrial cancer, breast primary TNBC and breast metastasis) and for most cases TLSs were positive for PVRL2. Staining was preformed using a proprietary rabbit mAb raised against the ECD of PVRL2 on a Dako Autostainer.

PVRIG UNIQUELY CLUSTERS WITH MARKERS OF EARLY MEMORY (STEM-LIKE) CD8+ T CELLS IN TME CD8+ T CELLS

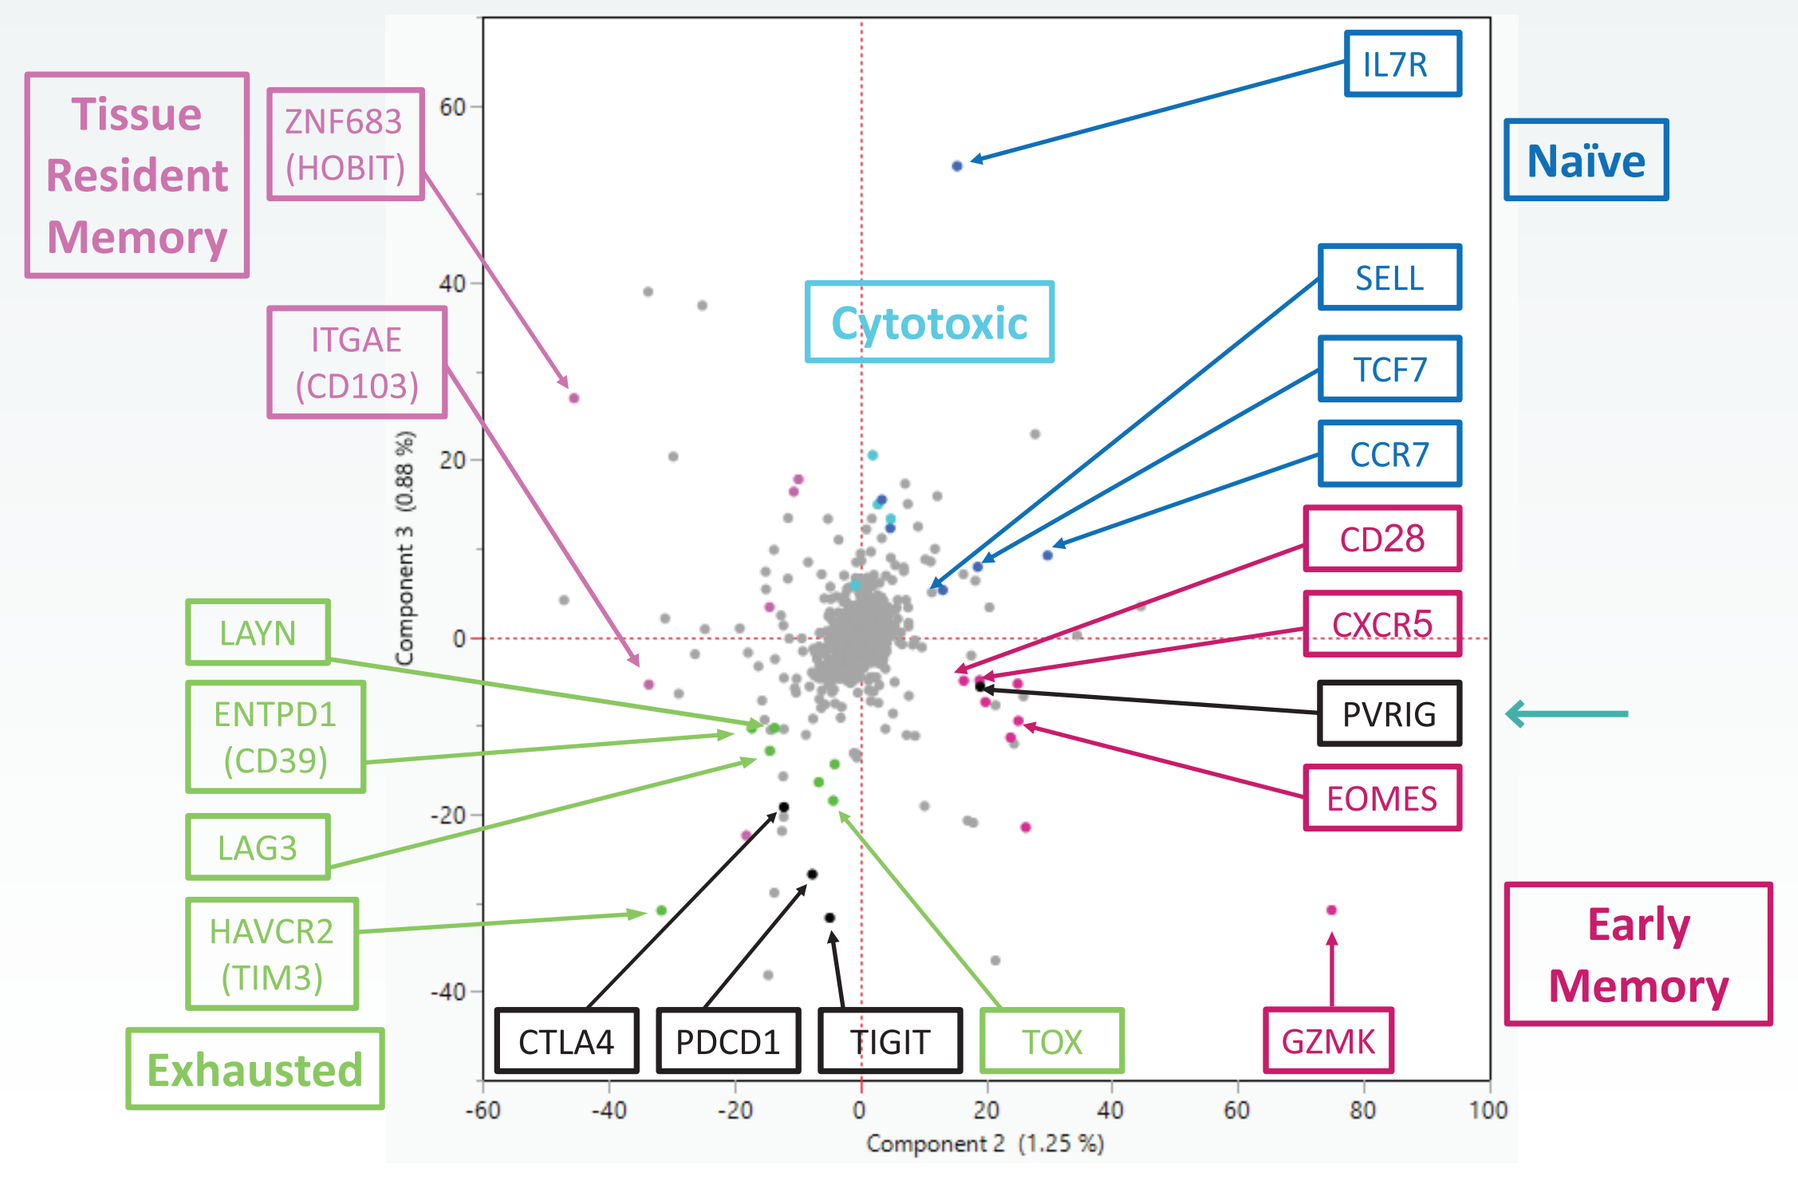
A.
Tissue Resident Memory
ZNF683 (HOBIT)
ITGAE (CD103)
LAYN
ENTPD1 (CD39)
LAG3
HAVCR2 (TIM3)
Exhausted
CTLA4
PDCD1
TIGIT
TOX
GZMK
Cytotoxic
IL7R
Naïve
SELL
TCF7
CCR7
CD28
CXCR5
PVRIG
EOMES
Early Memory
Component 3 (0.88 %)
Component 2 (1.25 %)

GSE99254_NSCLC Dataset internally analyzed

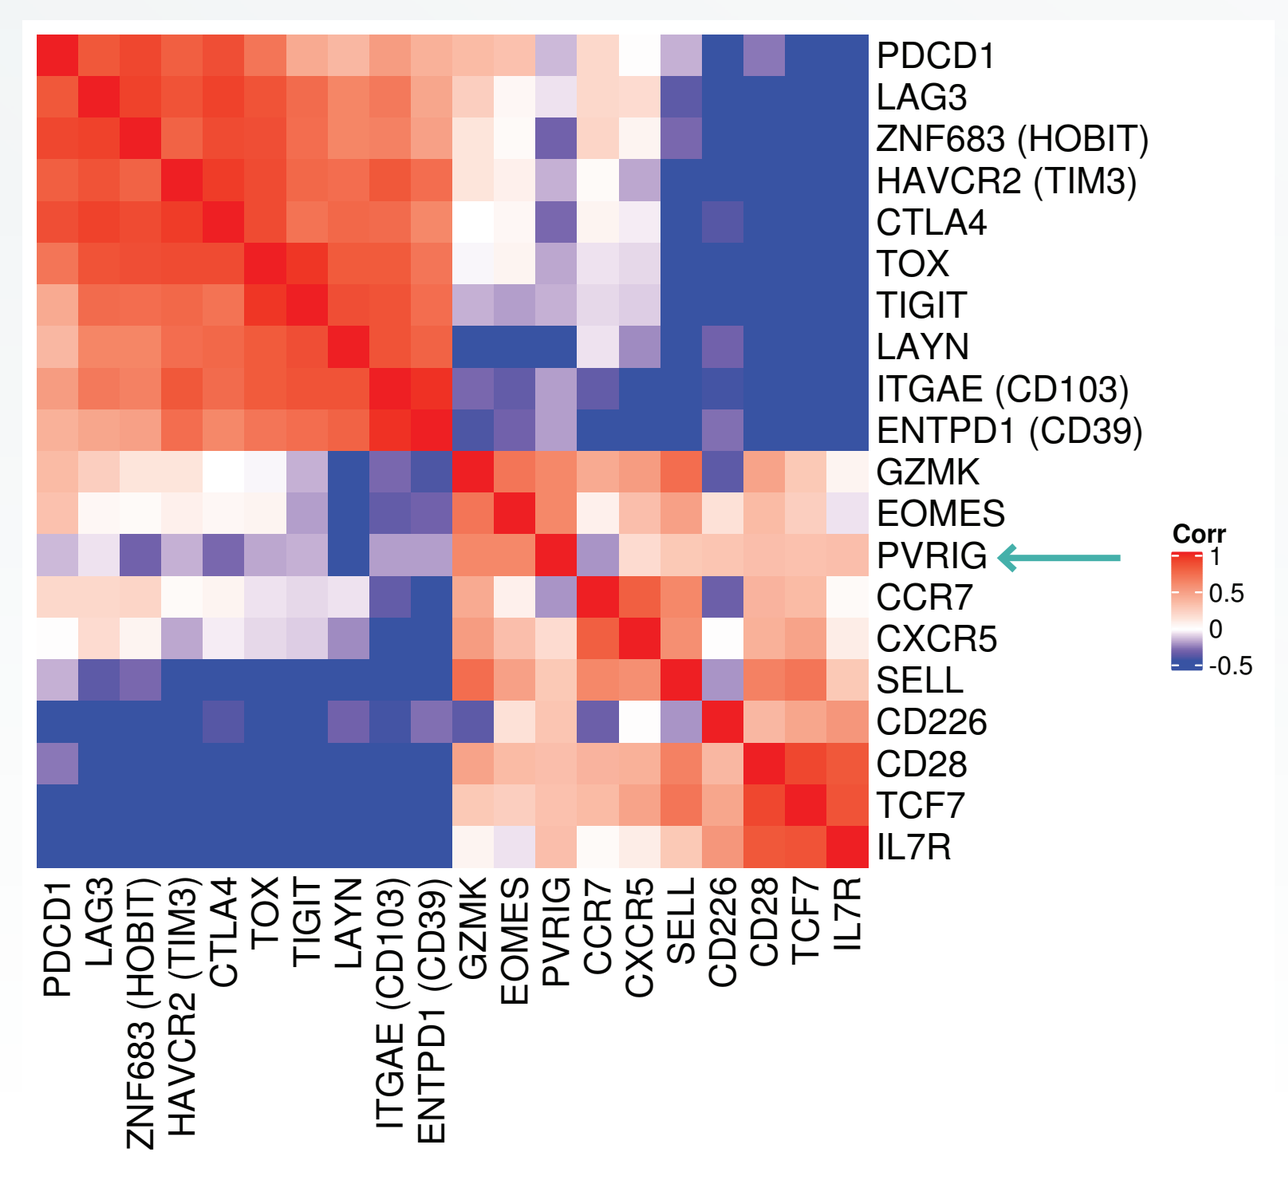
B.
PDCD1
LAG3
ZNF683 (HOBIT)
HAVCR2 (TIM3)
CTLA4
TOX
TIGIT
LAYN
ITGAE (CD103)
ENTPD1 (CD39)
GZMK
EOMES
PVRIG
CCR7
CXCR5
SELL
CD226
CD28
TCF7
IL7R
Corr
1
0.5
0
-0.5

GSE108989_CRC Dataset internally analyzed

A. Unsupervised PCA was performed on scRNA expression matrix of TME CD8+ T cells, which includes all variable genes. Using cells as features and genes as entries, co-expression pattern among genes known to be expressed on naïve, memory, and exhausted CD8+ T cells is shown. Nine publicly available datasets were analyzed, representative NSCLC data set is shown. **B.** Publicly available scRNA-seq datasets (CRC, NSCLC, HNSCC, Melanoma, Liver cancer, n=13) were analyzed for co-expression pattern among 19 genes, including genes known to be expressed on naïve (TCF7, IL7R, SELL), memory (GZMK, EOMES), and exhausted (PDCD1, LAG3, HAVCR2) CD8+ T cells. Representative dataset of CRC is presented.

PVRL2 IS DOMINANTLY EXPRESSED ON DENDRITIC CELLS

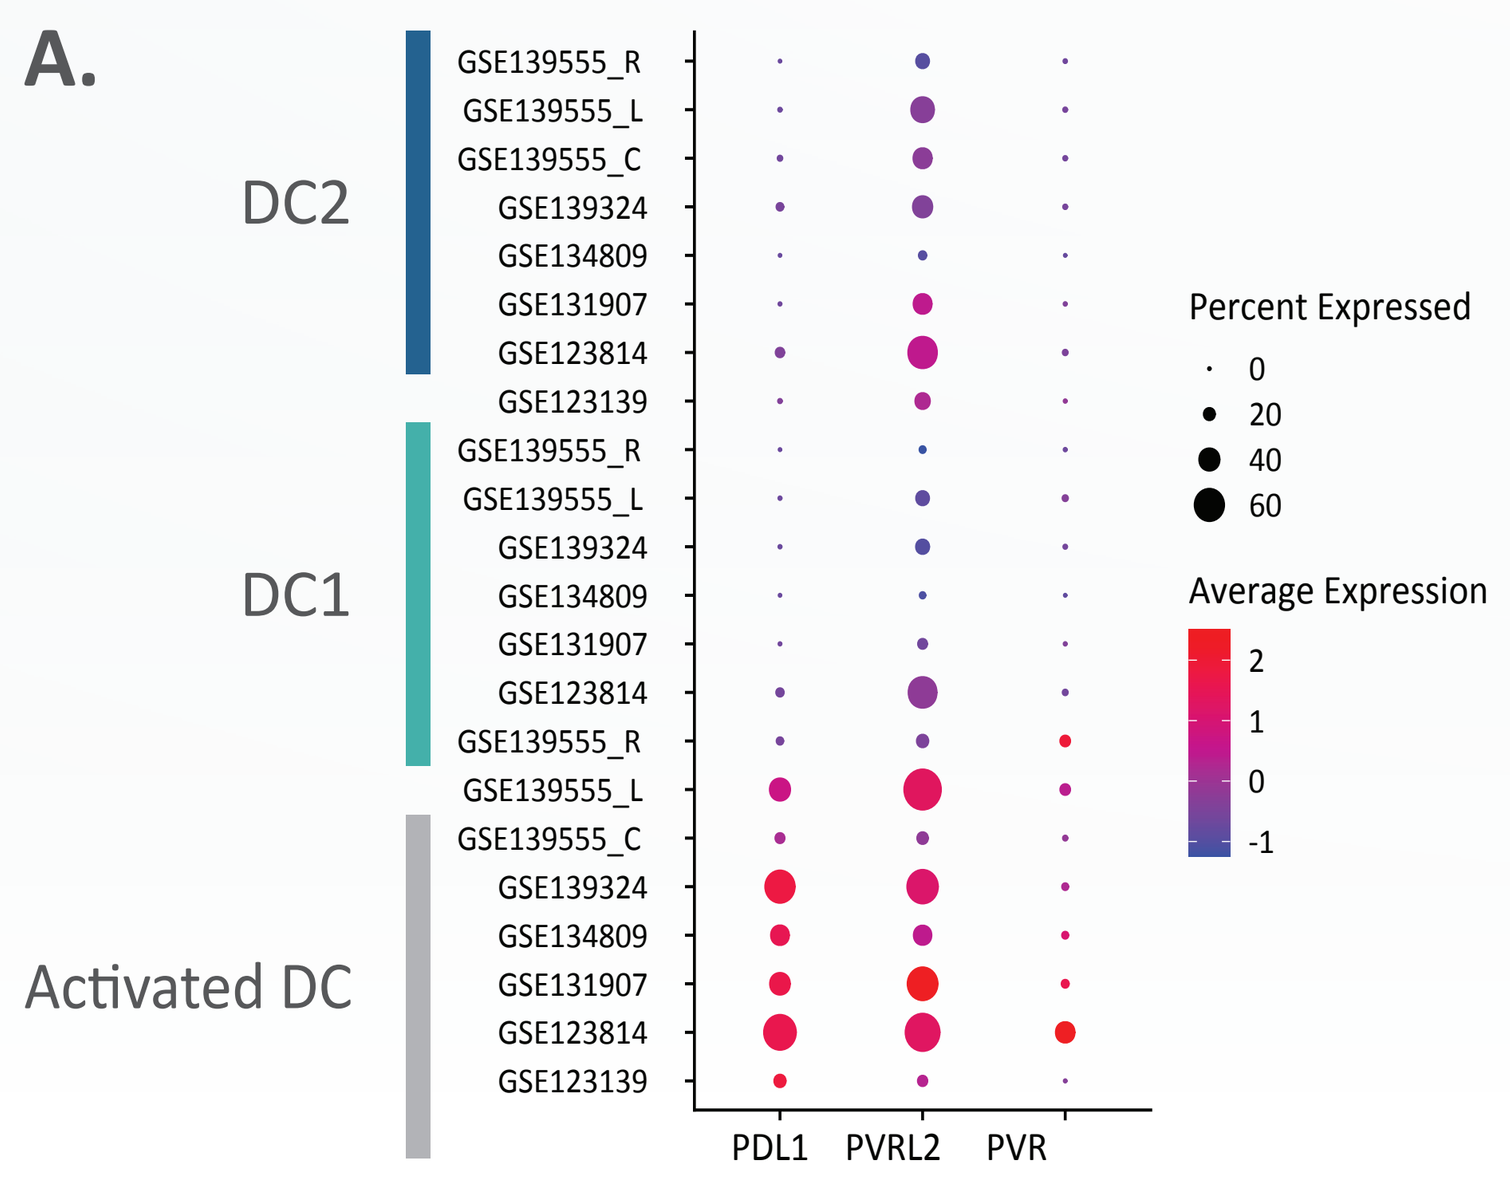
A.
DC2
DC1
Activated DC
GSE139555_R
GSE139555_L
GSE139555_C
GSE139324
GSE134809
GSE131907
GSE123814
GSE123139
PDL1
PVRL2
PVR
Percent Expressed
0
20
40
60
Average Expression
2
1
0
-1

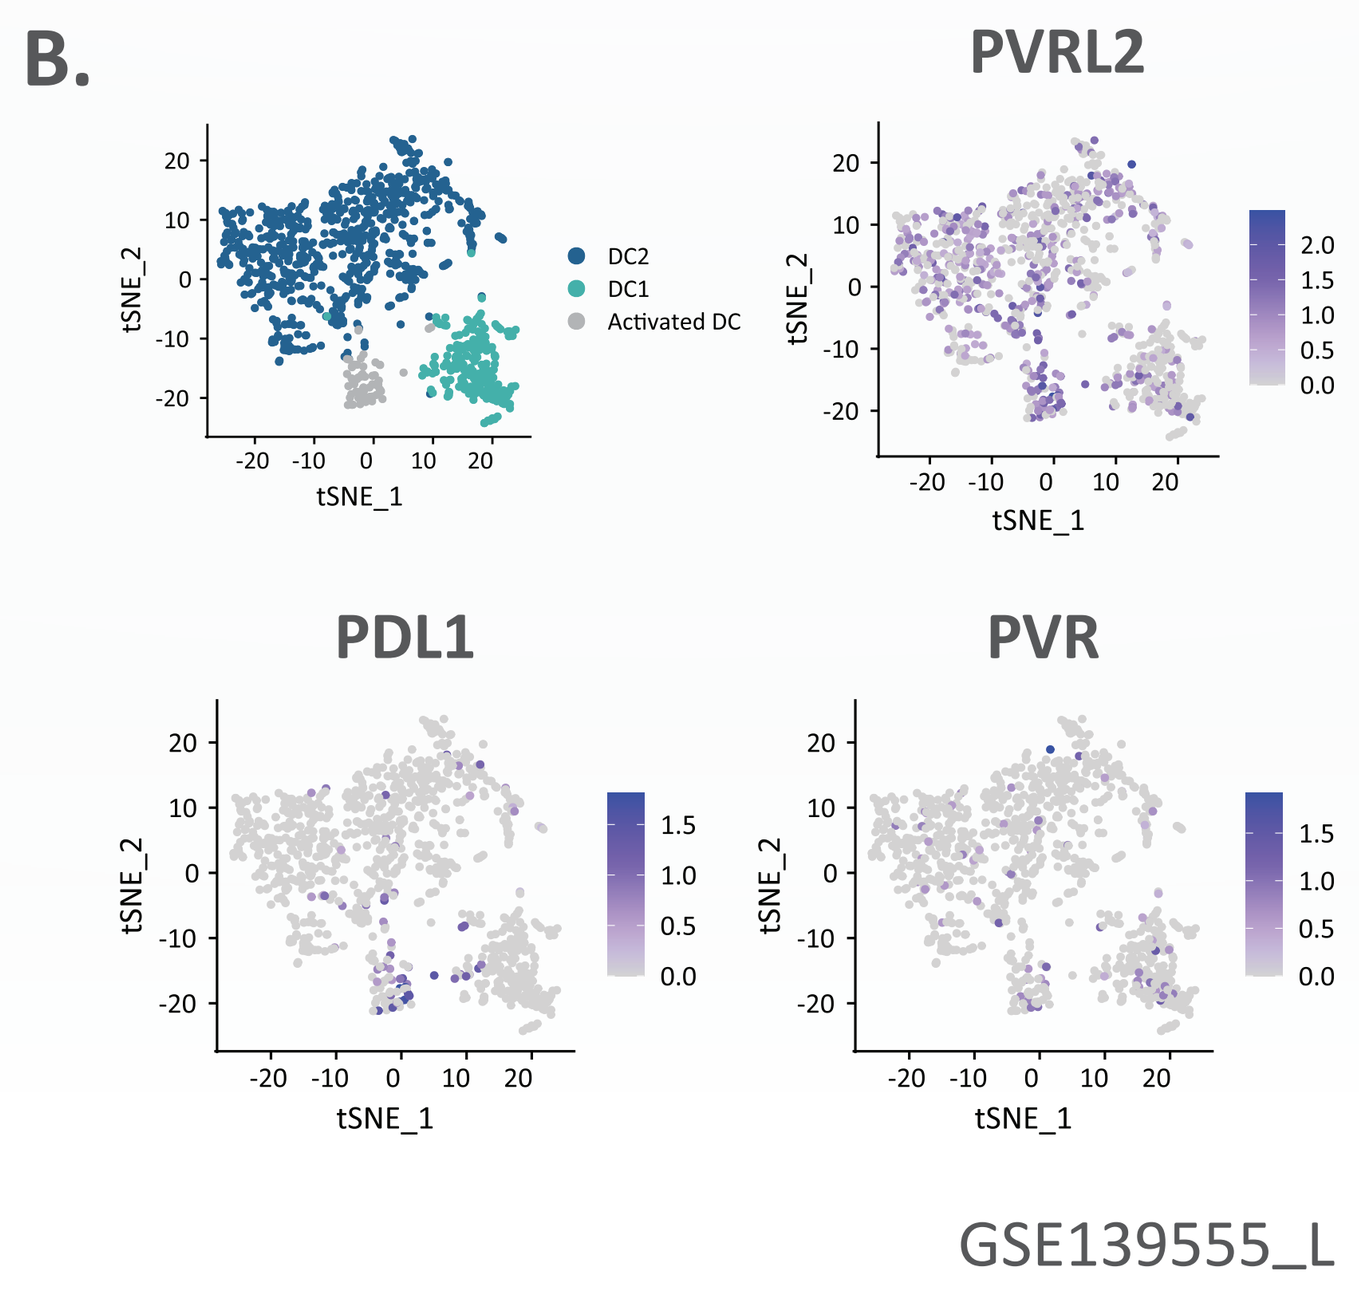
B.
DC2
DC1
Activated DC
PVRL2
PDL1
PVR
tSNE_1
tSNE_2
GSE139555_L

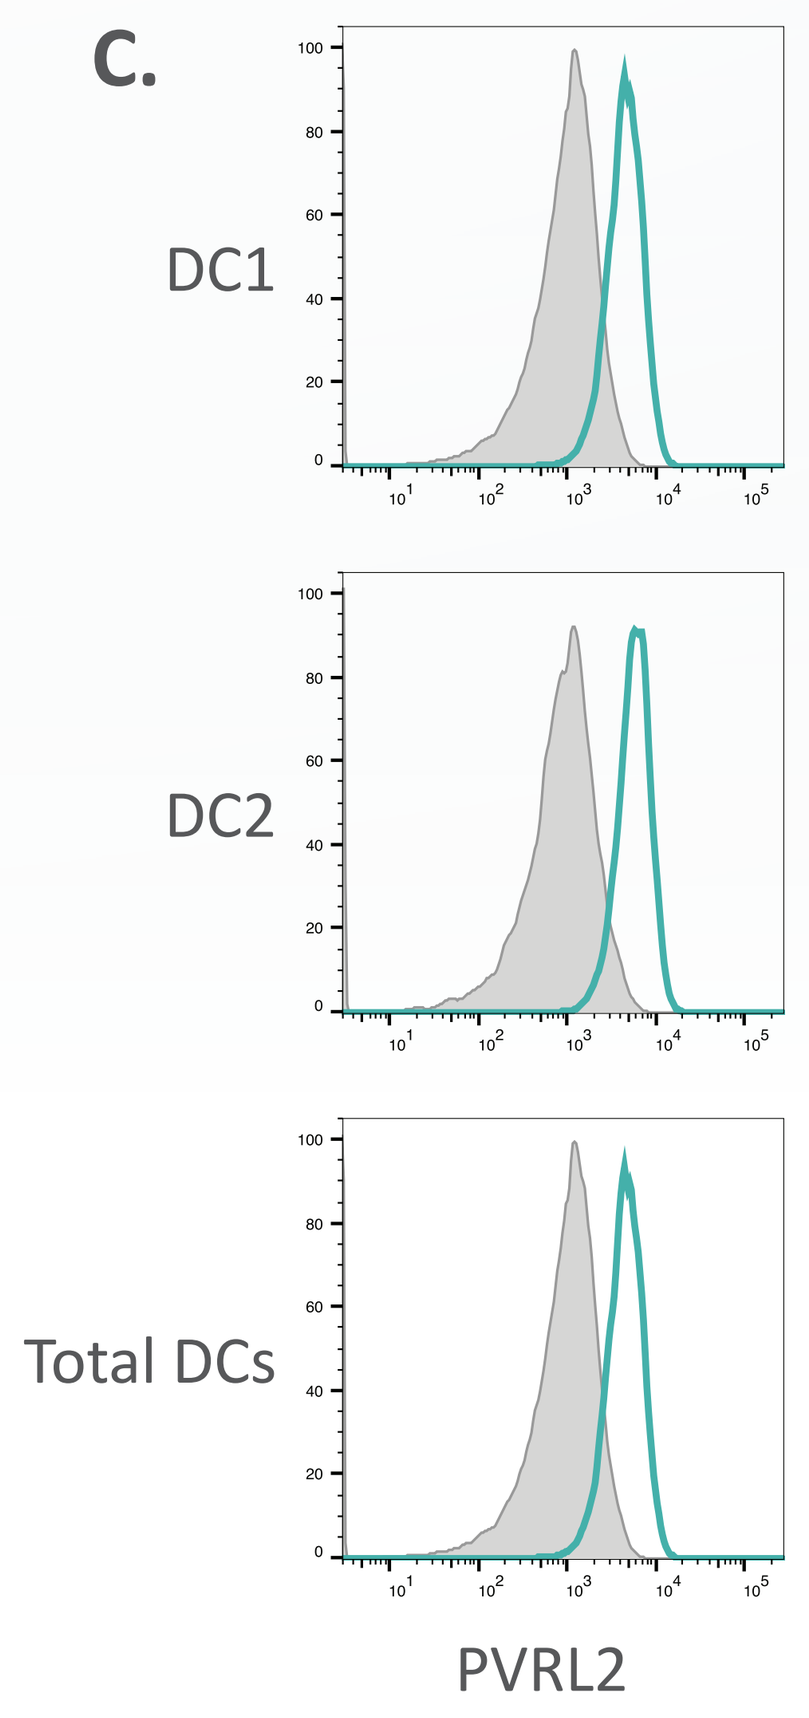
C.
DC1
DC2
Total DCs
PVRL2

A. Dot plots showing the percent of cells and average level of expression of PVR/PVRL2/PD-L1 in major dendritic cell subsets across multiple publicly available scRNA-seq cancer datasets. **B.** Representative tSNE map depicting the expression profile of PVR/PVRL2/PD-L1 in major dendritic cell subsets in NSCLC. **C.** PVRL2 protein expression across DC subsets in a representative ovarian cancer sample analyzed by flow-cytometry.

ELEVATED INDUCTION OF ACTIVATED-DC MARKERS IN PATIENTS THAT CLINICALLY RESPONDED TO COM701+NIVOLUMAB, COMPARED TO NON-RESPONDERS

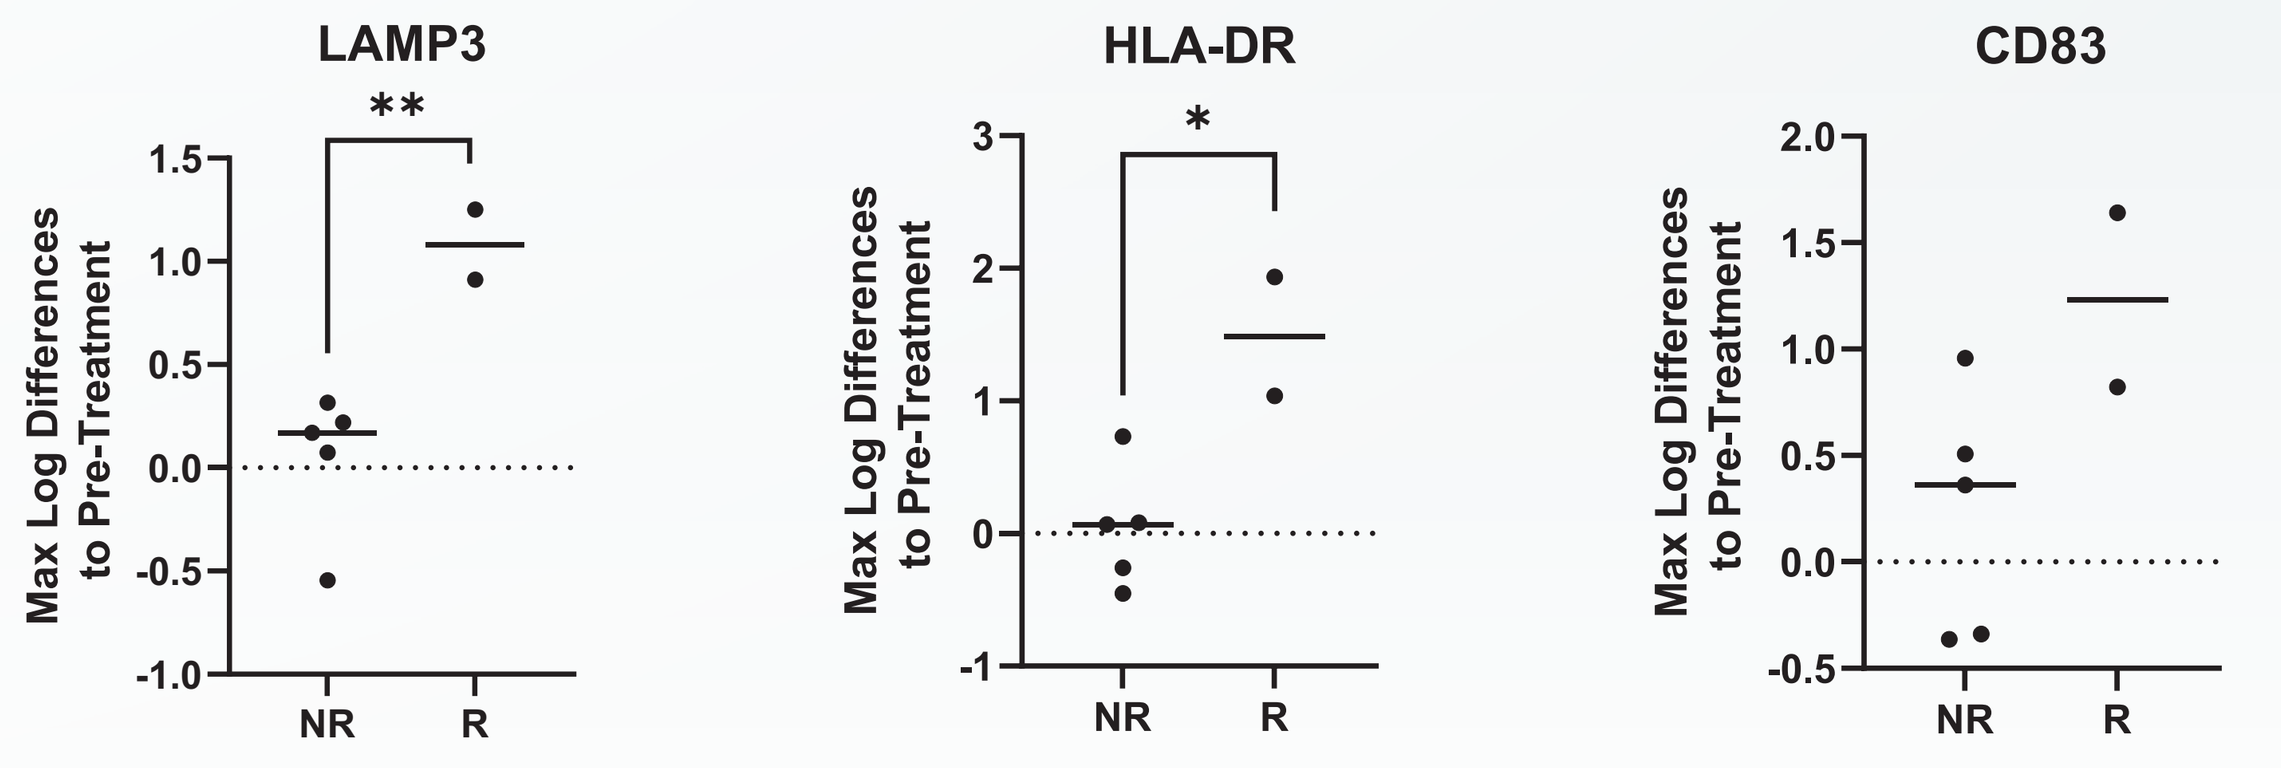
LAMP3
**
HLA-DR
*
CD83
Max Log Differences to Pre-Treatment
NR
R

Serum of 7 patients from the nivolumab+COM701 dose escalation arm, were analyzed using Olink Explore 1536. For each patient, the maximal difference of log2 expression between all on-treatment time points and the pre-treatment value was calculated for each protein. Maximal log2 differences were compared by Student's t-test, with patients grouped based on response, RECIST criteria (responders (R): CR+PR vs. non responders (NR): SD+PD). Out of 10 proteins most significant for on-treatment up-regulation in responders, 3 are markers of activated DCs (LAMP3 ,HLA-DR and CD83).

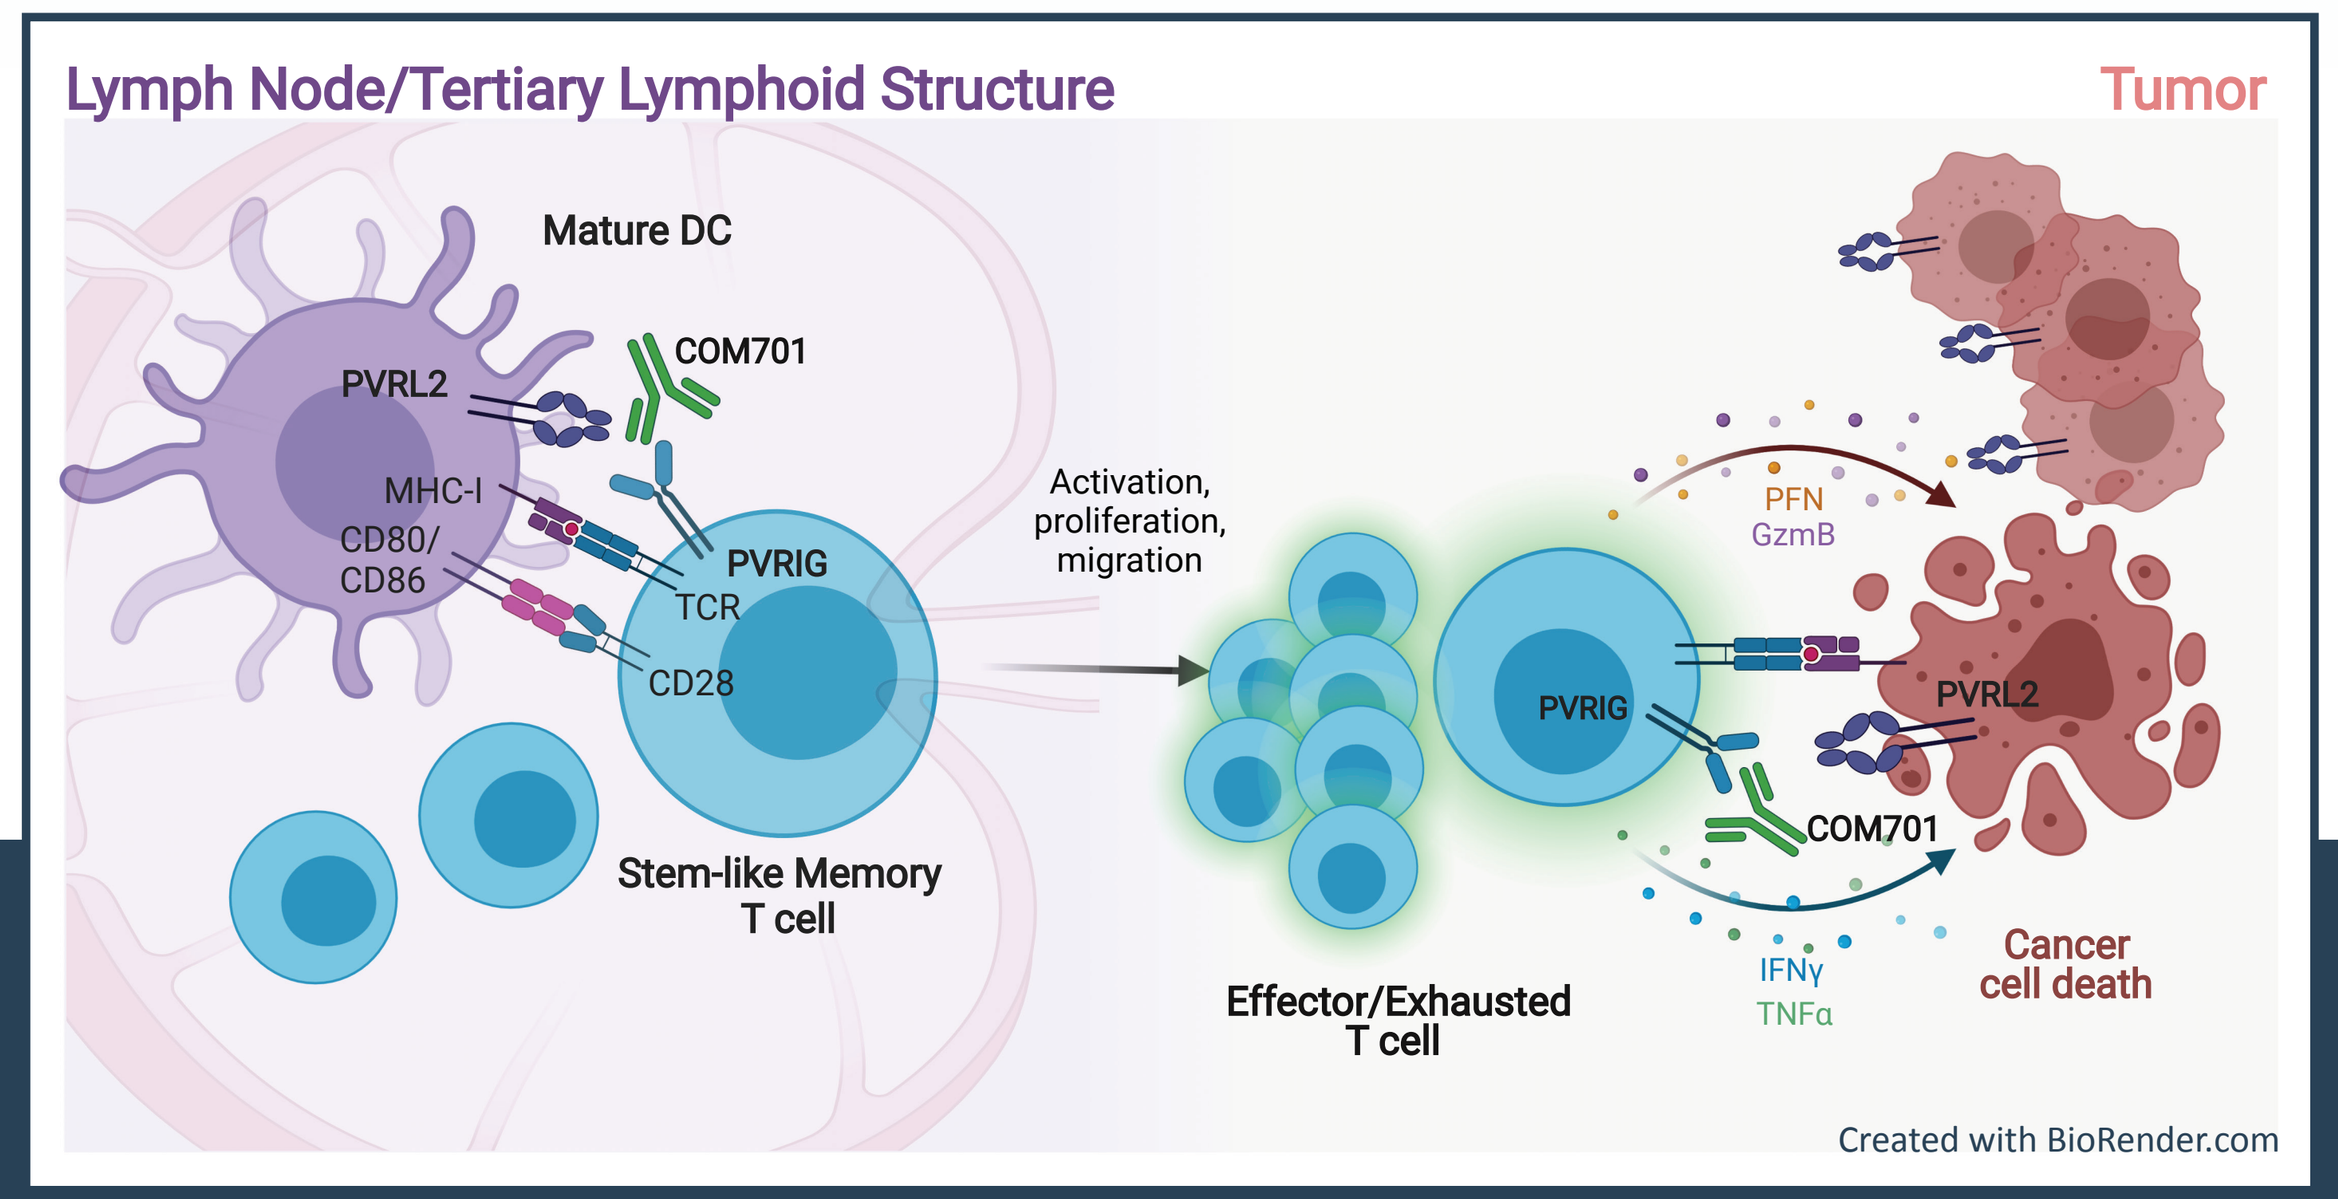
Lymph Node/Tertiary Lymphoid Structure
Tumor
Mature DC
PVRL2
COM701
MHC-I
CD80/
CD86
PVRIG
TCR
CD28
Stem-like Memory T cell
Activation, proliferation, migration
Effector/Exhausted T cell
PVRIG
PFN
GzmB
PVRL2
COM701
IFNγ
TNFα
Cancer cell death
Created with BioRender.com

CONCLUSION

PVRIG is expressed on stem-like and exhausted T cells but has a unique dominant expression on early memory cells, while PVRL2 is abundantly expressed across DC types. PVRIG blockade could therefore enhance memory T cells activation by DCs, resulting in their increased expansion and differentiation. Accordingly, early data shows increased induction of activated DC markers, potentially following efficient T-DC interaction, in serum of two patients responding to COM701+nivolumab.