Harnessing natural IL-18 activity through IL-18BP blockade reshapes the tumor microenvironment for potent anti-tumor immune response

Assaf Menachem, Zoya Alteber, Gady Cojocaru, Tal Fridman Kfir, Dan Blat, Olga Leiderman, Moran Galperin, Lital Sever, Nadav Cohen, Keren Cohen, Roy Granit, Sandra Vols, Masha Frenkel, Lior Faigenbloom, Liron Soffer, Karin Meyer, Keren Menachem, Hadas Galon Tilleman, Michal Perpinial, Evgeny Tatirovsky, Pierre Ferre, Eran Ophir

Compugen Ltd, Holon, Israel

compugen FROM CODE TO CURE®

Background

Conventional cytokines have limited anti-cancer efficacy mostly due to narrow therapeutic window and systemic adverse effects. IL-18 is an inflammasome induced proinflammatory cytokine that activates T and NK cells and stimulates IFNγ production. The activity of IL-18 is naturally blocked by a high affinity endogenous binding protein (IL-18BP), which is induced in response to IFNγ upregulation as a negative feedback mechanism. By assessing total and free IL-18 we examined whether bound-IL-18 levels in the tumor are above the level required for in-vitro human T-cell activation. To unleash endogenous bound IL-18 activity, COM503, an anti-human IL-18BP blocker Ab, was generated and examined in T and NK cell-based assays. In-vivo, IL-18BP blockade was evaluated in multiple mouse tumor models. Tumor microenvironment (TME) modulation was assessed by flow cytometry, scRNA sequencing and cytokine profiling.

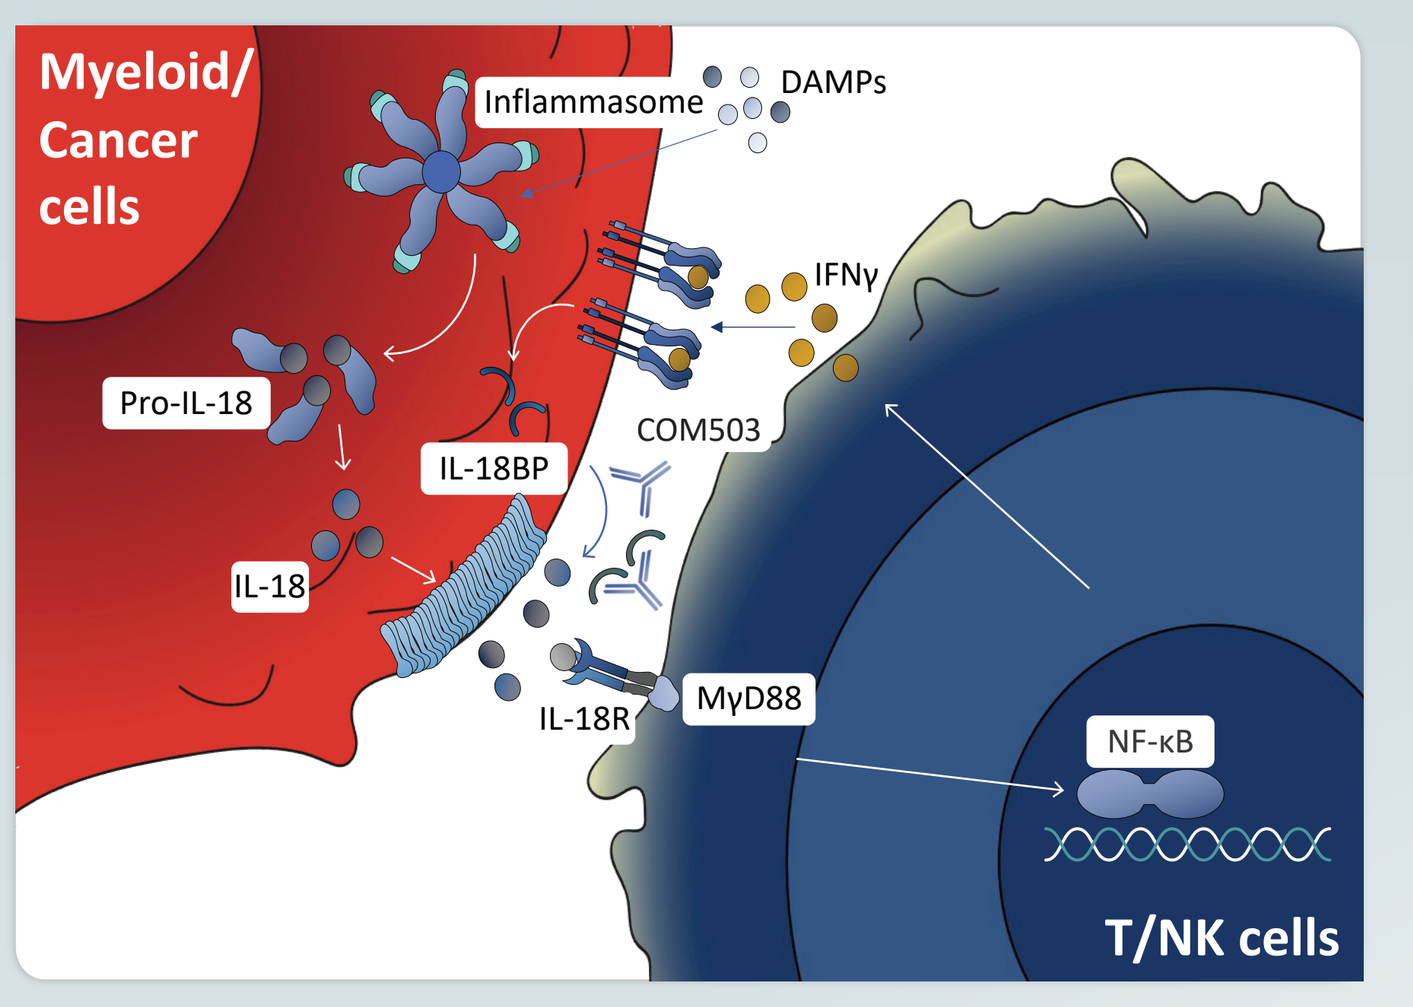
Myeloid/ Cancer cells
Inflammasome
DAMPs
IFNγ
Pro-IL-18
IL-18BP
COM503
IL-18
IL-18R
MyD88
NF-κB
T/NK cells

IL-18 is:
- An effector cytokine
- Secreted upon inflammasome activation
- Upregulated in the TME

IL-18BP:
- Binds IL-18 and blocks its immune stimulatory activity
- IL-18BP secretion is increased via an IL-18-mediated feedback mechanism

COM503:
- Has the potential to induce potent anti-tumor responses and pronounced TME-localized immune modulation

Inflammasome induced cytokines such as IL-18 and IL-1β are abundant in the TME

A.

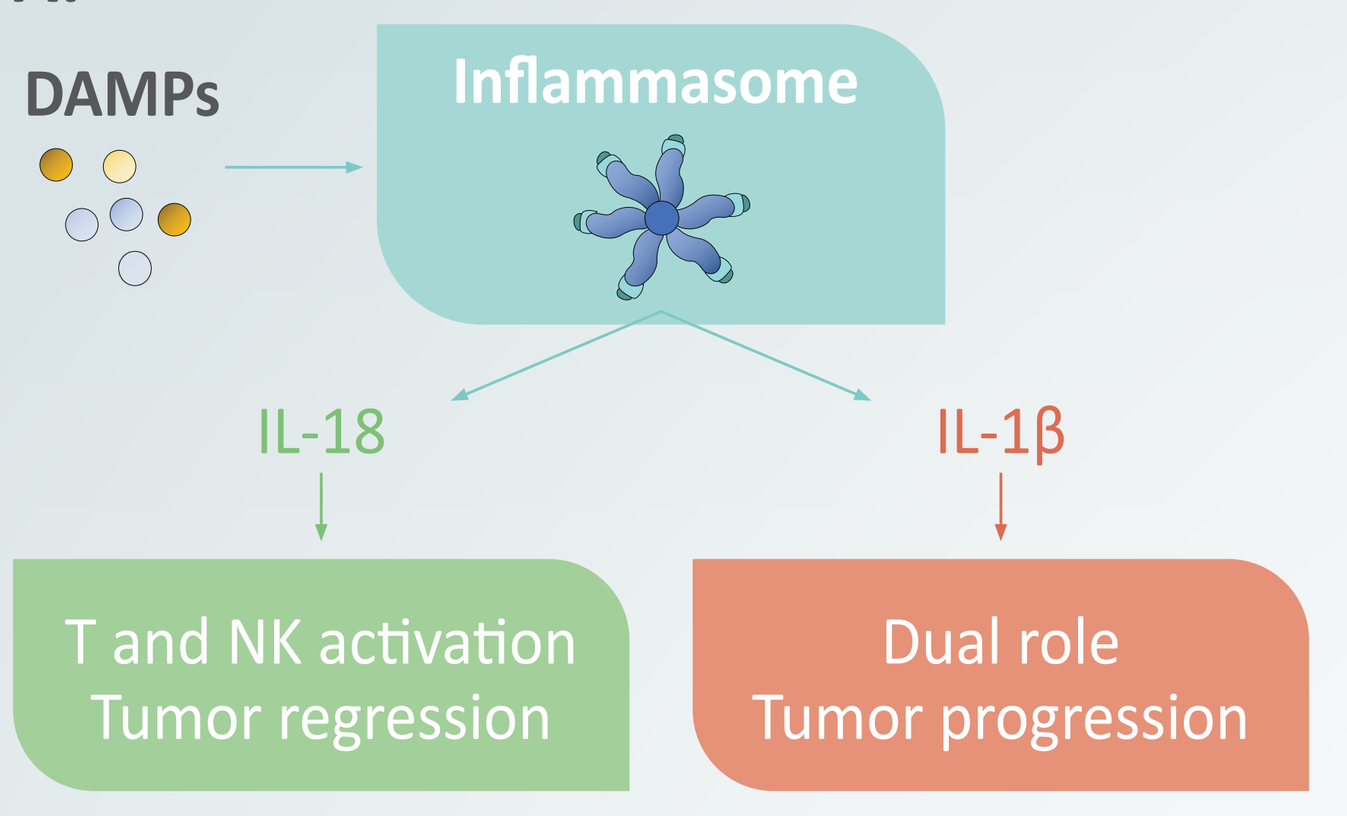
DAMPs
Inflammasome
IL-18
IL-1β
T and NK activation
Tumor regression
Dual role
Tumor progression

B.

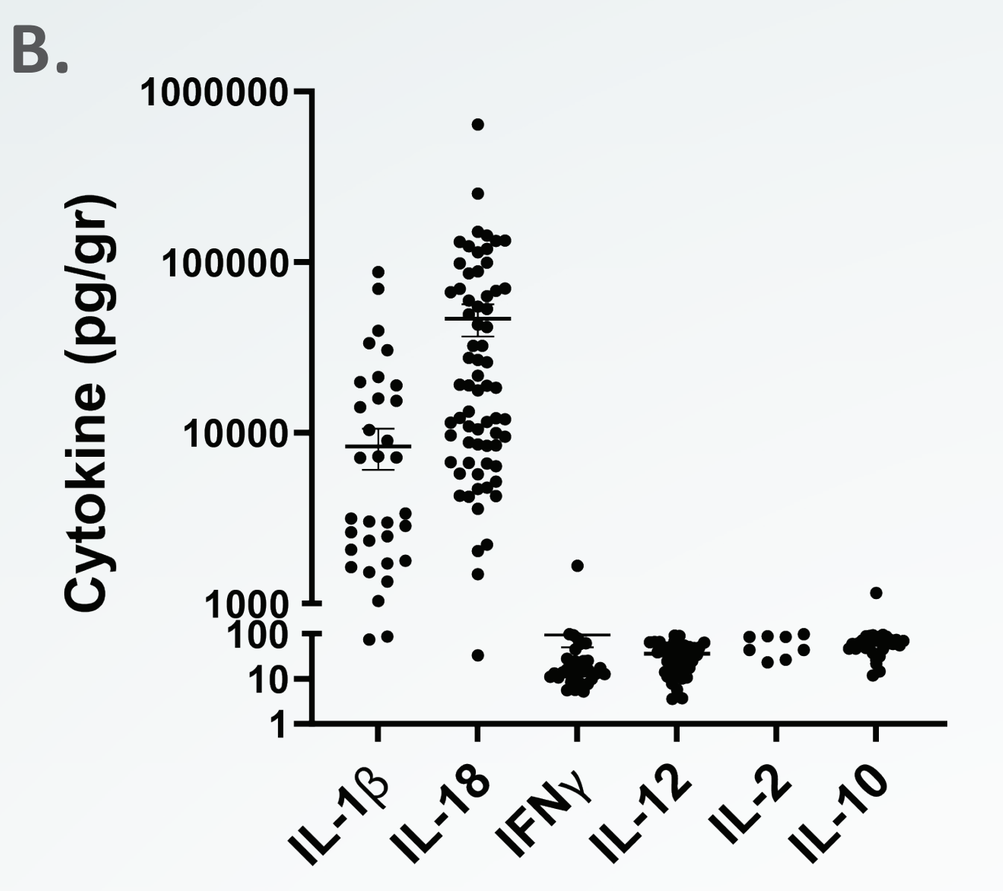
Cytokine (pg/gr)
IL-1β
IL-18
IFNγ
IL-12
IL-2
IL-10

List of indications:
Colorectal
NSCLC
Ovary
Renal
Breast
Endometrial
H&N
Stomach
Pancreas
Esophageal
Bladder
Rectal

A. IL-18 and IL-1β are inflammasome derived cytokines with opposite effects in the TME. **B.** Levels of cytokines in tumor derived supernatants (TDS) across indications.

IL-18 pathway is elevated in the TME across indications

A.

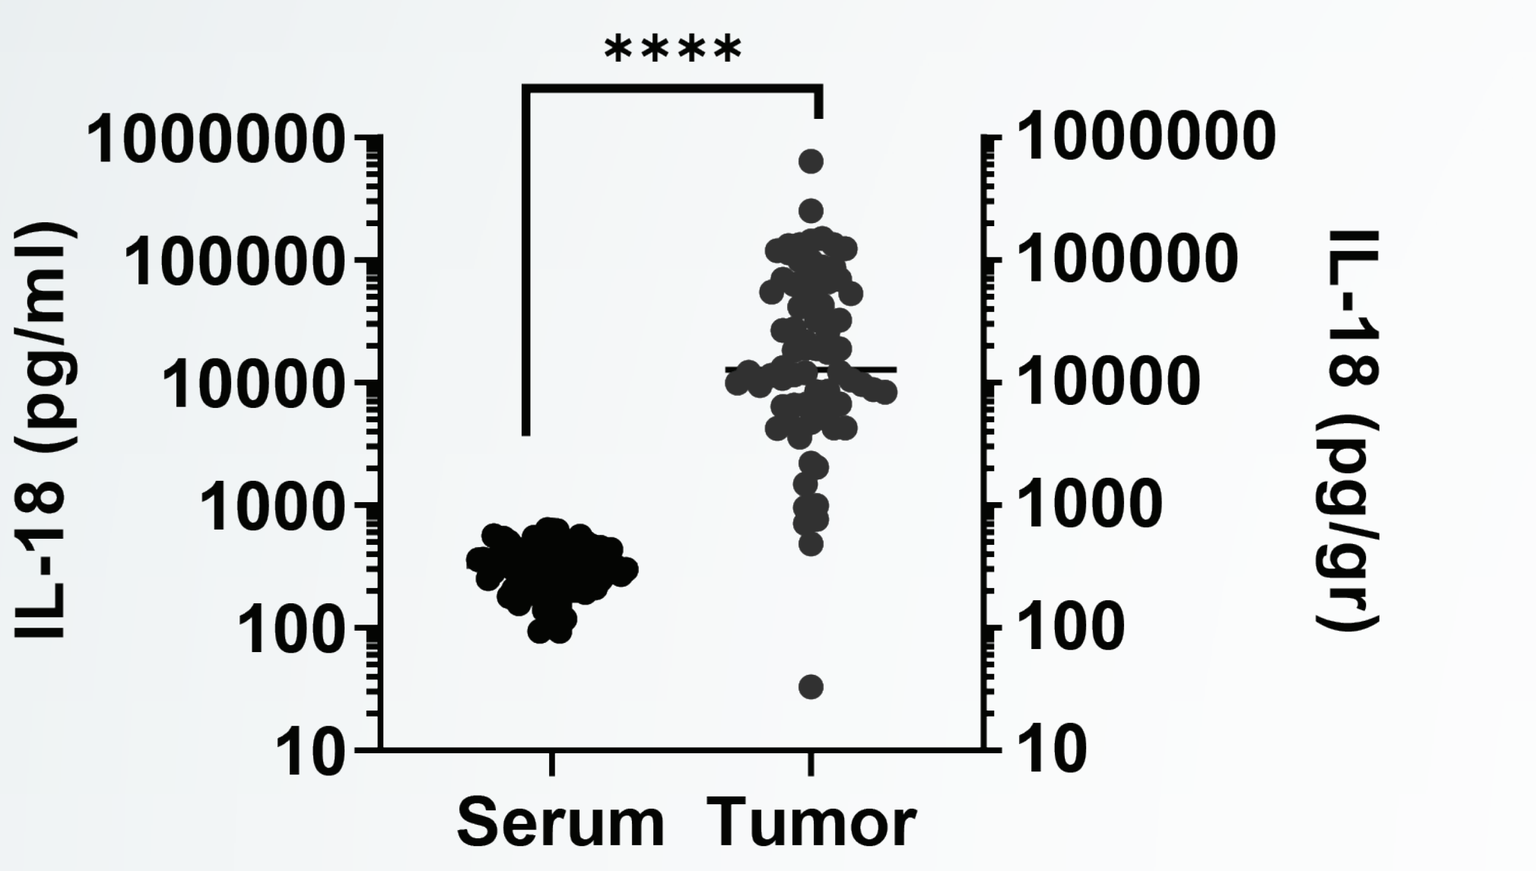

IL-18 (pg/ml)
IL-18 (pg/gr)
Serum
Tumor

C.

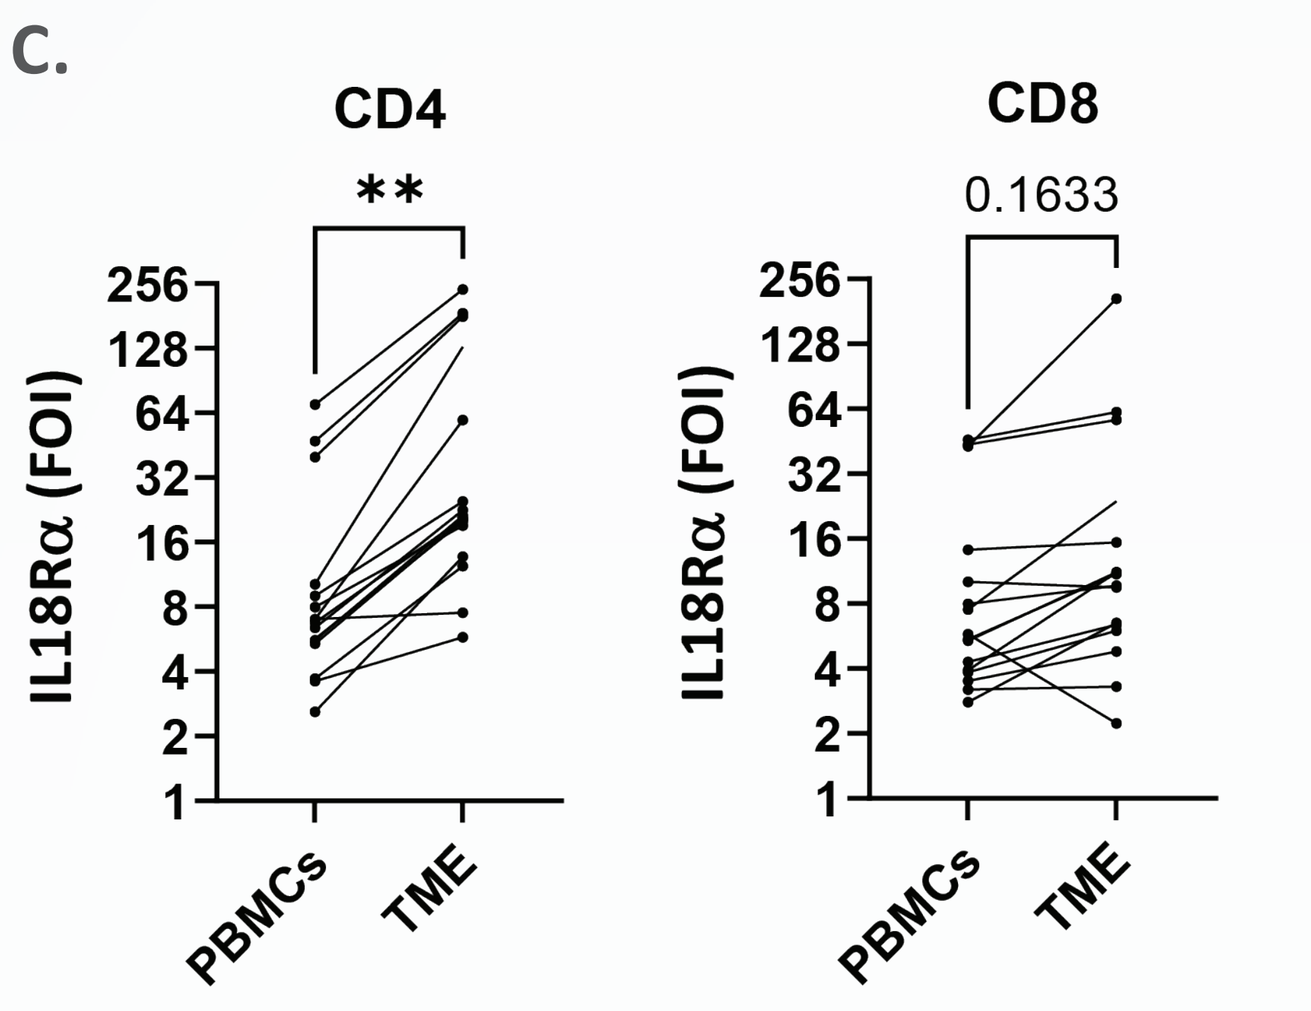
CD4
**
CD8
0.1633
IL18Rα (FOI)
PBMCs
TME

B.

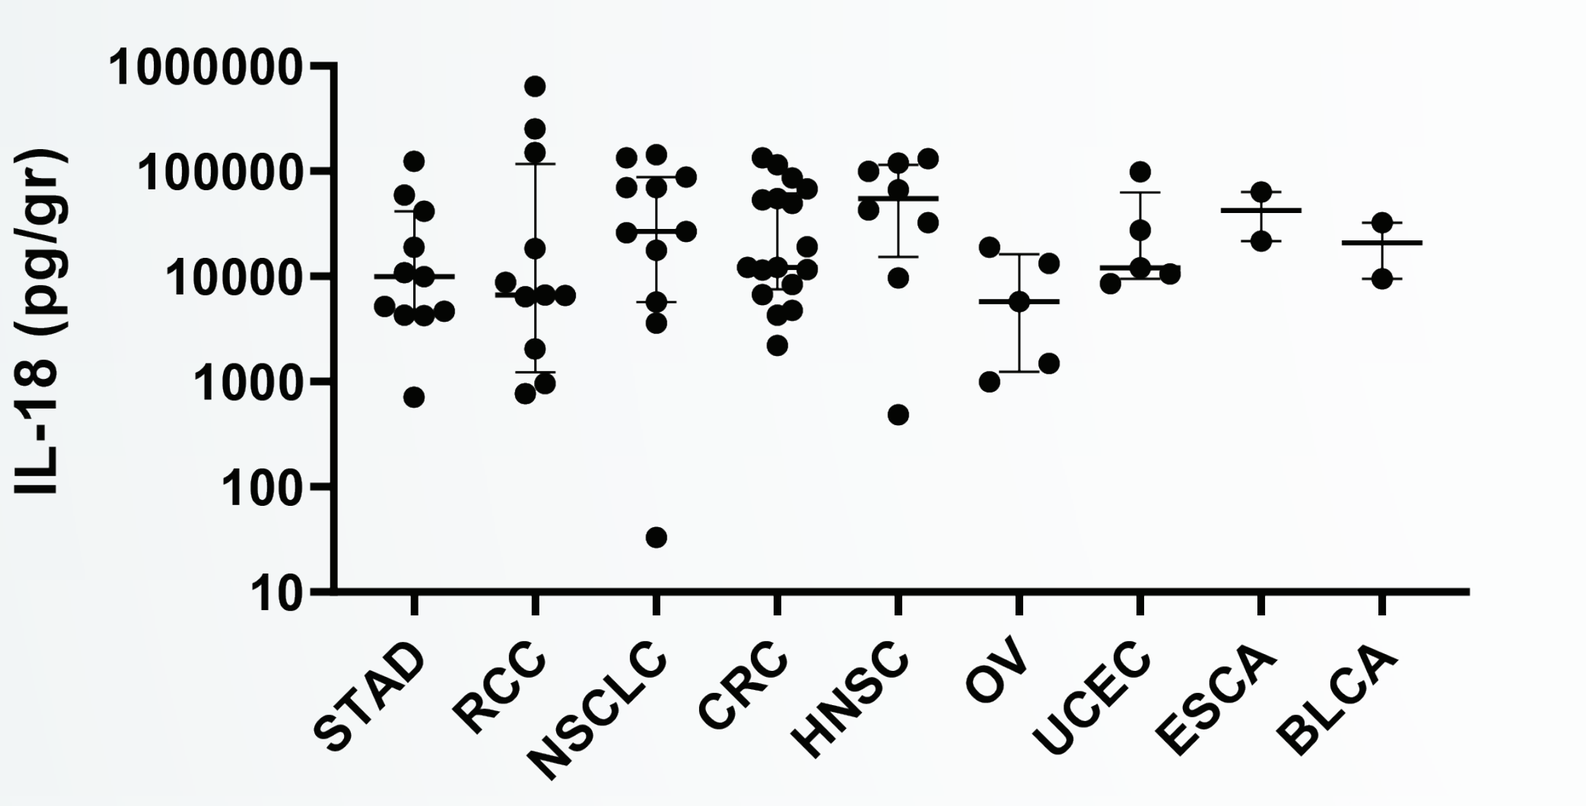
IL-18 (pg/gr)
STAD
RCC
NSCLC
CRC
HNSC
OV
UCEC
ESCA
BLCA

A. IL-18 levels in TDS are elevated compared to levels in serum in cancer patients. **B.** IL-18 levels in TDS across indications. **C.** IL-18 receptor α subunit (IL-18Rα) expression is induced on tumor infiltrating lymphocytes (TILs) in the TME compared to periphery. Statistical analysis was preformed using paired t test (two tailed). P ≤ 0.01**, P ≤ 0.0005****

IL-18BP-bound IL-18 levels in the TME are above the amount required for T cell activation in vitro

A.

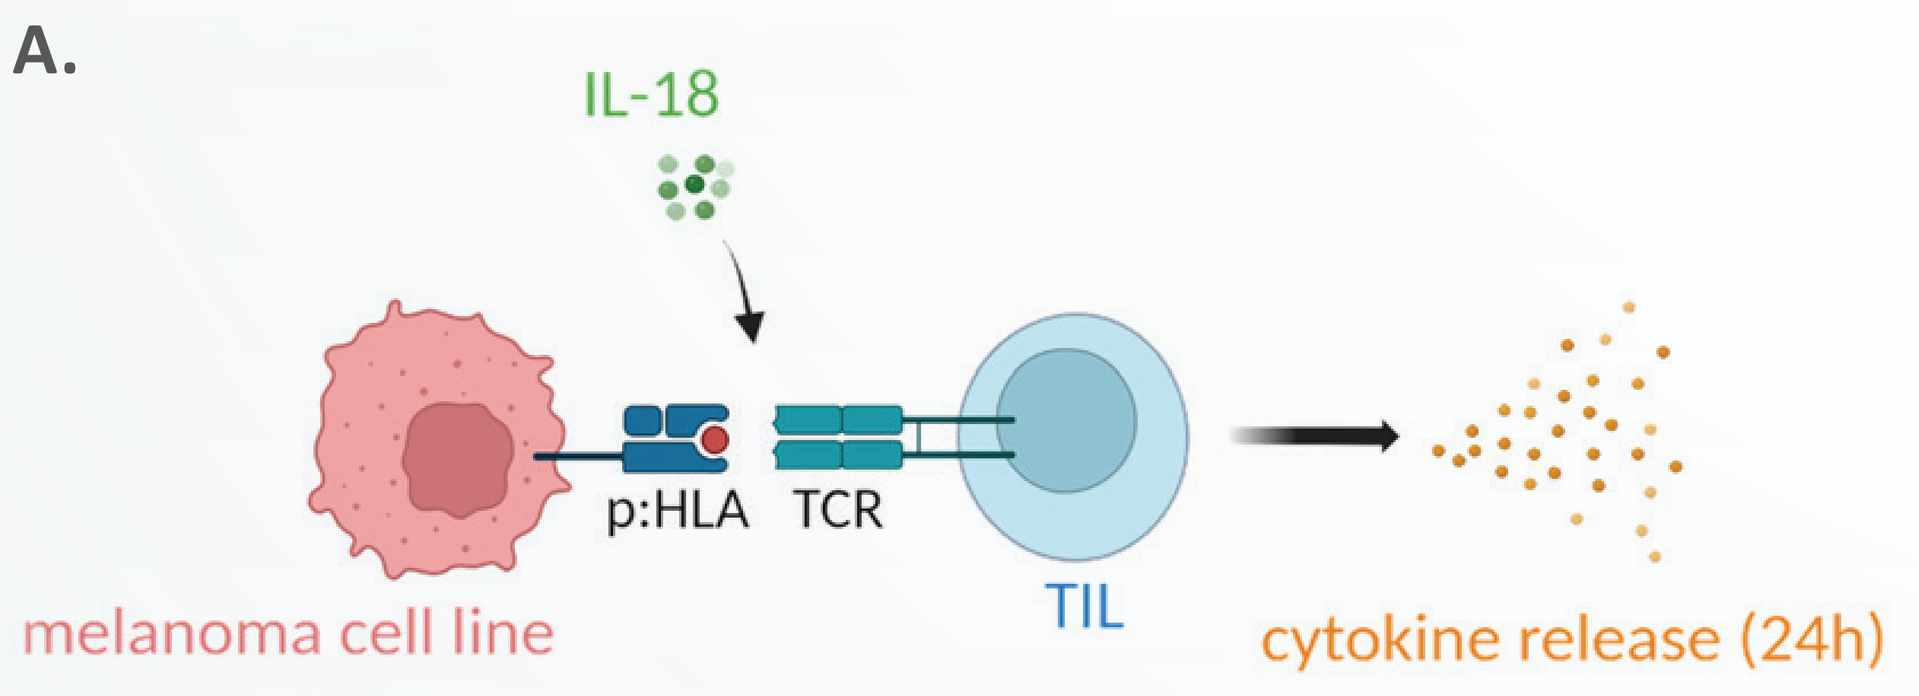
IL-18
p:HLA
TCR
melanoma cell line
TIL
cytokine release (24h)

B.

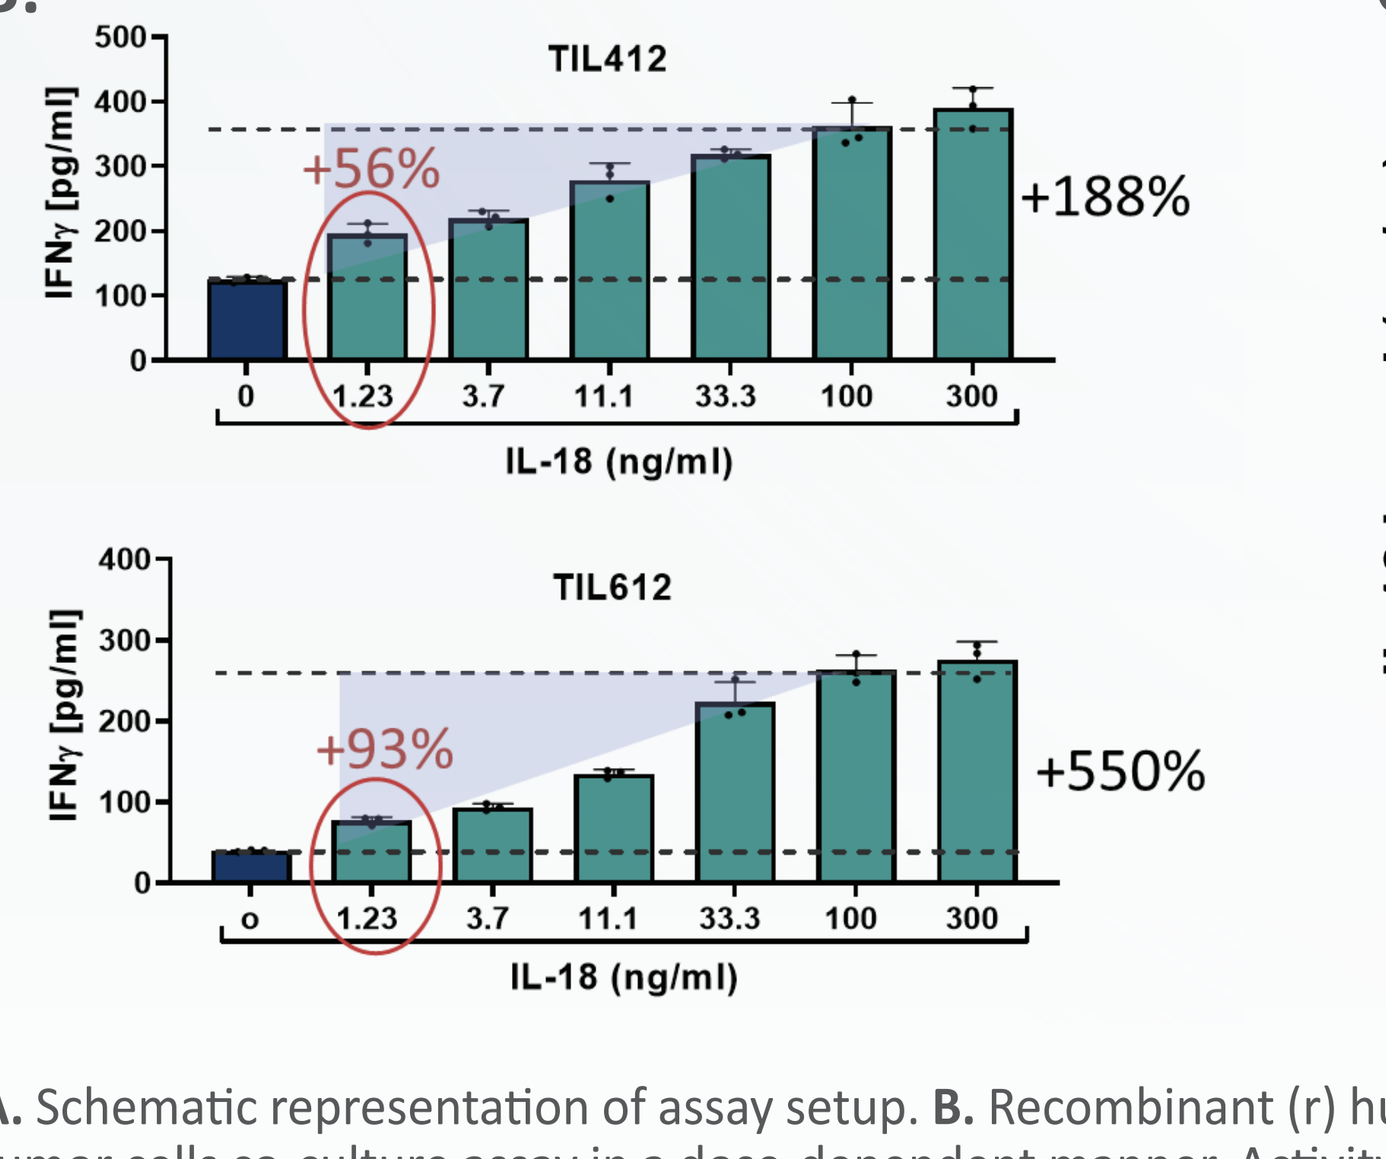
TIL412
IFNγ [pg/ml]
+56%
+188%
IL-18 (ng/ml)
TIL612
+93%
+550%

C.

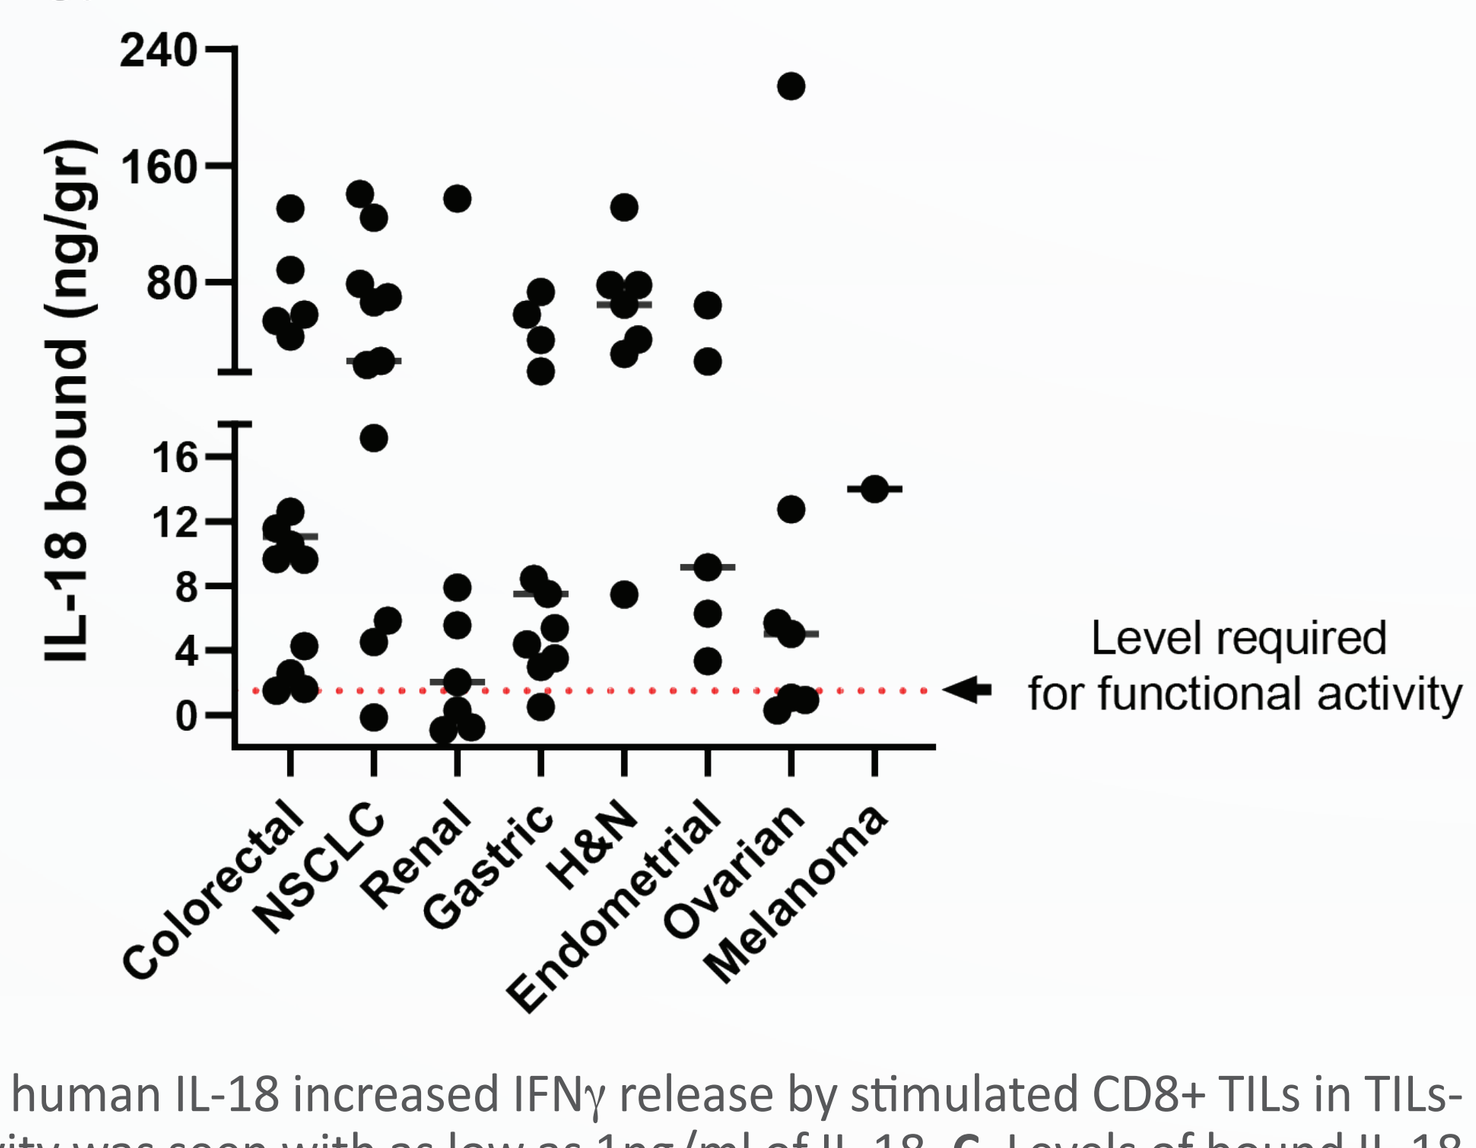
IL-18 bound (ng/gr)
Level required for functional activity
Colorectal
NSCLC
Renal
Gastric
H&N
Endometrial
Ovarian
Melanoma

A. Schematic representation of assay setup. **B.** Recombinant (r) human IL-18 increased IFNγ release by stimulated CD8+ TILs in TILs-tumor cells co-culture assay in a dose-dependent manner. Activity was seen with as low as 1ng/ml of IL-18. **C.** Levels of bound IL-18 in TDS across indications are mostly above the level required for in vitro T cell activation. IL-18BP-bound IL-18 levels were calculated by deducting IL-18 free from total IL-18 measured for each sample by two separate ELISA assays. Dashed red line represent the level required for functional activity (1.5ng/gr).

IL-18BP binds IL-18 with high affinity and inhibits its activity in HEK293 reporter system

A.

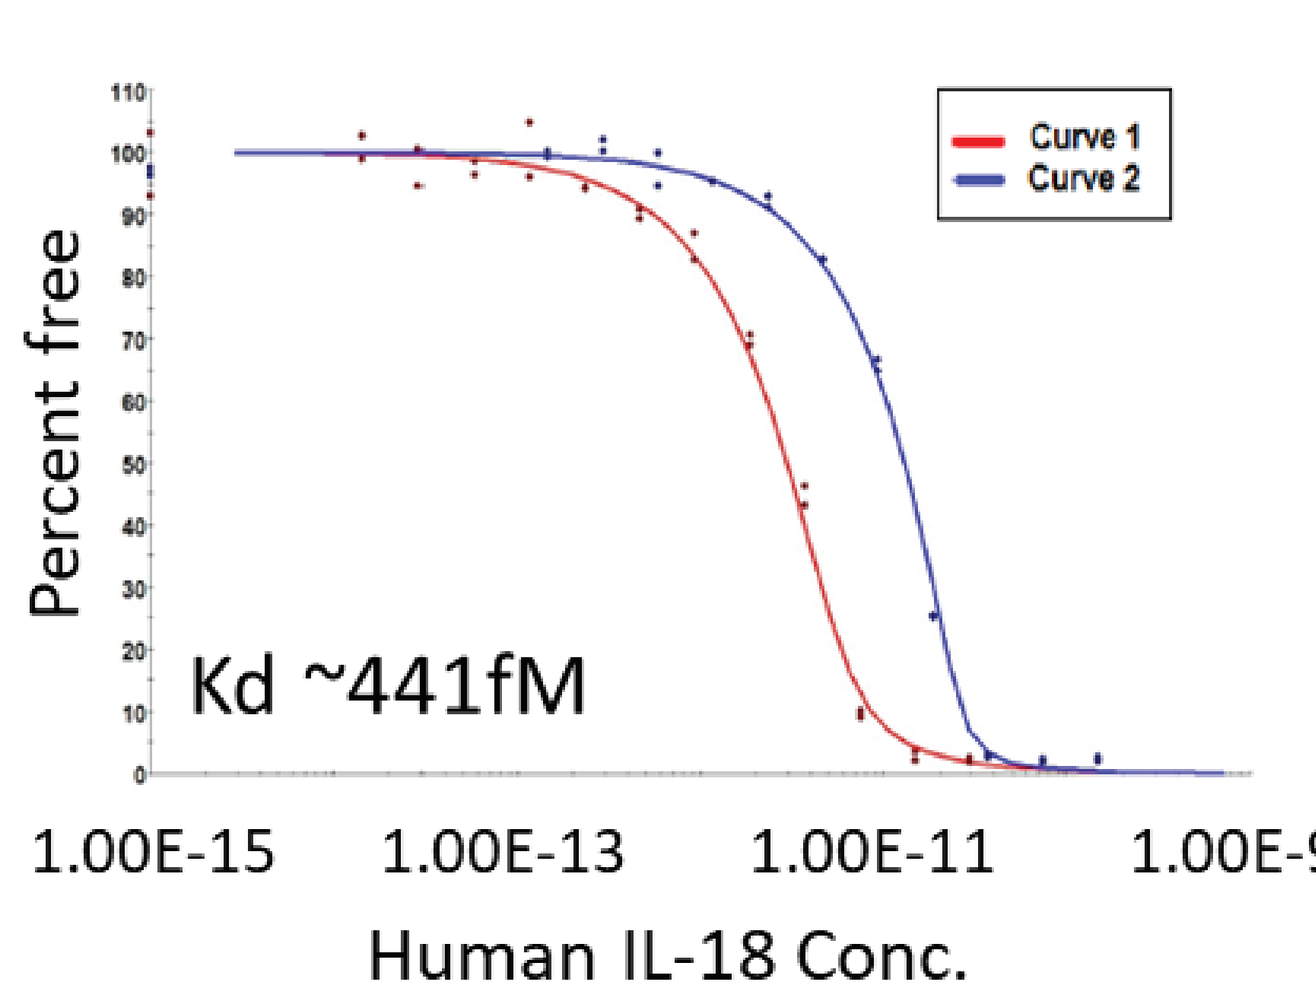
Percent free
Curve 1
Curve 2
Kd ~441fM
1.00E-15
1.00E-13
1.00E-11
1.00E-9
Human IL-18 Conc.

B.

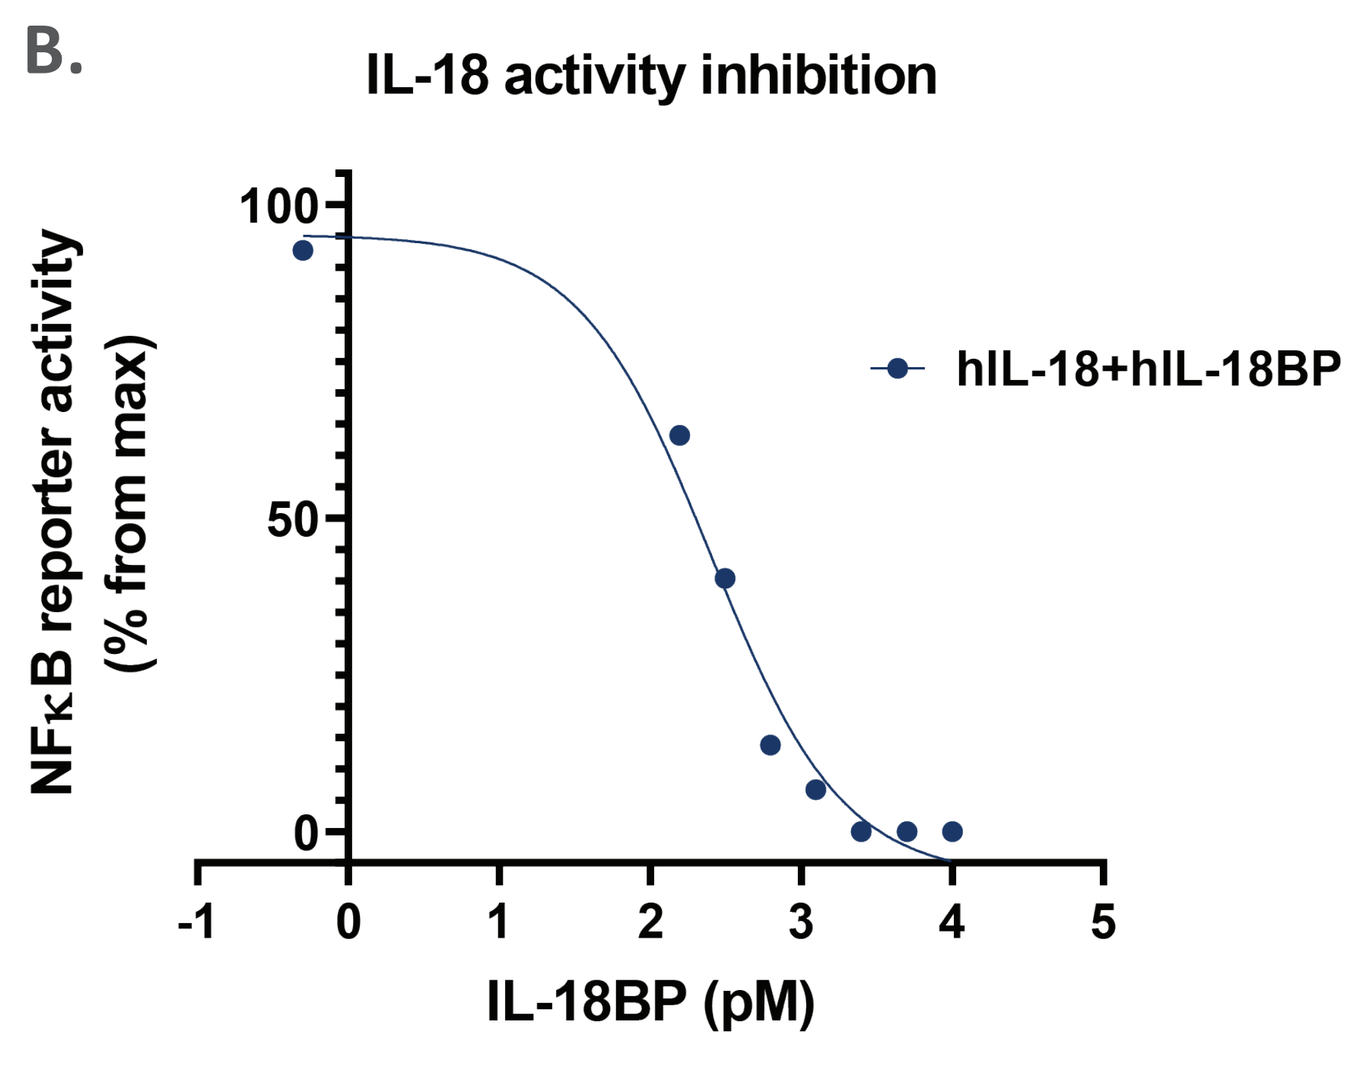
IL-18 activity inhibition
NFκB reporter activity (% from max)
hIL-18+hIL-18BP
IL-18BP (pM)

A. Human IL-18-IL-18BP interaction measurements in KinExA. For each run in KinExA, two curves with different column binding protein (CBP) concentrations were run and analyzed using n-curve analysis to determine the Kd. **B.** IL-18BP inhibits IL-18 activity in NFkB-HEK293-IL-18R reporter system

IL-18BP is upregulated in tumor associated macrophages in the TME and expressed across multiple human cancer indications

A.

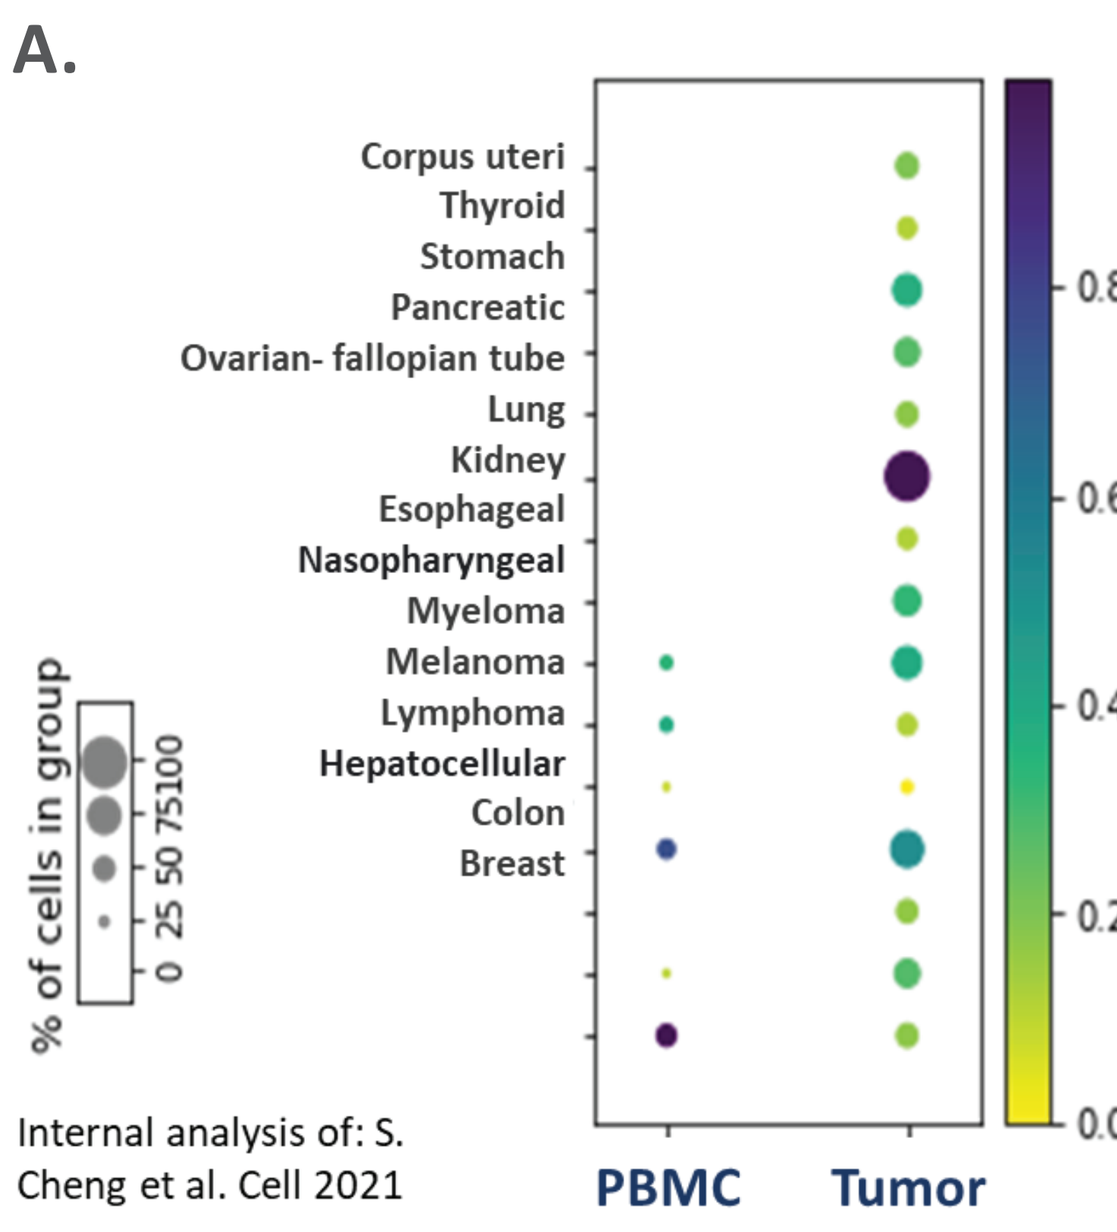
Corpus uteri
Thyroid
Stomach
Pancreatic
Ovarian- fallopian tube
Lung
Kidney
Esophageal
Nasopharyngeal
Myeloma
Melanoma
Lymphoma
Hepatocellular
Colon
Breast
% of cells in group
PBMC
Tumor

Internal analysis of: S. Cheng et al. Cell 2021

B.

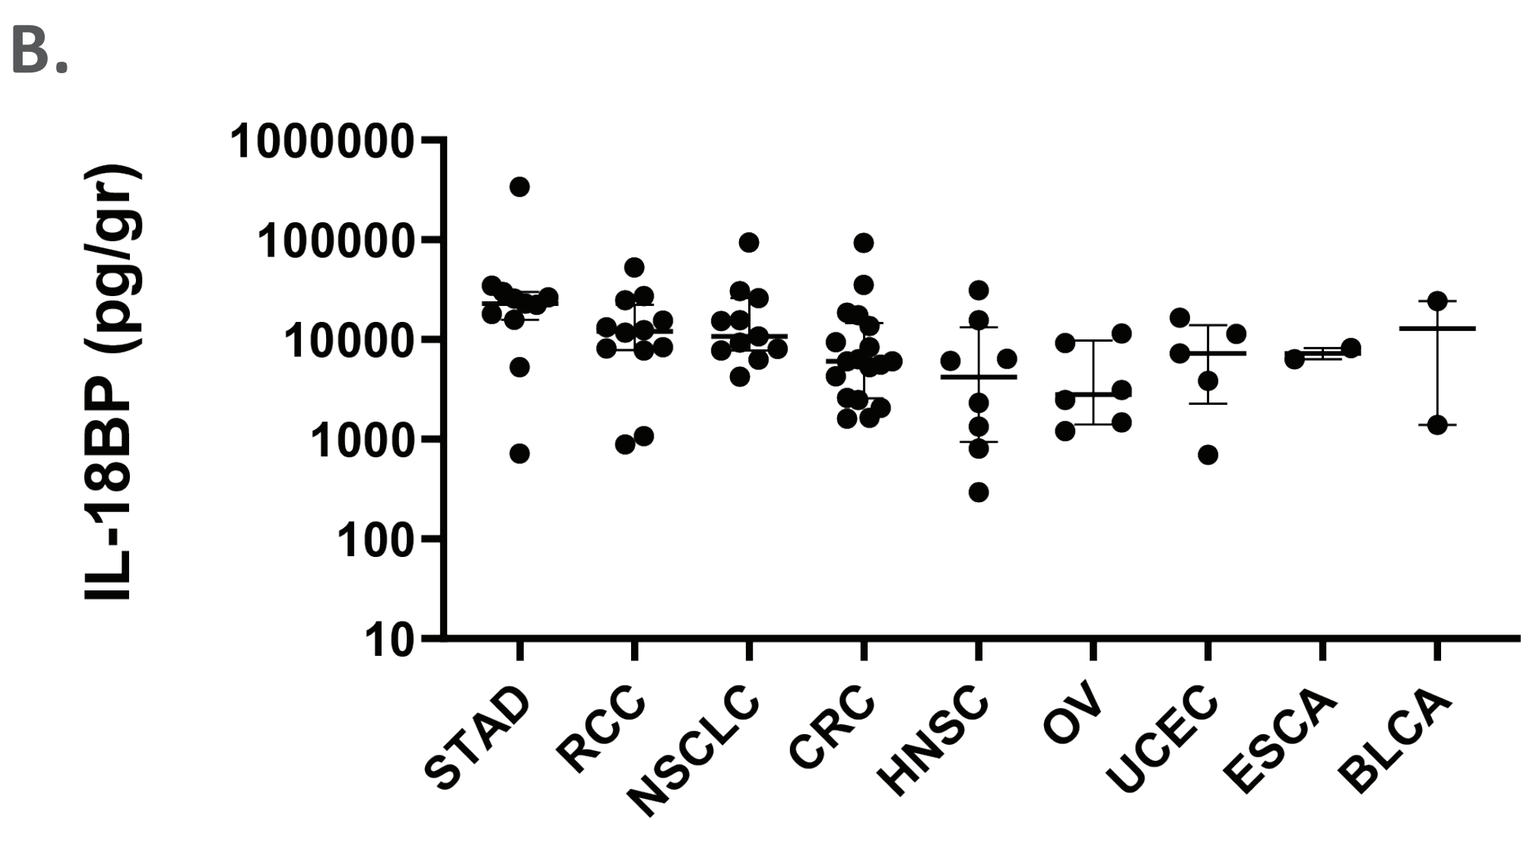
IL-18BP (pg/gr)
STAD
RCC
NSCLC
CRC
HNSC
OV
UCEC
ESCA
BLCA

A. scRNA analyses of tumor-infiltrating myeloid cells, across indications showing that IL-18BP is up-regulated in myeloid population in the TME compared to the periphery (PBMCs). **B.** IL-18BP protein levels in TDS across indications.

COM503- a fully human high affinity anti-IL-18BP blocker antibody restores human T and NK cell activity

A.

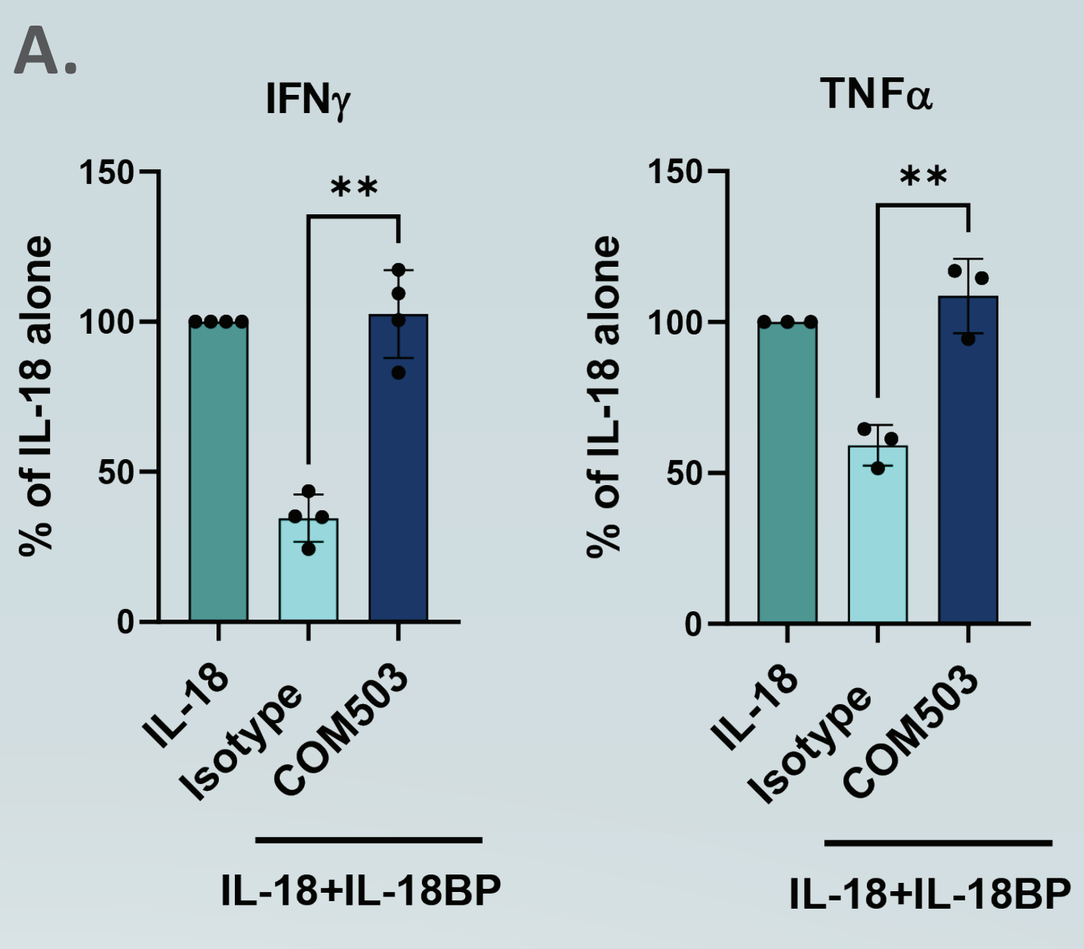
IFNγ
TNFα
% of IL-18 alone
IL-18
Isotype
COM503
IL-18+IL-18BP

B.

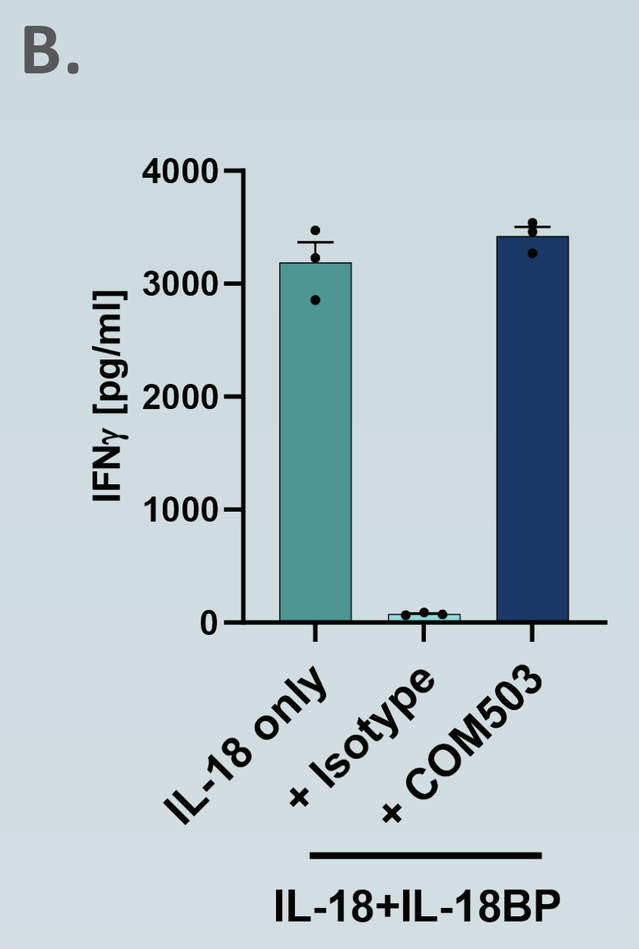
IFNγ [pg/ml]
IL-18 only
+ Isotype
+ COM503
IL-18+IL-18BP

A. COM503 displaced pre-complexed IL-18-IL-18BP to increase IFNγ and TNFα release by stimulated human CD8+ TILs in TILs- tumor cells co-culture assay in the presence of rIL-18BP and rIL-18. **B.** COM503 displaced pre-complexed IL-18-IL-18BP to increase IFNγ release by NK cells in the presence of IL-12. **C.** COM503 increased TNFα, IFNγ, IL-2 and Granzyme B (GZMB) release by human tumor dissociated cells (TDCs). Surgically removed cancer specimens were dissociated into single cell suspension and cultured with anti-CD3/anti-CD28 mAbs in the presence or absence of COM503 (10ug/ml), anti-PD1 (pembrolizumab, 10ug/ml) or both for 72 hours, after which cytokines and granzyme B secretion were measured in supernatants. Representative example from a human ovarian TDC sample is shown.

C.

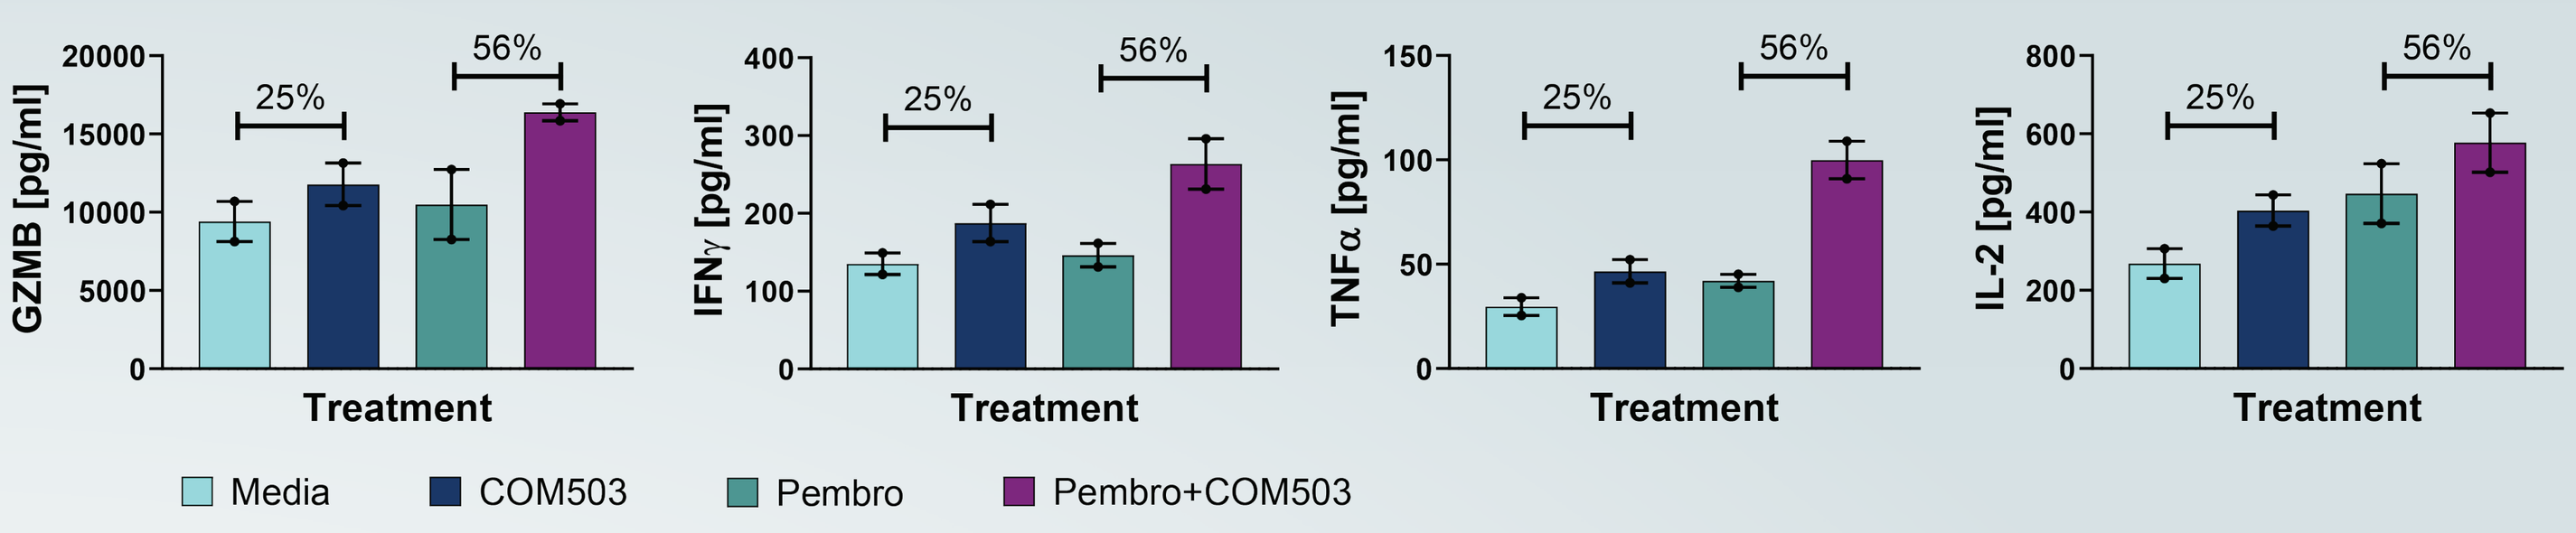
GZMB [pg/ml]
IFNγ [pg/ml]
TNFα [pg/ml]
IL-2 [pg/ml]
25%
56%
Treatment
Media
COM503
Pembro
Pembro+COM503

Anti-mouse IL-18BP antibody demonstrates monotherapy activity across murine syngeneic tumor models

A.

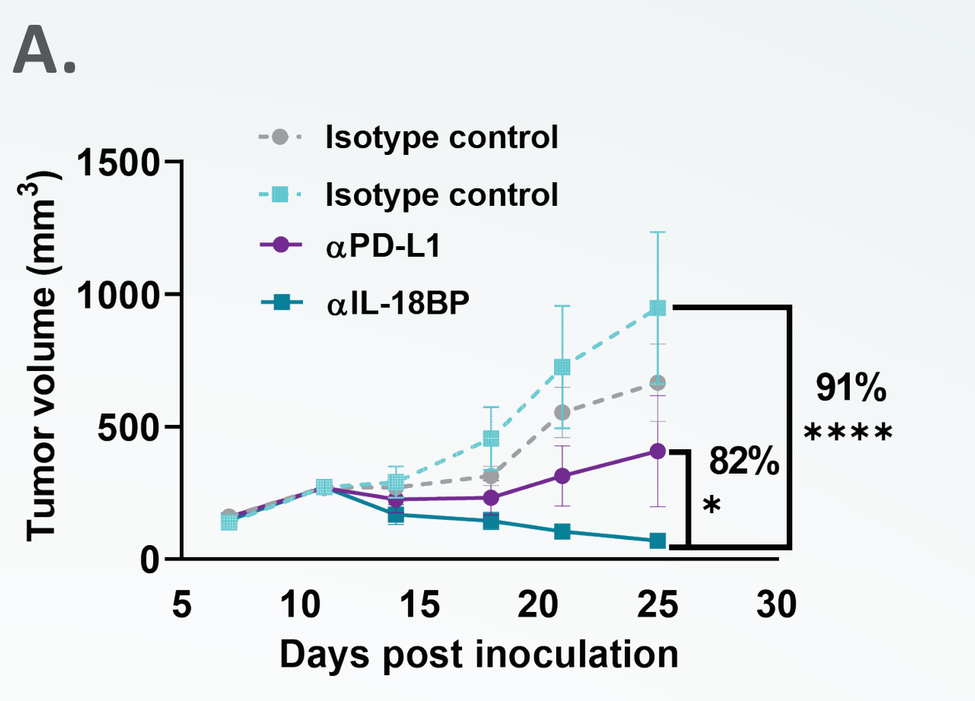
Isotype control
Isotype control
αPD-L1
αIL-18BP
Tumor volume (mm³)
91%

82%
*
Days post inoculation

B.

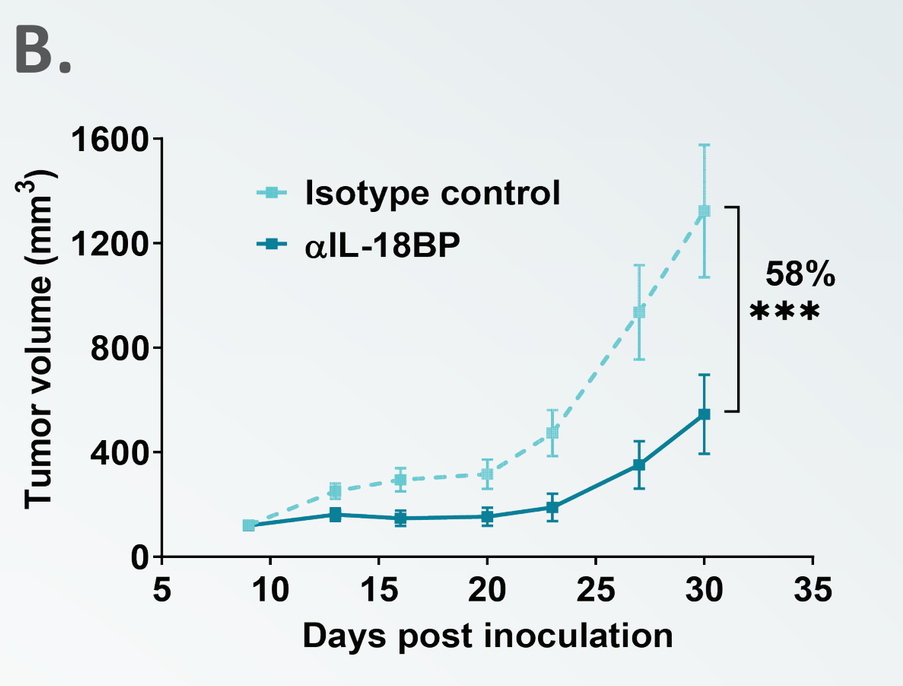
Isotype control
αIL-18BP
Tumor volume (mm³)
58%

Days post inoculation

C.

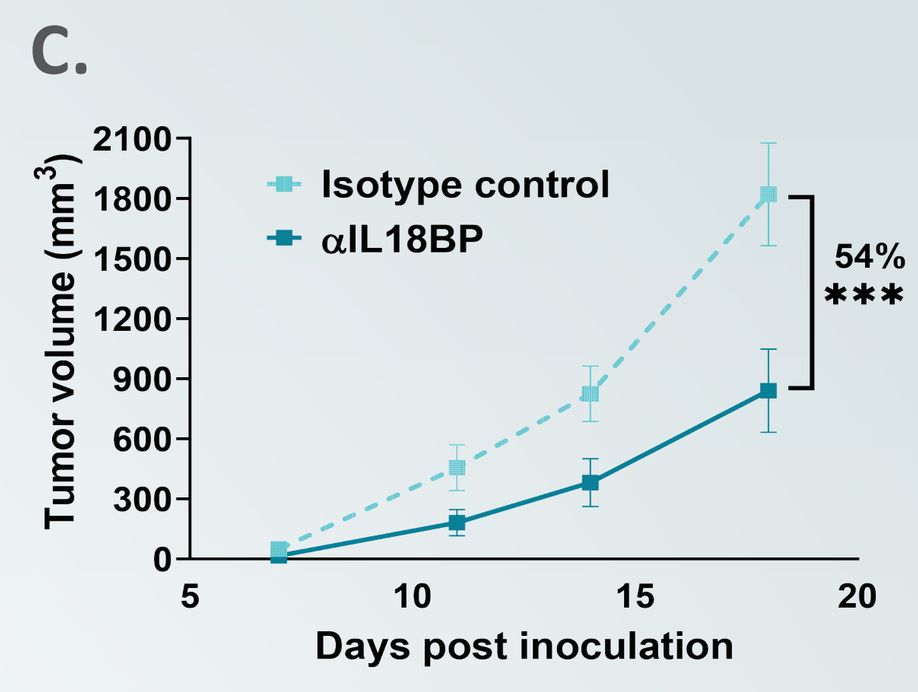
Isotype control
αIL18BP
Tumor volume (mm³)
54%

Days post inoculation

A-C. Anti-mouse IL-18BP Ab inhibited tumor growth in E0771 orthotopic mouse breast tumor model (A) MC38ova mouse CRC tumor model (B) and B16F10-hmgp100 mouse melanoma tumor model (C). Tumor volumes are represented as the Mean volume + SEM. Treatment initiated at tumor volume of 120mm³ and 270mm³ for MC38ova and E0771 respectively, and on day 4 for B16F10-hmgp100, and was injected twice a week for 6 doses.

Anti-mouse IL-18BP antibody demonstrates combo activity with anti-PD-L1 Ab in E0771 tumor model

A.

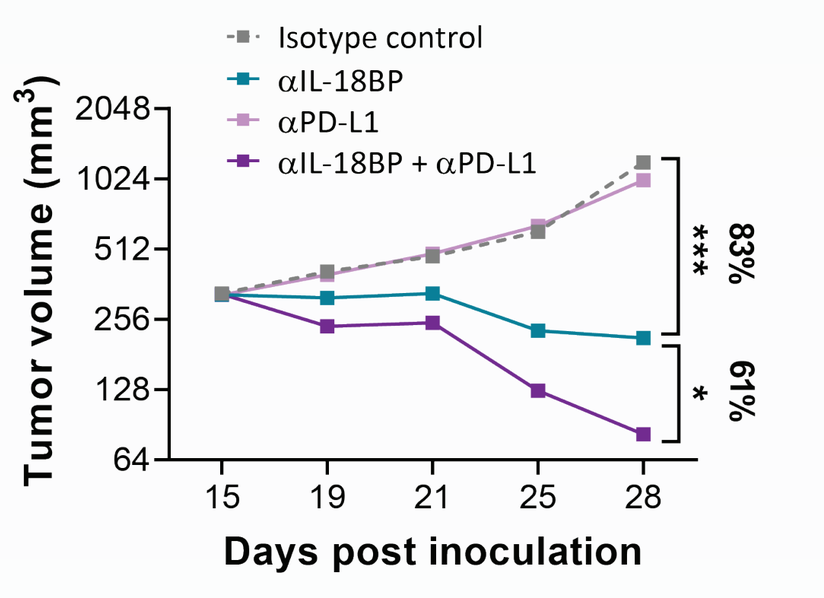
Isotype control
αIL-18BP
αPD-L1
αIL-18BP + αPD-L1
Tumor volume (mm³)
83%

61%
*
Days post inoculation

B.

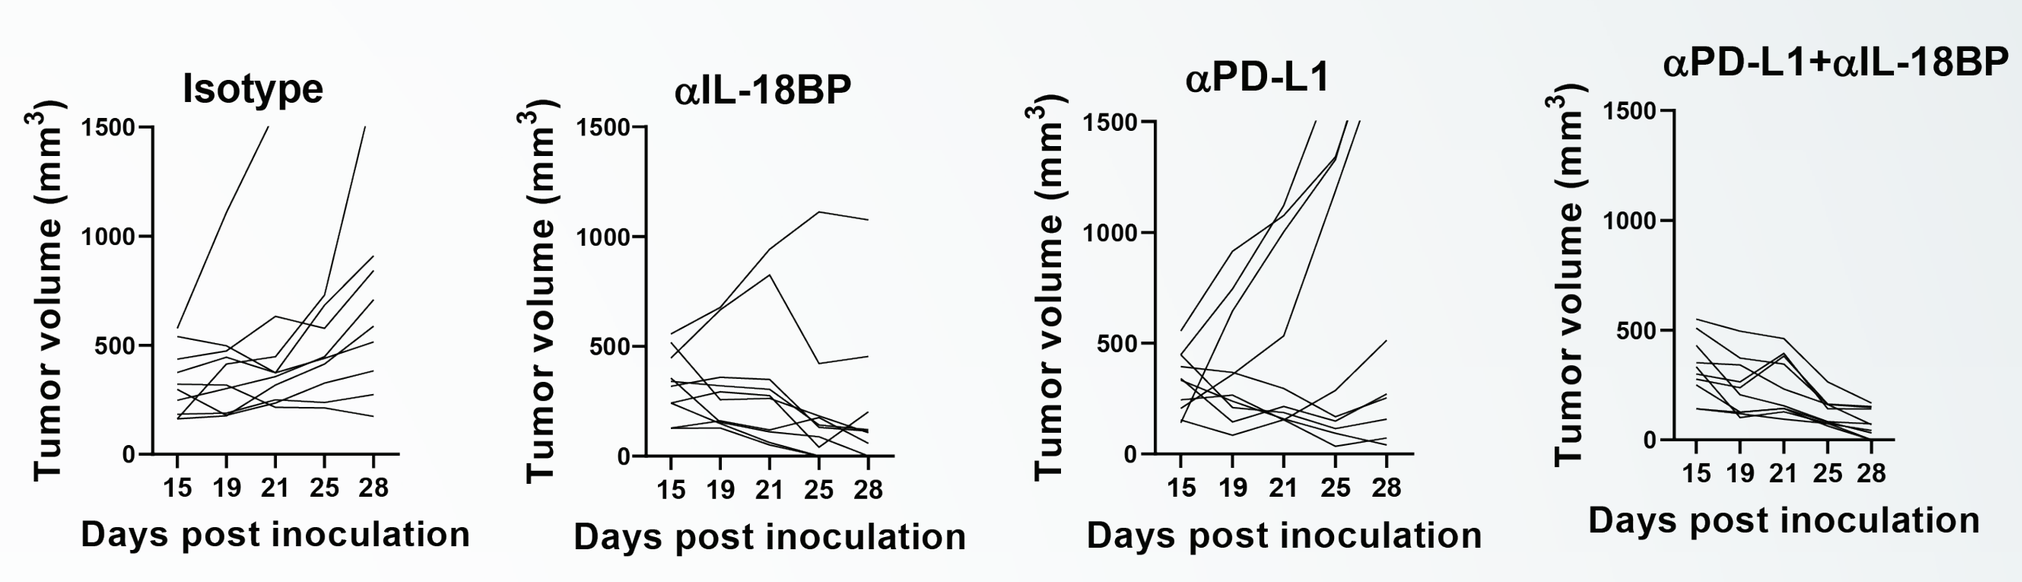
Isotype
αIL-18BP
αPD-L1
αPD-L1+αIL-18BP
Tumor volume (mm³)
Days post inoculation

A. Anti-mouse IL-18BP Ab in combination with anti PD-L1 Ab inhibited tumor growth in E0771 mouse tumor model. **B.** Individual E0771 tumors measurements for each mouse are depicted. Tumor volumes are represented as the Mean volume + SEM. Treatment initiated at tumor volume of 270mm³ and was injected twice a week for 6 doses.

Anti-mouse IL-18BP Ab induces the expansion of polyfunctional non exhausted T cells in the TME

A.

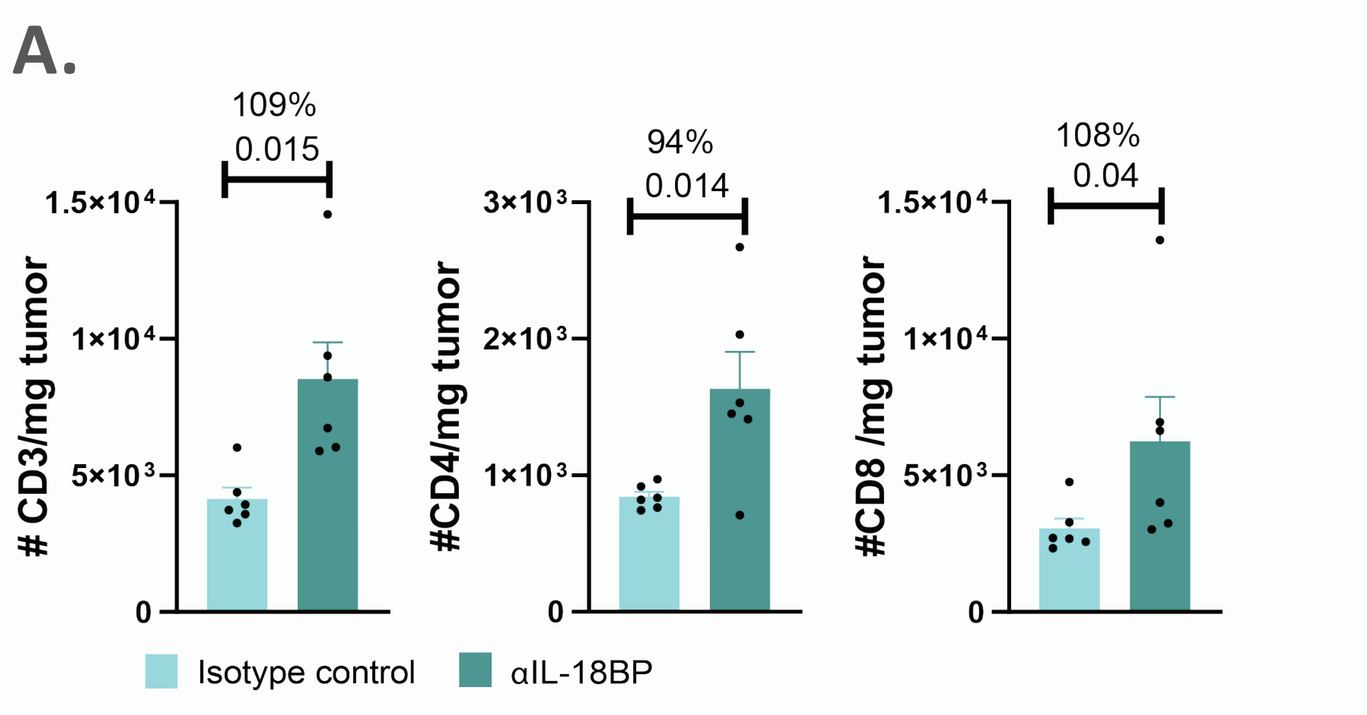
109%
0.015
94%
0.014
108%
0.04
CD3/mg tumor
#CD4/mg tumor
#CD8 /mg tumor
Isotype control
αIL-18BP

B.

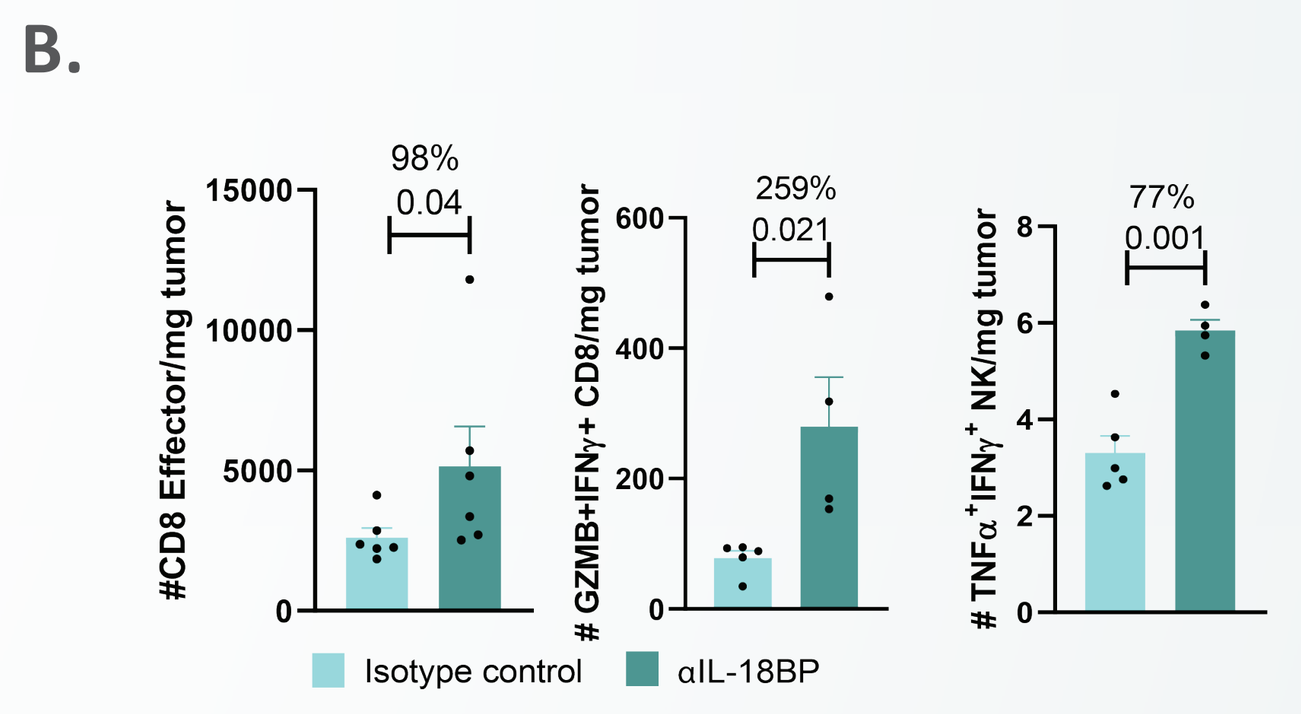
98%
0.04
259%
0.021
77%
0.001
#CD8 Effector/mg tumor
GZMB+IFNγ+ CD8/mg tumor
TNFα+IFNγ+ NK/mg tumor
Isotype control
αIL-18BP

C.

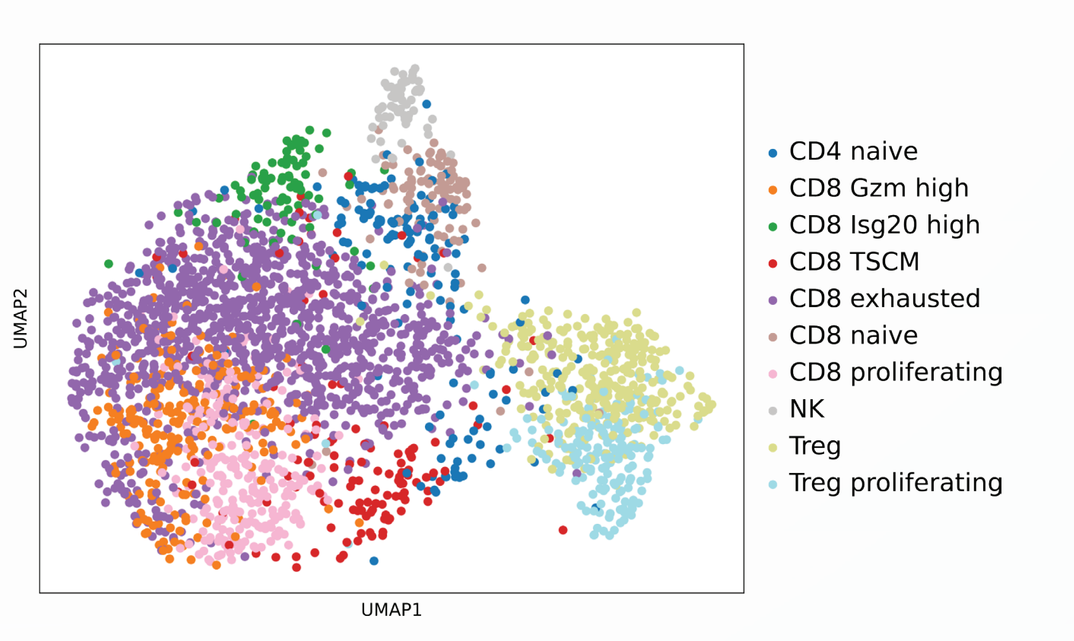
CD4 naive
CD8 Gzm high
CD8 Isg20 high
CD8 TSCM
CD8 exhausted
CD8 naive
CD8 proliferating
NK
Treg
Treg proliferating
UMAP1
UMAP2

D.

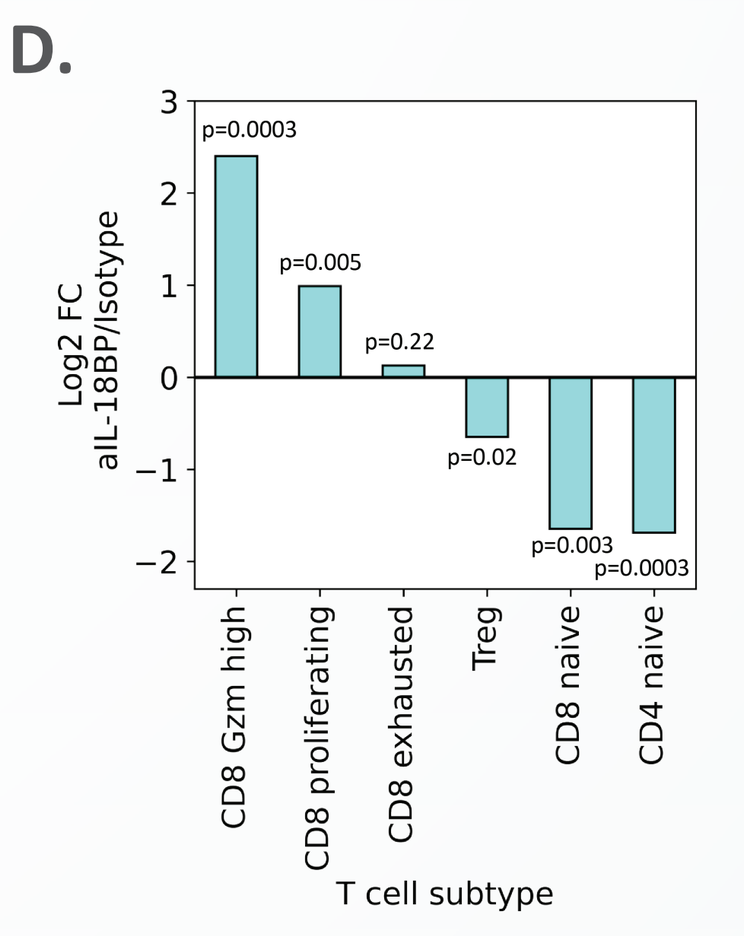
Log2 FC aIL-18BP/Isotype
p=0.0003
p=0.005
p=0.22
p=0.02
p=0.003
p=0.0003
CD8 Gzm high
CD8 proliferating
CD8 exhausted
Treg
CD8 naive
CD4 naive
T cell subtype

E.

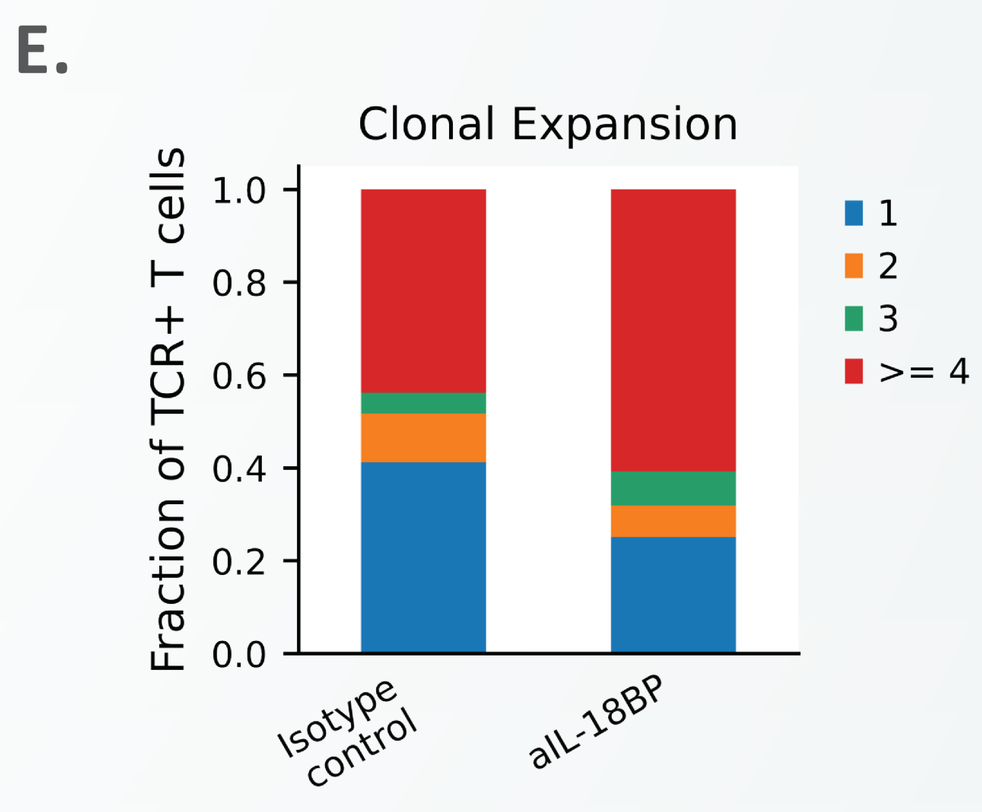
Clonal Expansion
Fraction of TCR+ T cells
1
2
3
>= 4
Isotype control
aIL-18BP

E0771 tumor cells were inoculated in C57Bl/6 mice and treated either with anti-mouse IL-18BP Ab or isotype control. TME modulation was assessed by flow cytometry, scRNA sequencing and cytokines profiling **A.** Anti-IL-18BP Ab increased CD3+, CD8+ and CD4+ T cell numbers in the tumor. **B.** Anti-IL-18BP Ab increased effector CD8+ T cells (CD44⁺CD62L⁻), as well as polyfunctional GZMB⁺IFNγ⁺CD8⁺ T cells, and TNFα+IFNγ⁺-secreting NK cells. **C-D.** Anti-IL-18BP Ab induced polyfunctional non exhausted T cells in the TME. **C.** UMAP projection showing T and NK cells present in E0771 tumors treated with anti-IL-18BP or isotype control. **D.** Enrichment of major T cell subpopulation frequencies in anti-IL-18BP Ab treatment was compared to the control group. **E.** Anti-IL-18BP Ab increased T cell clonal expansion suggesting Ag-specific immune response. Quantification of clonal expansion frequencies in anti-IL-18BP Ab treatment was compared to the control group in TCR+ T cells.

Anti-mouse IL-18BP Ab increases the expansion of proinflammatory myeloid populations and cytokine secretion in the TME

A.

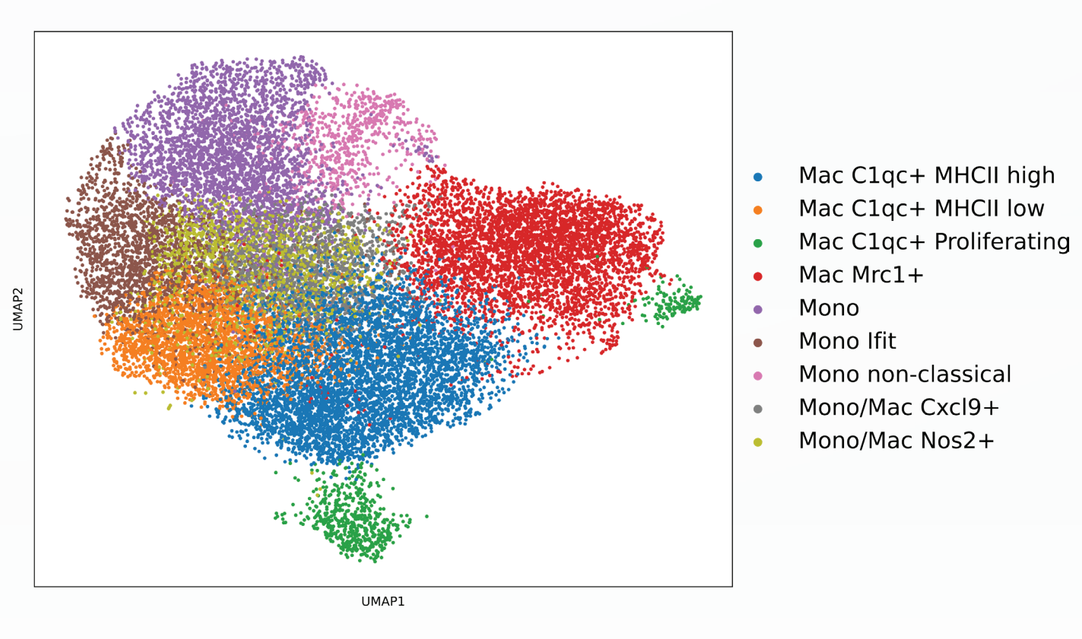
Mac C1qc+ MHCII high
Mac C1qc+ MHCII low
Mac C1qc+ Proliferating
Mac Mrc1+
Mono
Mono Ifit
Mono non-classical
Mono/Mac Cxcl9+
Mono/Mac Nos2+

B.

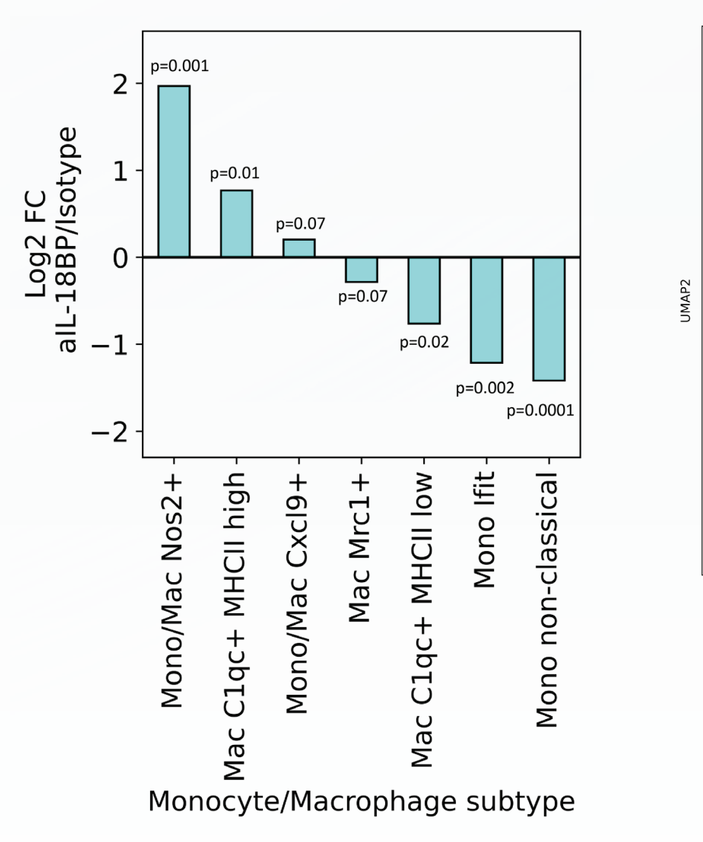
Log2 FC aIL-18BP/Isotype
p=0.001
p=0.01
p=0.07
p=0.07
p=0.02
p=0.002
p=0.0001
Mono/Mac Nos2+
Mac C1qc+ MHCII high
Mono/Mac Cxcl9+
Mac Mrc1+
Mac C1qc+ MHCII low
Mono Ifit
Mono non-classical
Monocyte/Macrophage subtype

C.

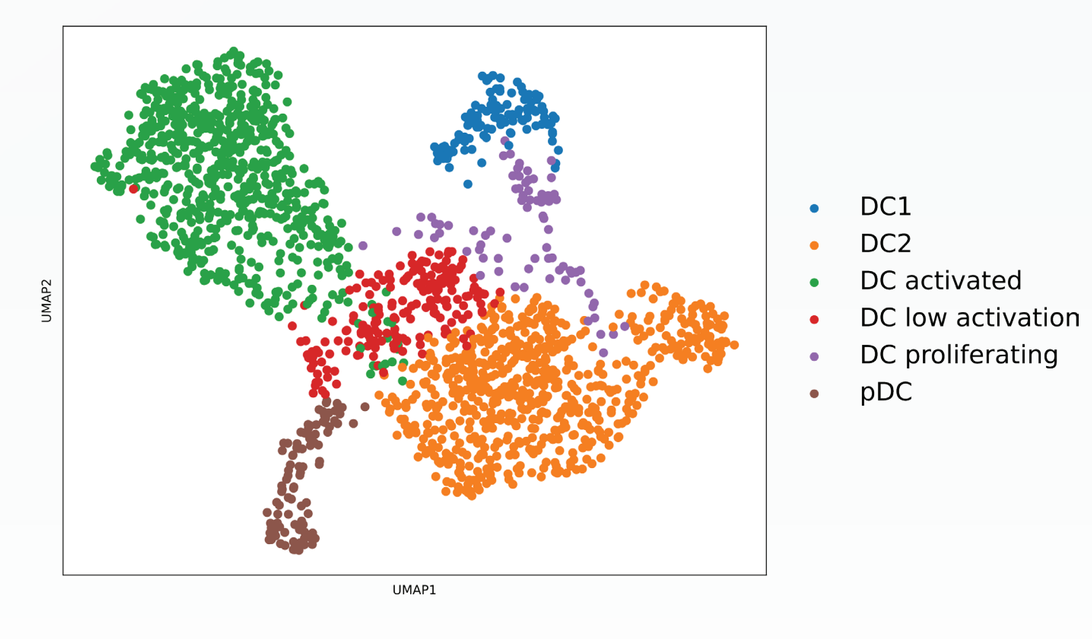
DC1
DC2
DC activated
DC low activation
DC proliferating
pDC

D.

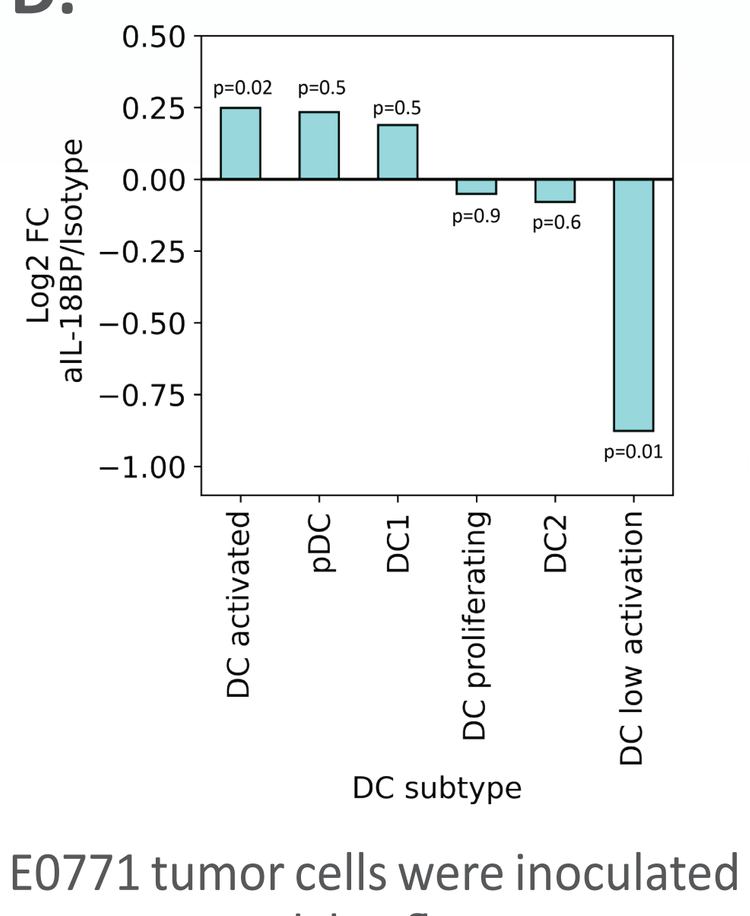
Log2 FC aIL-18BP/Isotype
p=0.02
p=0.5
p=0.5
p=0.9
p=0.6
p=0.01
DC activated
pDC
DC1
DC proliferating
DC2
DC low activation
DC subtype

E.

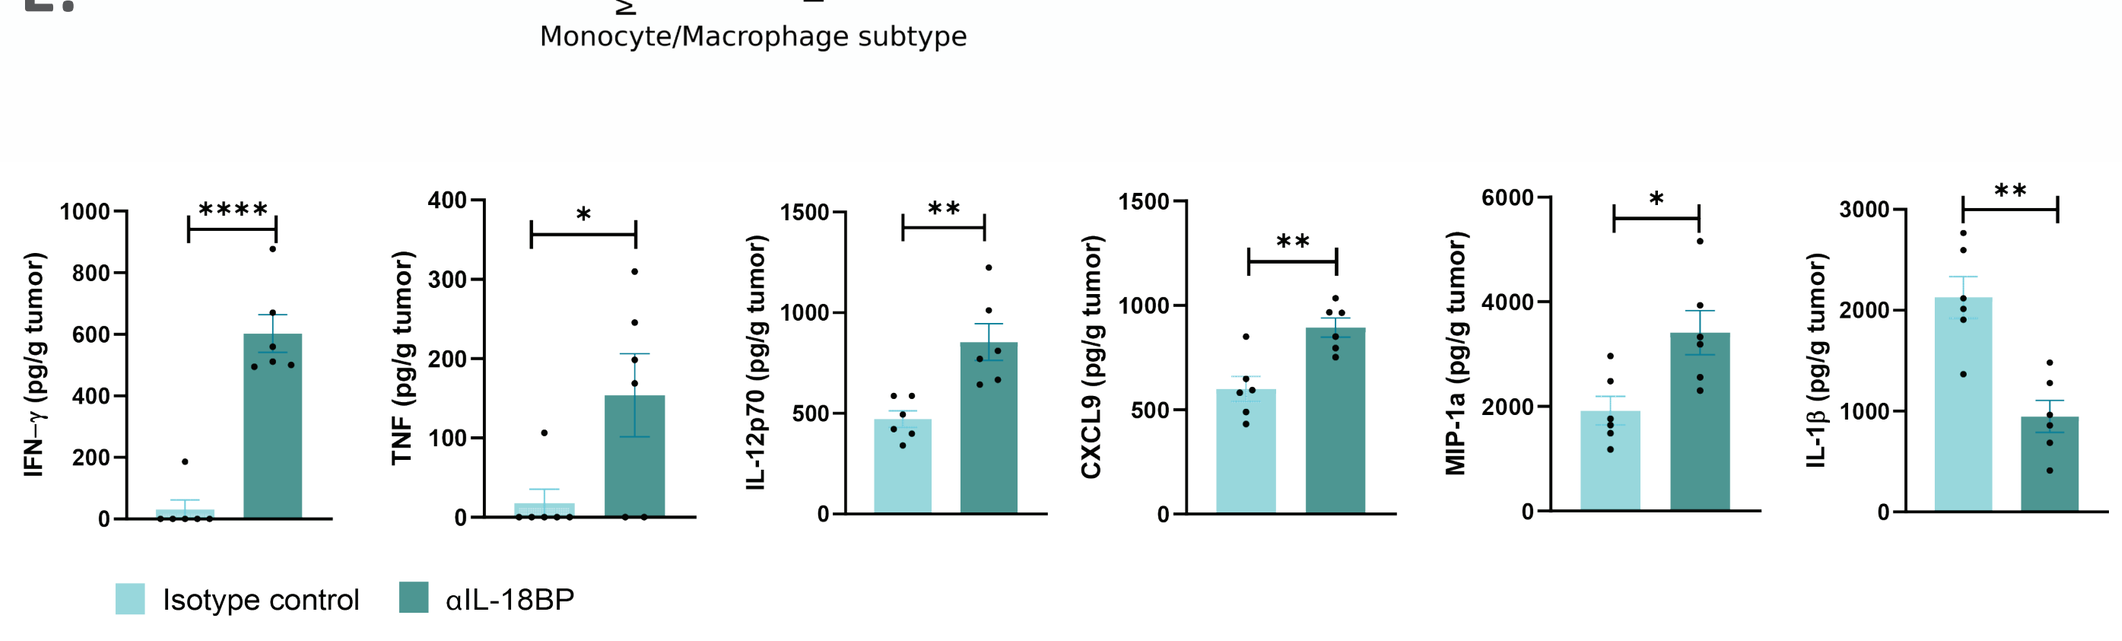
IFN-γ (pg/g tumor)
TNF (pg/g tumor)
IL-12p70 (pg/g tumor)
CXCL9 (pg/g tumor)
MIP-1a (pg/g tumor)
IL-1β (pg/g tumor)
Isotype control
αIL-18BP

E0771 tumor cells were inoculated in C57Bl/6 mice and treated either with anti-mouse IL-18BP Ab or isotype control. TME modulation was assessed by flow cytometry, scRNA sequencing and cytokine profiling. **A-B.** Anti- IL-18BP Ab increased proinflammatory macrophages in the TME. **A.** UMAP projection showing tumor-associated monocyte and macrophage subpopulations present in E0771 tumors treated with anti-IL-18BP or isotype control. **B.** Enrichment of major tumor-associated monocytes and macrophages frequencies in anti-IL-18BP Ab treatment was compared to the control group. **C-D.** Anti-IL-18BP Ab increased activated DC population in the TME. **C.** UMAP projection of DC subpopulations in E0771 tumors treated with anti-IL-18BP or isotype control. **D.** Enrichment of DC subpopulations frequencies in anti-IL-18BP Ab treatment compared to the control group. **E.** Anti-IL-18BP Ab increased proinflammatory cytokine secretion in the TME.

Anti-mouse IL-18BP Ab modulates the TME without affecting the periphery

A.

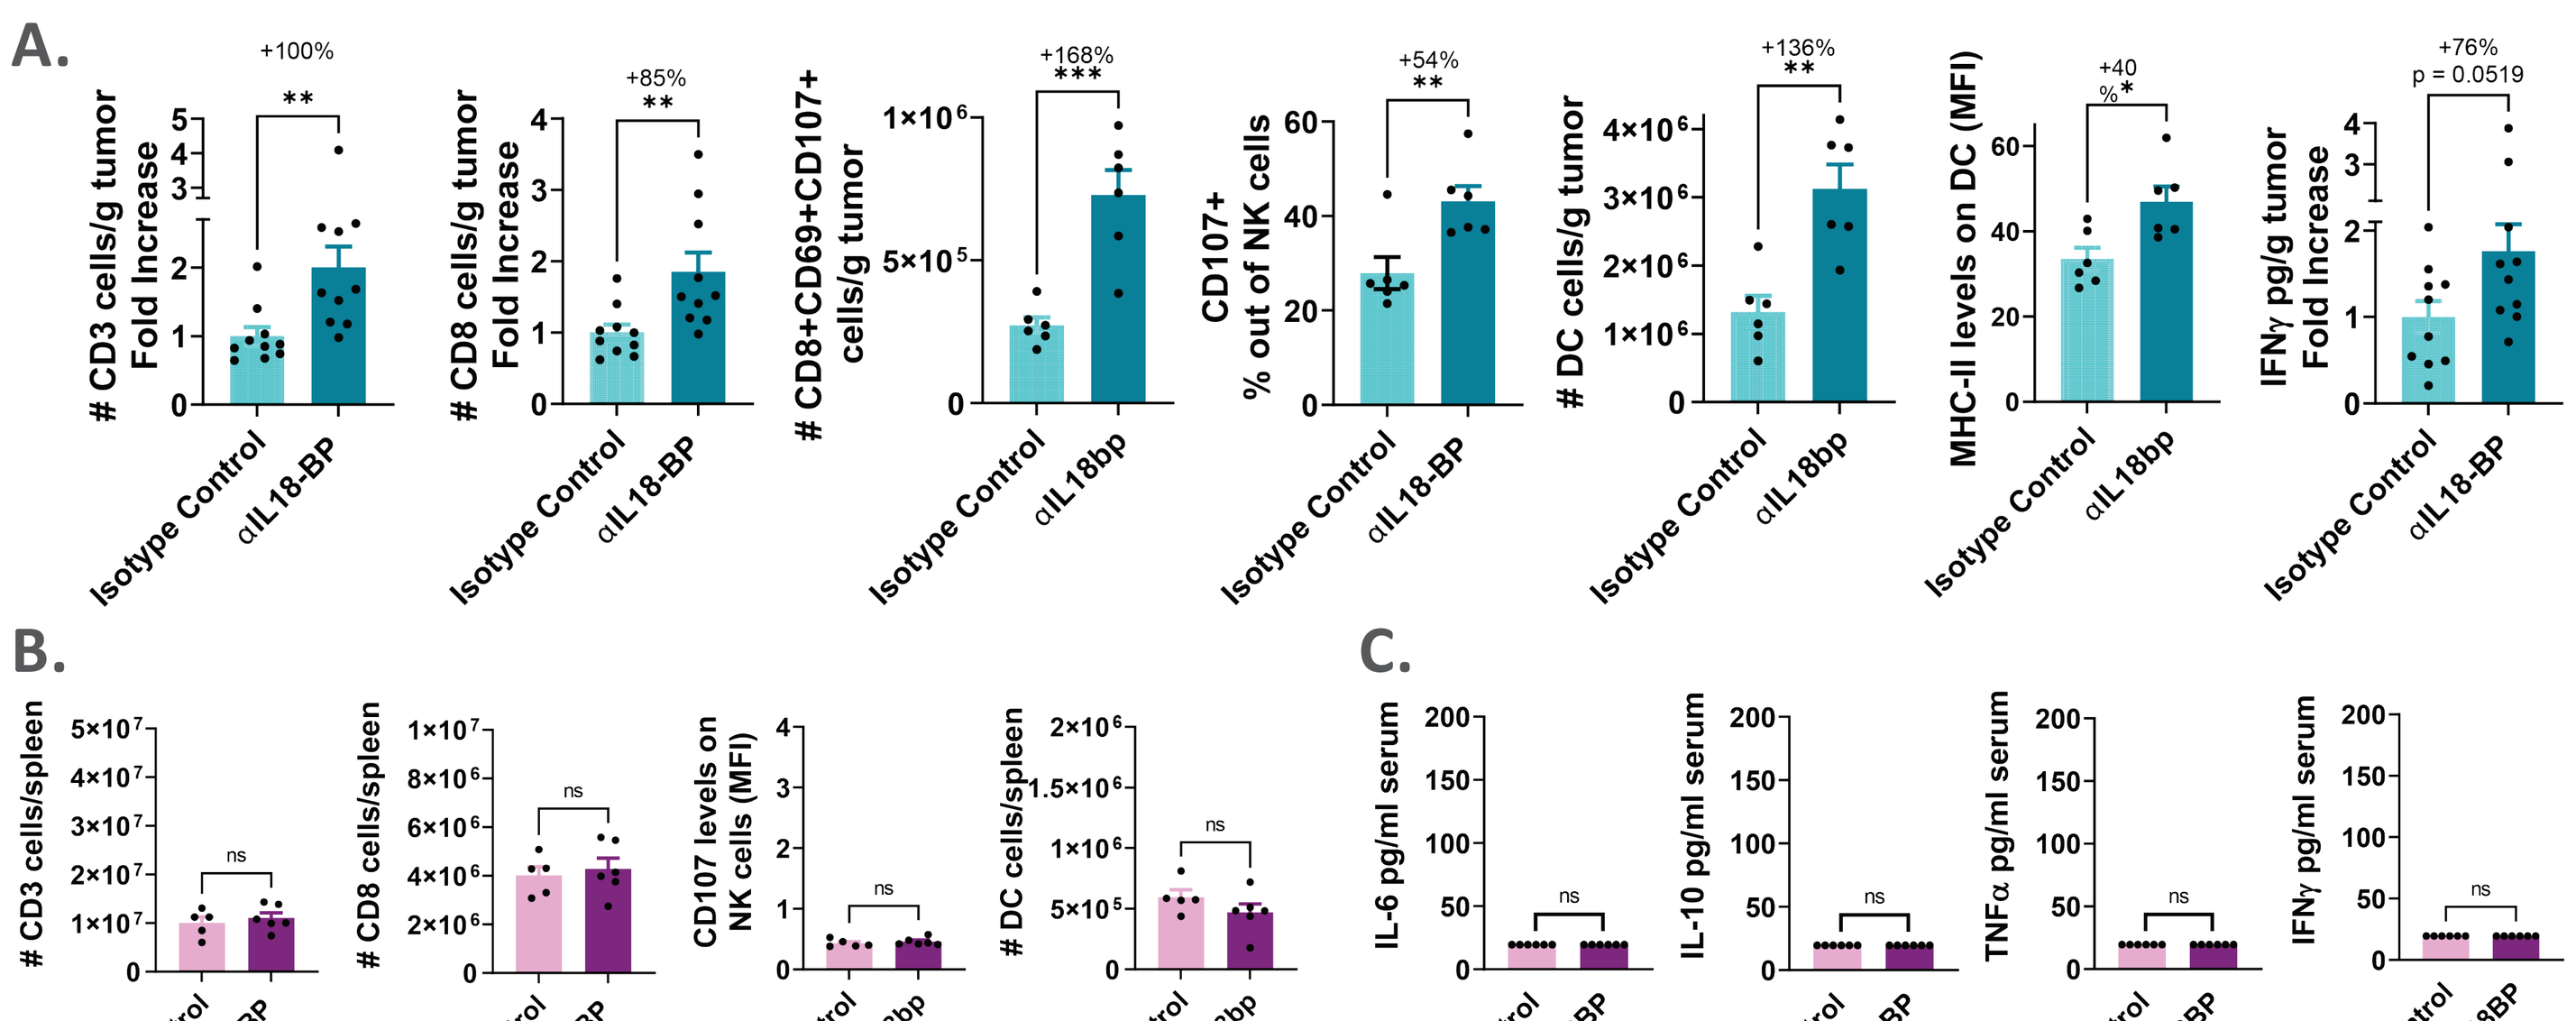
+100%
+85%
+168%
+54%
+136%
+40%
+76%
p = 0.0519
CD3 cells/g tumor Fold Increase
CD8 cells/g tumor Fold Increase
CD8+CD69+CD107+ cells/g tumor
CD107+ % out of NK cells
DC cells/g tumor
MHC-II levels on DC (MFI)
IFNγ pg/g tumor Fold Increase
Isotype Control
αIL18-BP

B.

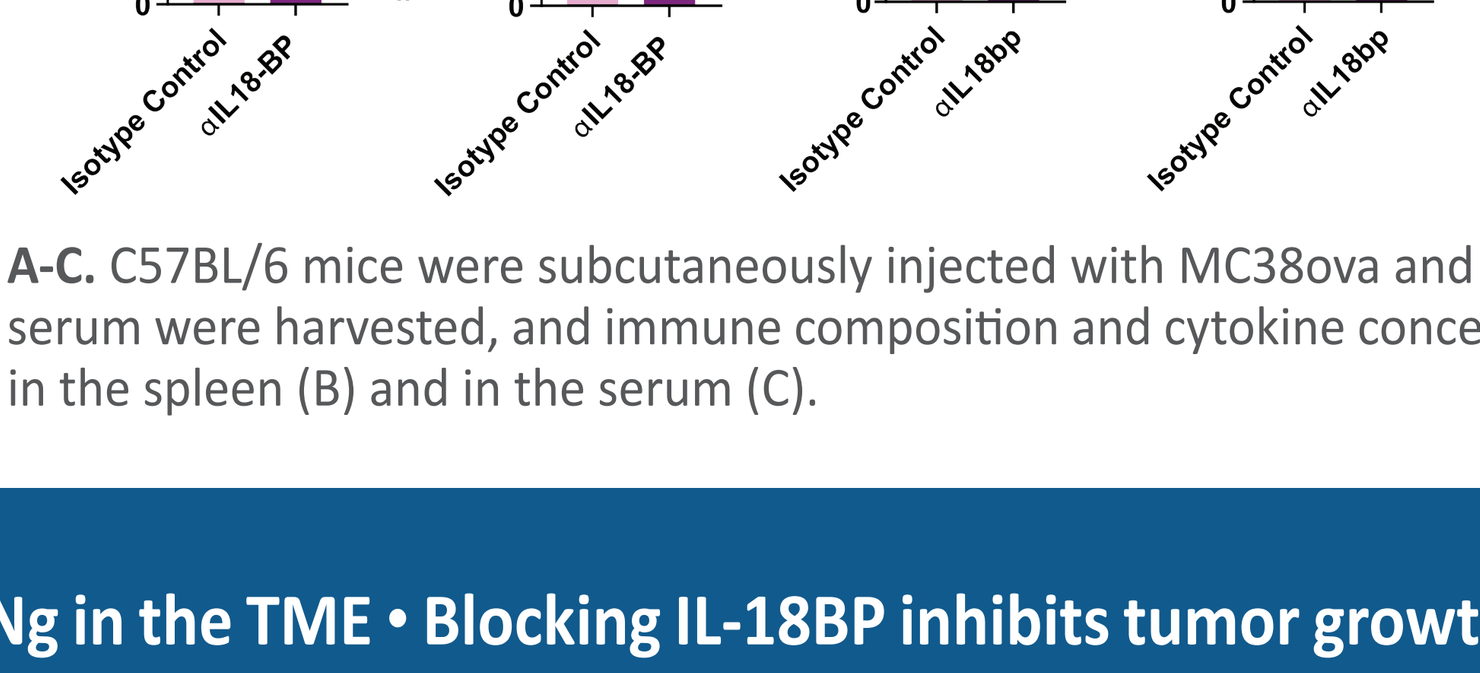
CD3 cells/spleen
CD8 cells/spleen
CD107 levels on NK cells (MFI)
DC cells/spleen
ns
Isotype Control
αIL18-BP

C.

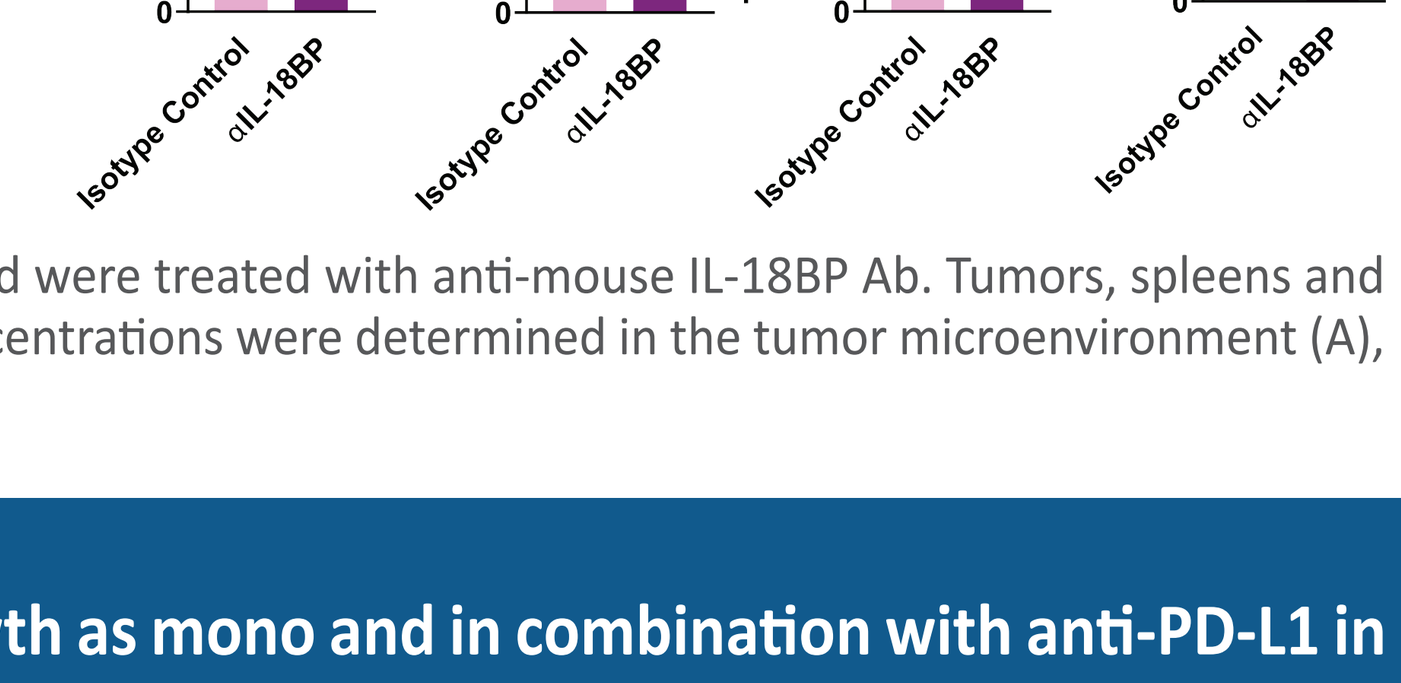
IL-6 pg/ml serum
IL-10 pg/ml serum
TNFα pg/ml serum
IFNγ pg/ml serum
ns
Isotype Control
αIL-18BP

A-C. C57BL/6 mice were subcutaneously injected with MC38ova and were treated with anti-mouse IL-18BP Ab. Tumors, spleens and serum were harvested, and immune composition and cytokine concentrations were determined in the tumor microenvironment (A), in the spleen (B) and in the serum (C).

CONCLUSION

• IL-18, upregulated in the TME, is inhibited by IL-18BP, which is induced by IFNg in the TME • Blocking IL-18BP inhibits tumor growth as mono and in combination with anti-PD-L1 in murine cancer models • Immune modulation following treatment with anti-IL-18BP Ab is restricted to the TME suggesting favorable therapeutic window, in contrast to recombinant therapeutic cytokines that are active systemically • COM503, a human IgG4 high affinity anti-IL-18BP blocker Ab, releases IL-18 to activate T and NK cells • IND expected in 2024