Unleashing natural IL-18 activity using an anti-IL-18BP blocker antibody induces potent immune stimulation and anti-tumor activity

Assaf Menachem, Zoya Alteber, Gady Cojocaru, Tal Fridman Kfir, Dan Blat, Olga Leiderman, Moran Galperin, Lital Sever, Nadav Cohen, Keren Cohen, Liron Soffer, Karin Meyer, Keren Menachem, Hadas Galon Tilleman, Michal Perpinial, Evgeny Tatirovsky, Pierre Ferre, Eran Ophir

Compugen Ltd, Holon, Israel

compugen
FROM CODE TO CURE®

Background

Conventional cytokines have limited anti-cancer efficacy mostly due to narrow therapeutic window and systemic adverse effects. IL-18 is an inflammasome induced proinflammatory cytokine that enhances T and NK cell activity and stimulates IFNg production. The activity of IL-18 is naturally blocked by a high affinity endogenous binding protein (IL-18BP). To study the expression of IL-18 and IL-18BP in the tumor, we measured their levels in tumor derived supernatants (TDS) by ELISA assays. To unleash endogenous bound IL-18 activity we generated COM503, high affinity anti human IL-18BP antibody (Ab). COM503 activity was examined in T and NK cell-based assays. For mouse in vivo studies, a surrogate Ab was generated and examined in multiple models either as monotherapy or in combinations with immune checkpoints (ICP) blockers.

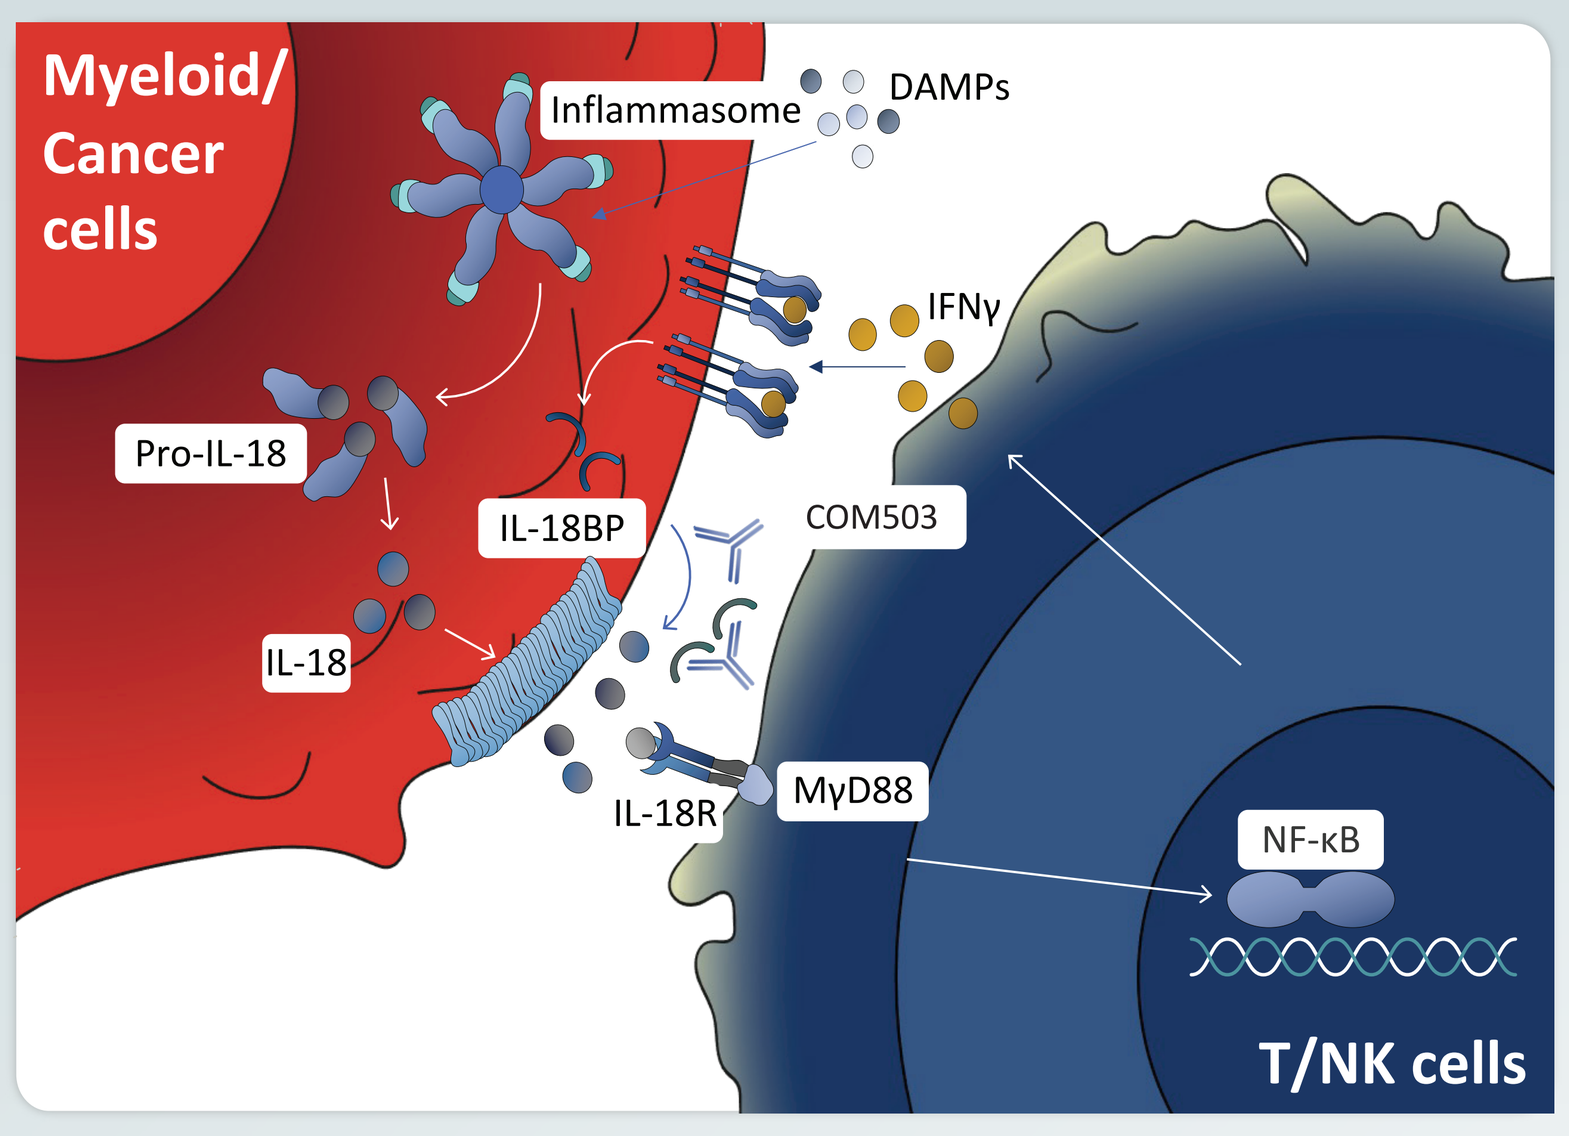
Myeloid/ Cancer cells
Inflammasome
DAMPs
IFNγ
Pro-IL-18
IL-18BP
COM503
IL-18
IL-18R
MyD88
NF-κB
T/NK cells

IL-18 is:
- An effector cytokine
- Secreted upon Inflammasome activation
- Upregulated in the TME

IL-18BP:
- Binds IL-18 and blocks its immune stimulatory activity
- L-18BP secretion is increased via an IL-18-mediated feedback mechanism

COM503:
- Has the potential to Induce potent anti tumor responses and pronounced TME-constrained immune modulation

Inflammasome induced cytokines such as IL-18 and IL-1b are abundant in the tumor microenvironment

A.

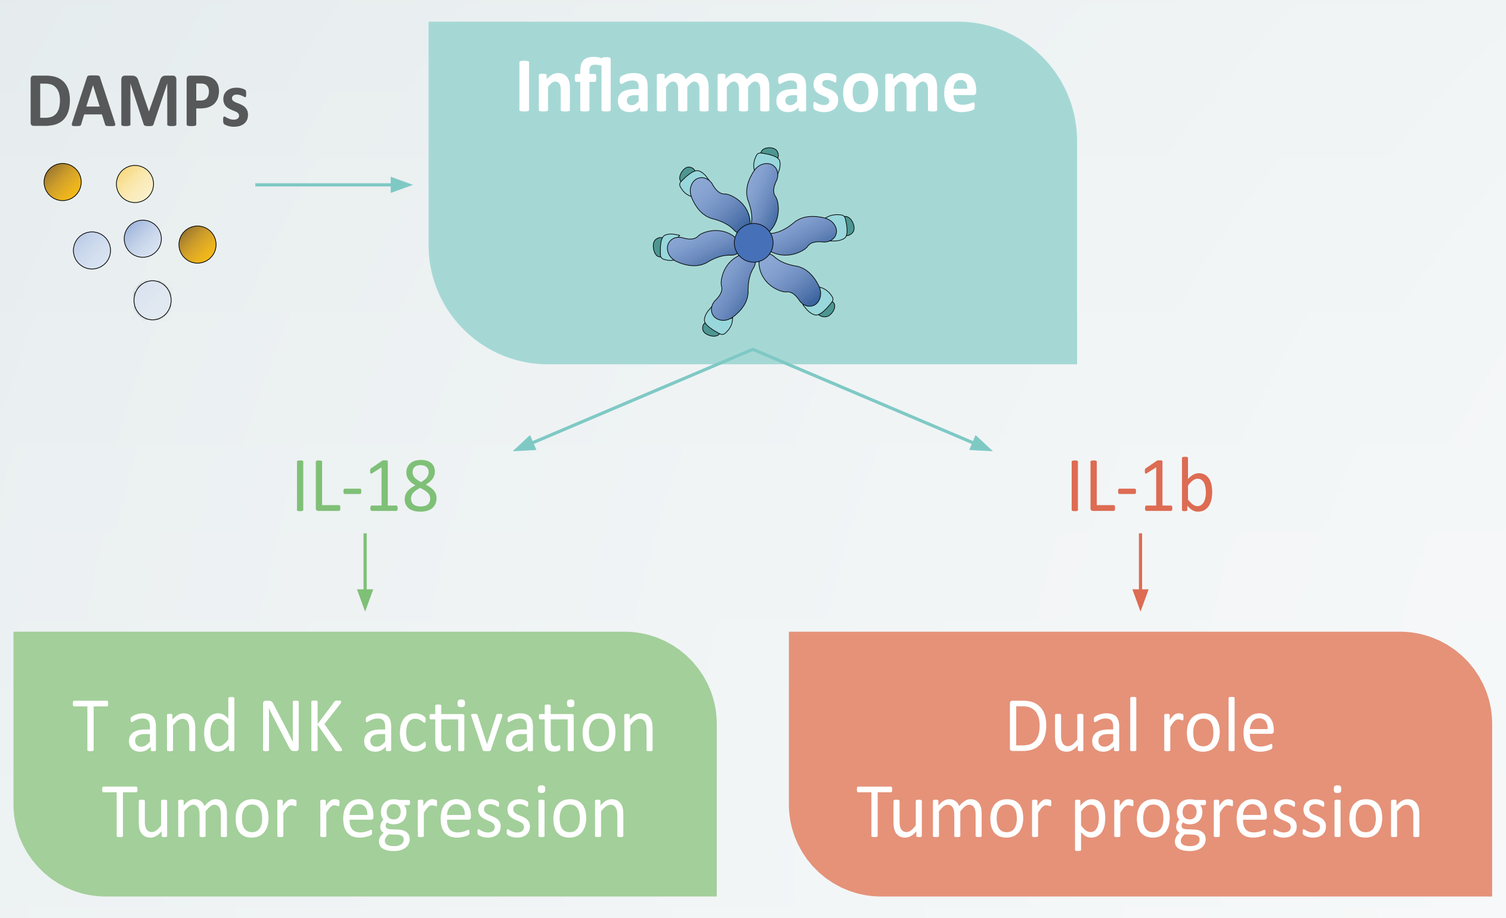
DAMPs
Inflammasome
IL-18
IL-1b
T and NK activation
Tumor regression
Dual role
Tumor progression

B.

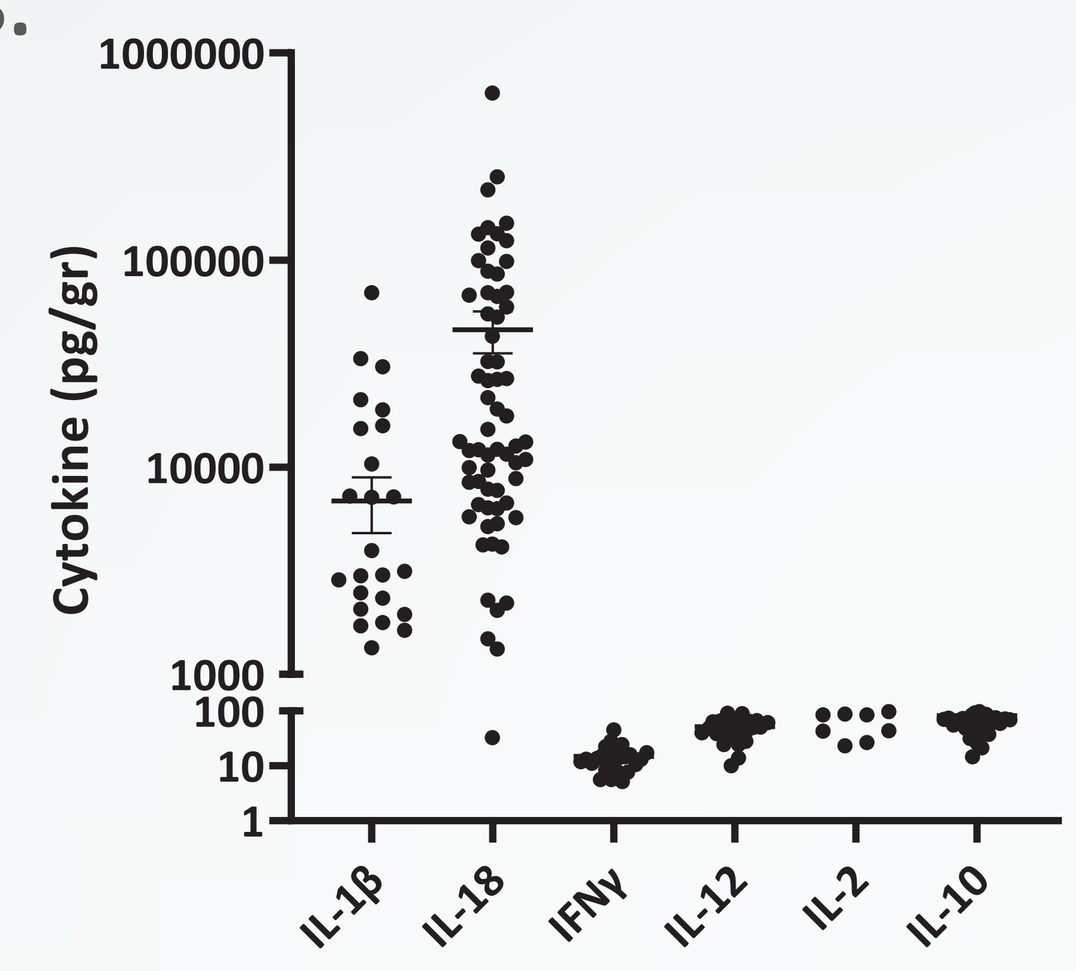
Cytokine (pg/gr)
IL-1β
IL-18
IFNγ
IL-12
IL-2
IL-10

List of indications:
Colorectal
NSCLC
Ovary
Renal
Breast
Endometrial
H&N
Stomach
Pancreas
Esophageal
Bladder
Rectal

A. IL-18 and IL-1b are inflammasome derived cytokines with opposite effects in the tumor microenvironment (TME). **B.** Levels of cytokines in TDS across indications.

IL-18 is upregulated in cancer patient serum and pathway is elevated in the TME across indications

A.

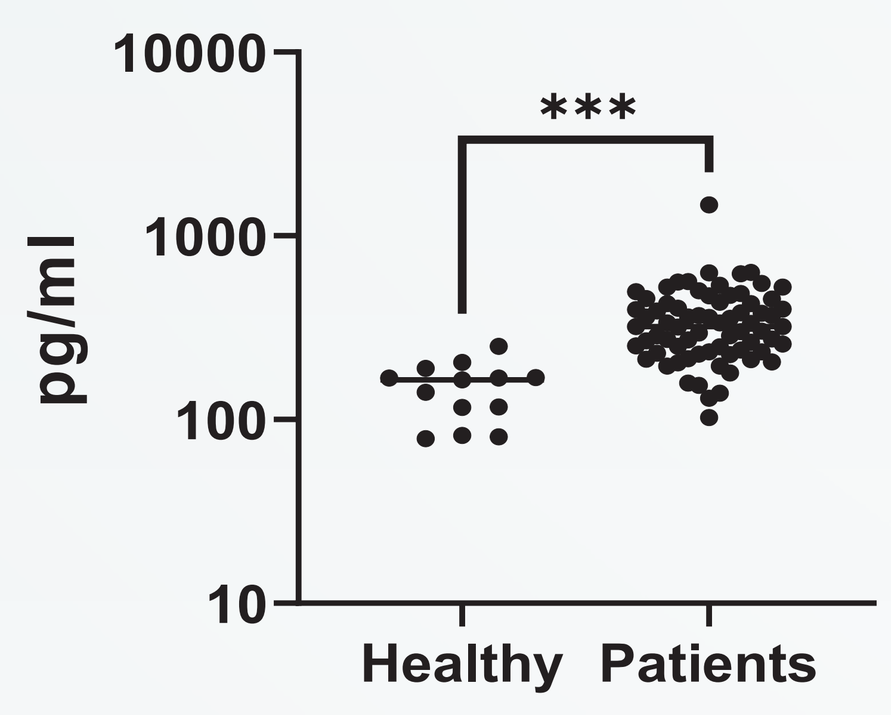
pg/ml

Healthy
Patients

B.

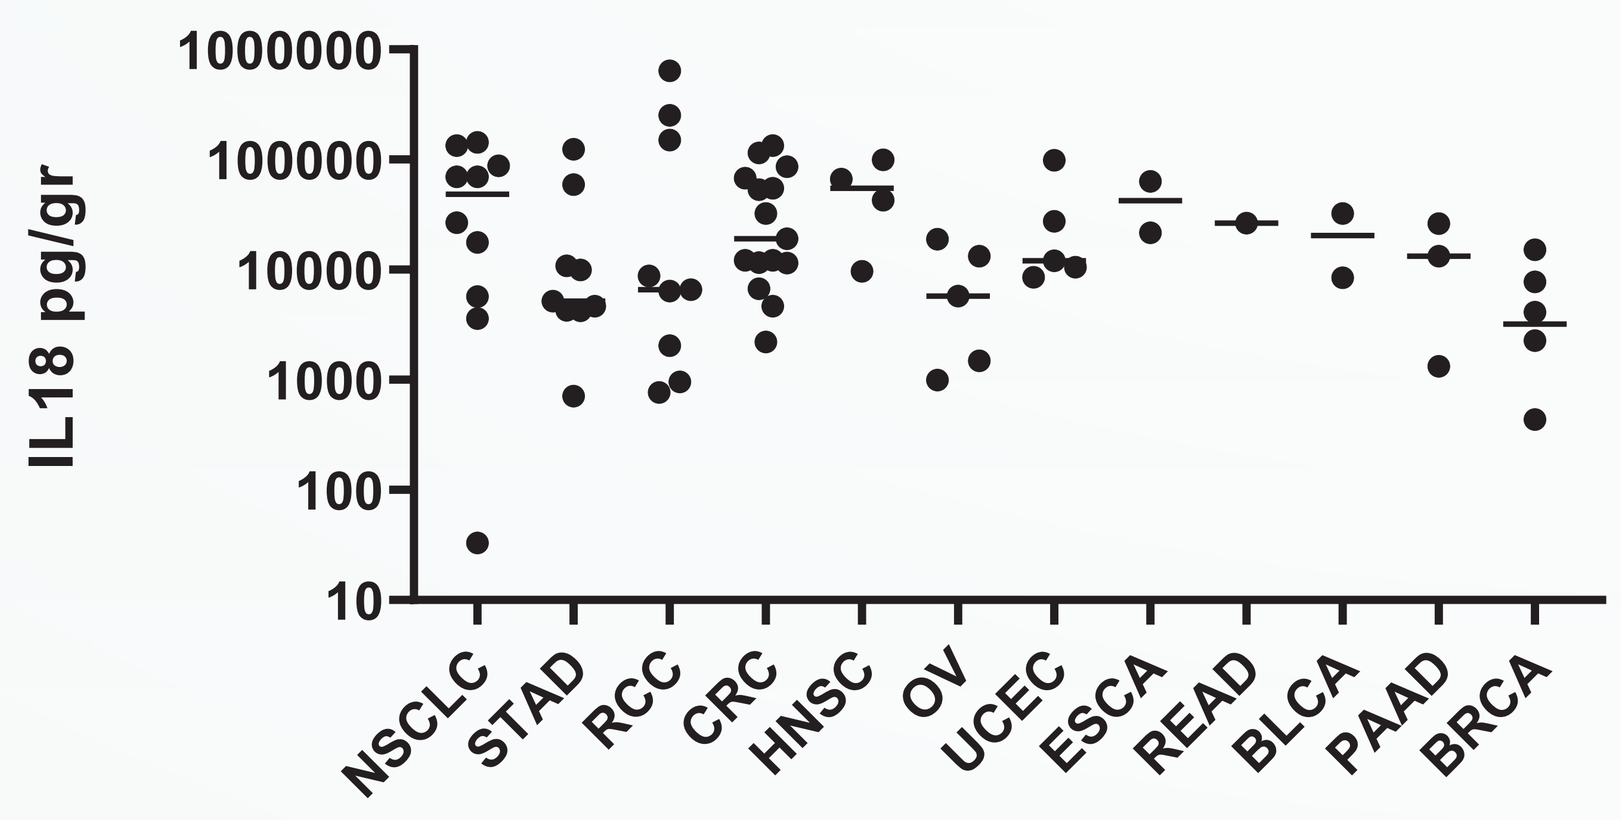
IL18 pg/gr
NSCLC
STAD
RCC
CRC
HNSC
OV
UCEC
ESCA
READ
BLCA
PAAD
BRCA

C.

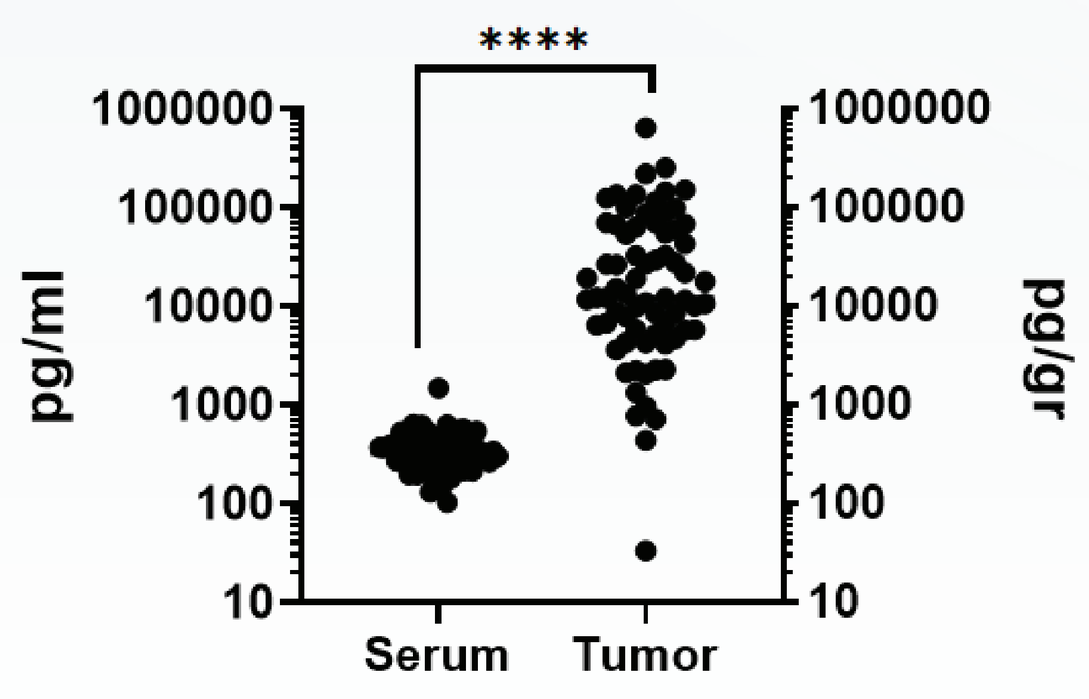
pg/ml

pg/gr
Serum
Tumor

D.

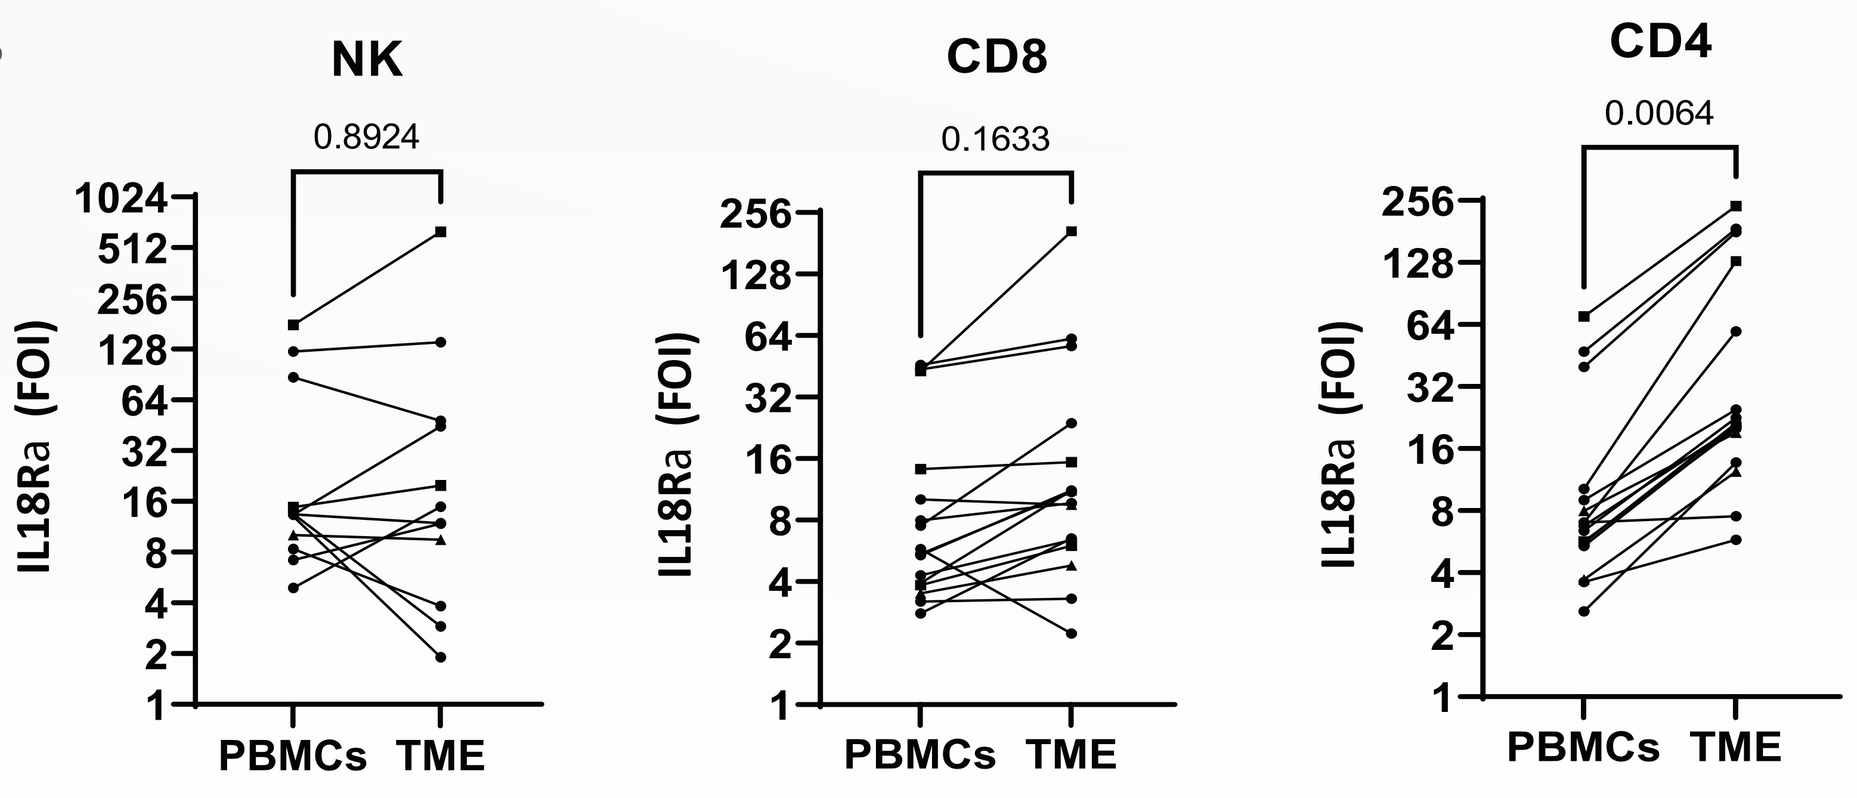
NK
0.8924
CD8
0.1633
CD4
0.0064
IL18Ra (FOI)
PBMCs
TME

A. IL-18 levels in serum of cancer patients are increased compared to levels in healthy donor serum. **B.** IL-18 levels in TDS across indications. **C.** IL-18 levels in TDS are increased compared to levels in serum. **D.** IL-18 receptor a subunit (IL-18Ra) expression is induced on tumor infiltrating lymphocytes (TILs) in the TME compared to periphery. Statistical analysis was preformed using paired t test (two tailed). $P \leq 0.001$***, $P \leq 0.0005$****

IL-18BP-Bound IL-18 levels in the TME are above amount required for T cell activation in vitro

A.

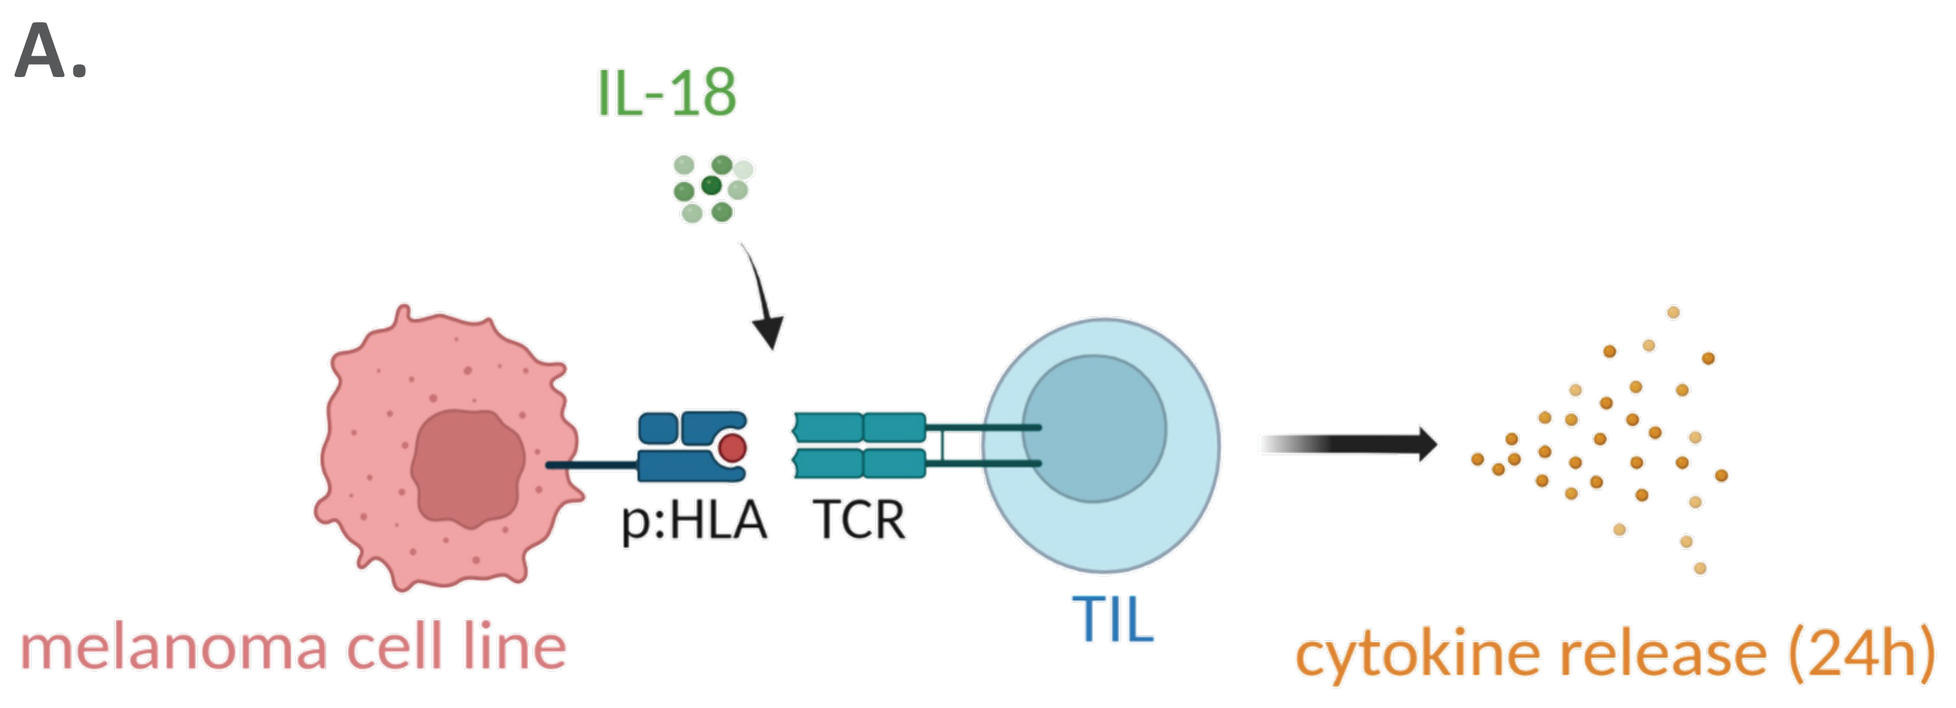
IL-18
p:HLA
TCR
melanoma cell line
TIL
cytokine release (24h)

C.

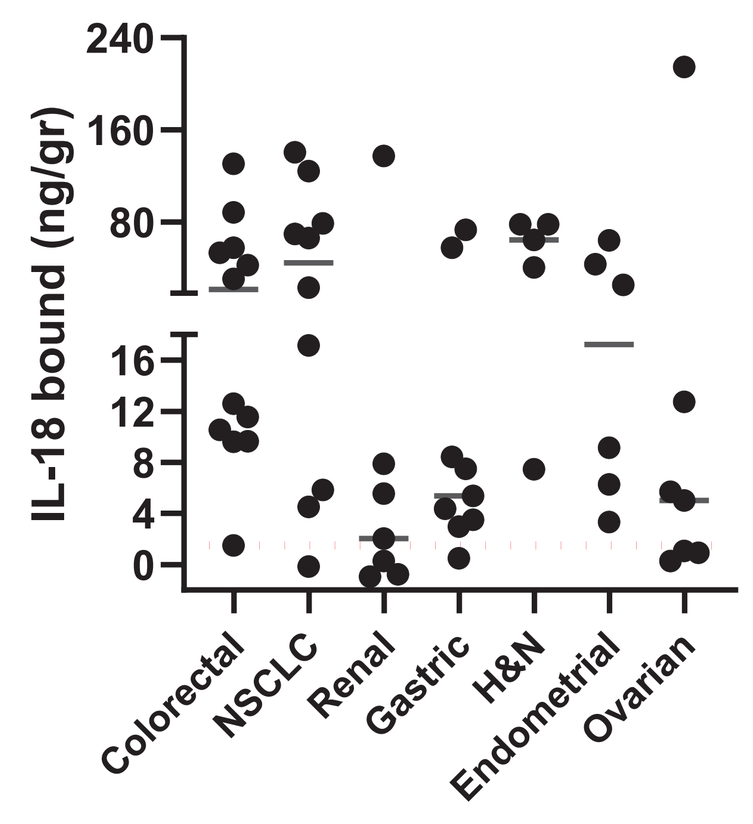
IL-18 bound (ng/gr)
Colorectal
NSCLC
Renal
Gastric
H&N
Endometrial
Ovarian

B.

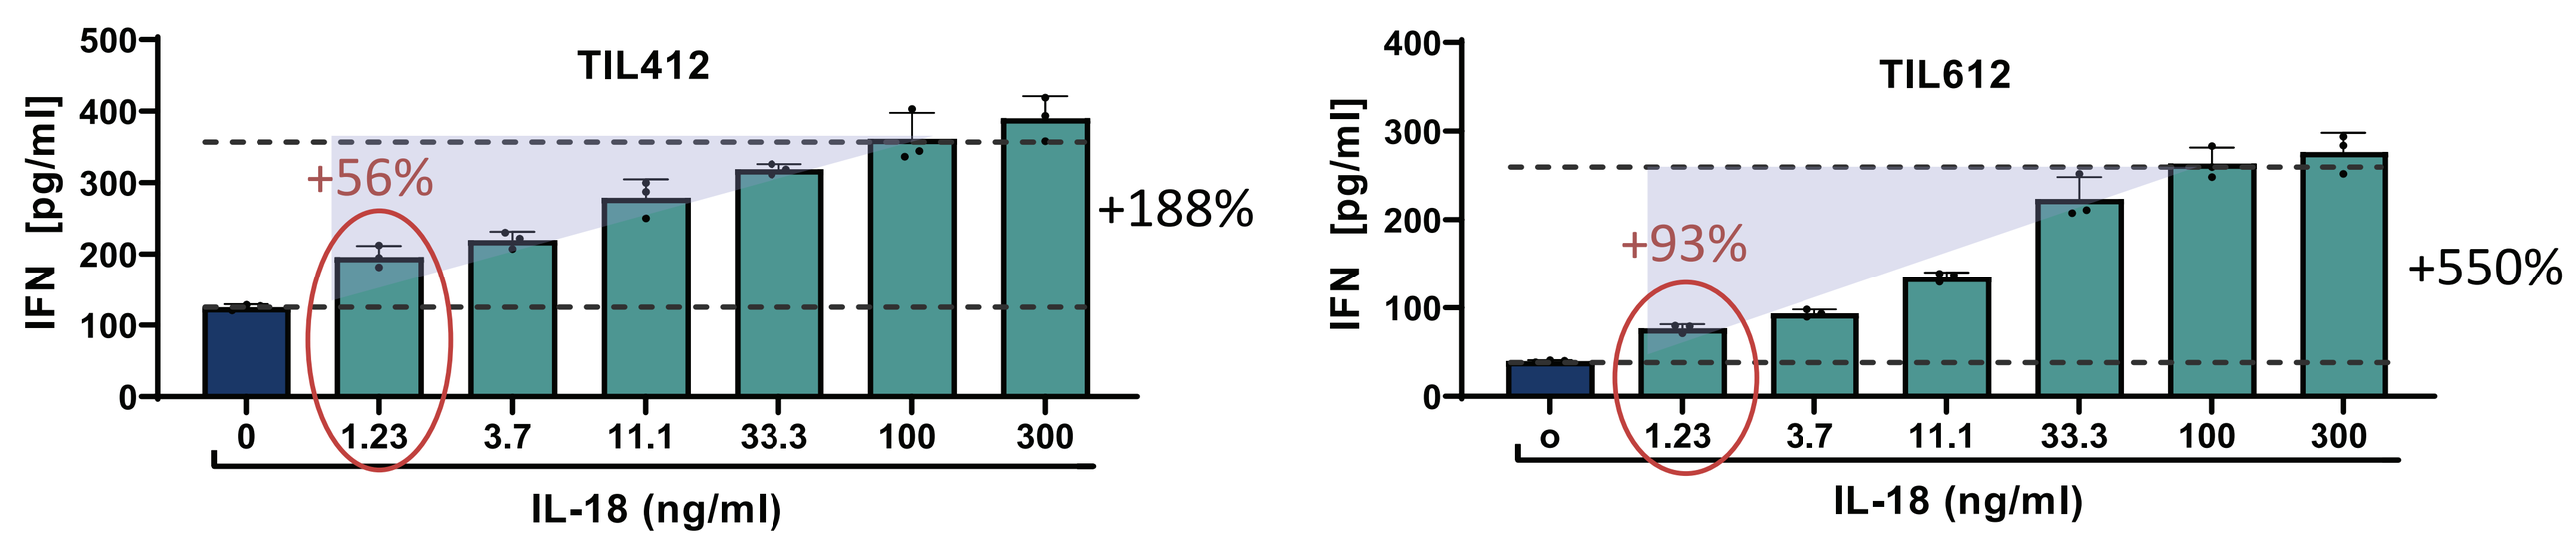
TIL412
+56%
+188%
TIL612
+93%
+550%
IFN [pg/ml]
IL-18 (ng/ml)

A. Schematic representation of assay setup. **B.** Recombinant (r) human IL-18 increased IFNg release by stimulated CD8+ tumor infiltrating lymphocytes (TILs) in TILs-tumor cells co-culture assay in a dose-dependent manner. Activity was seen with as low as 1ng/ml of IL-18. **C.** Levels of bound IL-18 in TDS across indications are mostly above the level required for in vitro T cell activation. IL-18BP-bound IL-18 levels were calculated by deducting IL-18 free from total IL-18 measured for each sample by two separate ELISA kits. Dashed red line represent the level required for functional activity (1.5ng/gr).

IL-18BP binds IL-18 with high affinity in human and mouse

A. HUMAN

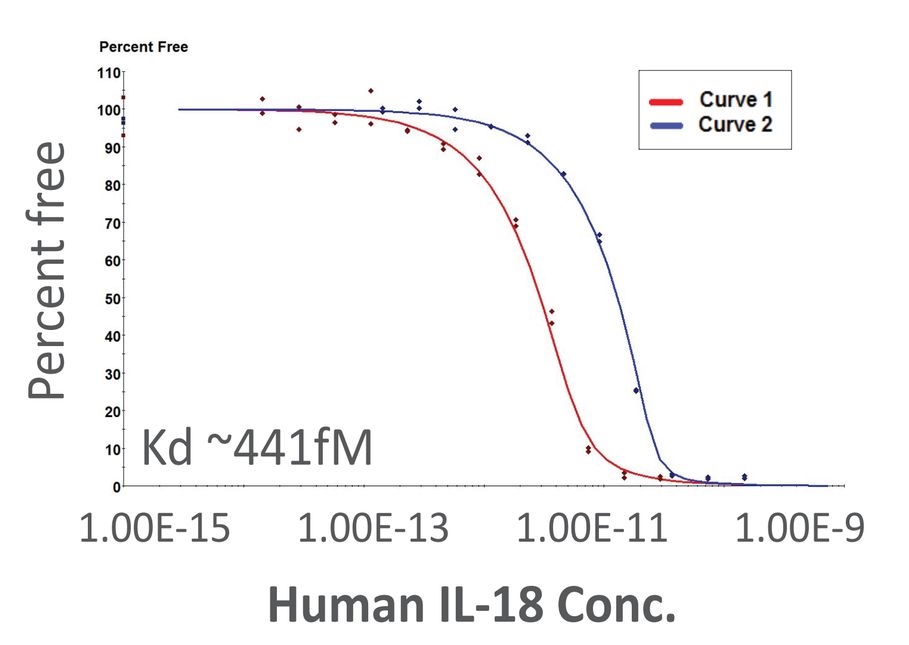
Percent free
Curve 1
Curve 2
Kd ~441fM
Human IL-18 Conc.

B. MOUSE

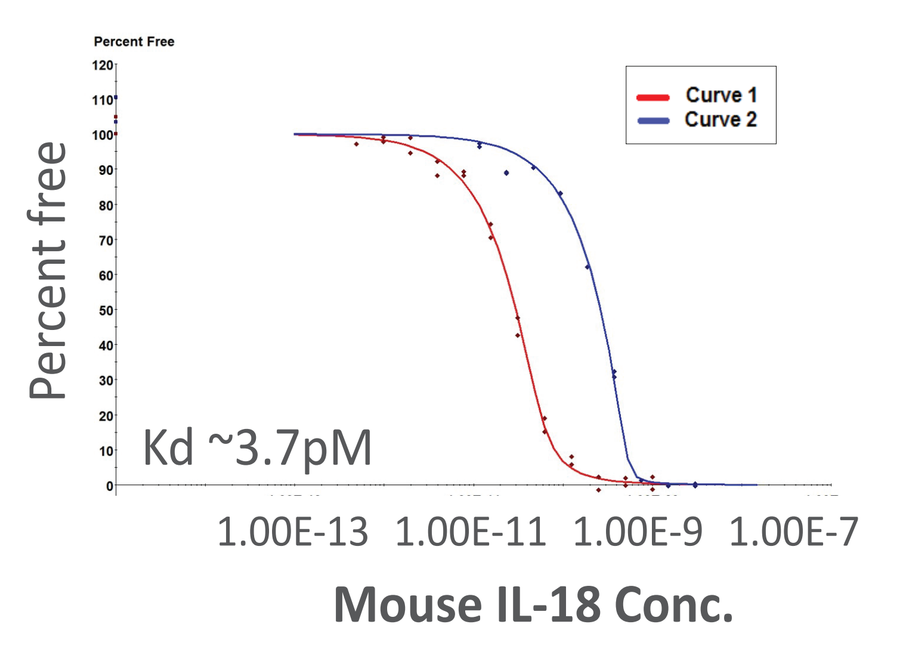
Percent free
Curve 1
Curve 2
Kd ~3.7pM
Mouse IL-18 Conc.

A-B. Human (A) mouse (B) IL18-IL18BP interaction measurements in KinExA. For each run in KinExA, two curves with different column binding protein (CBP) concentrations were run and analyzed using n-curve analysis to determine the Kd.

IL-18BP is upregulated in tumor associated macrophages in the TME and expressed across multiple human cancer indications

A.

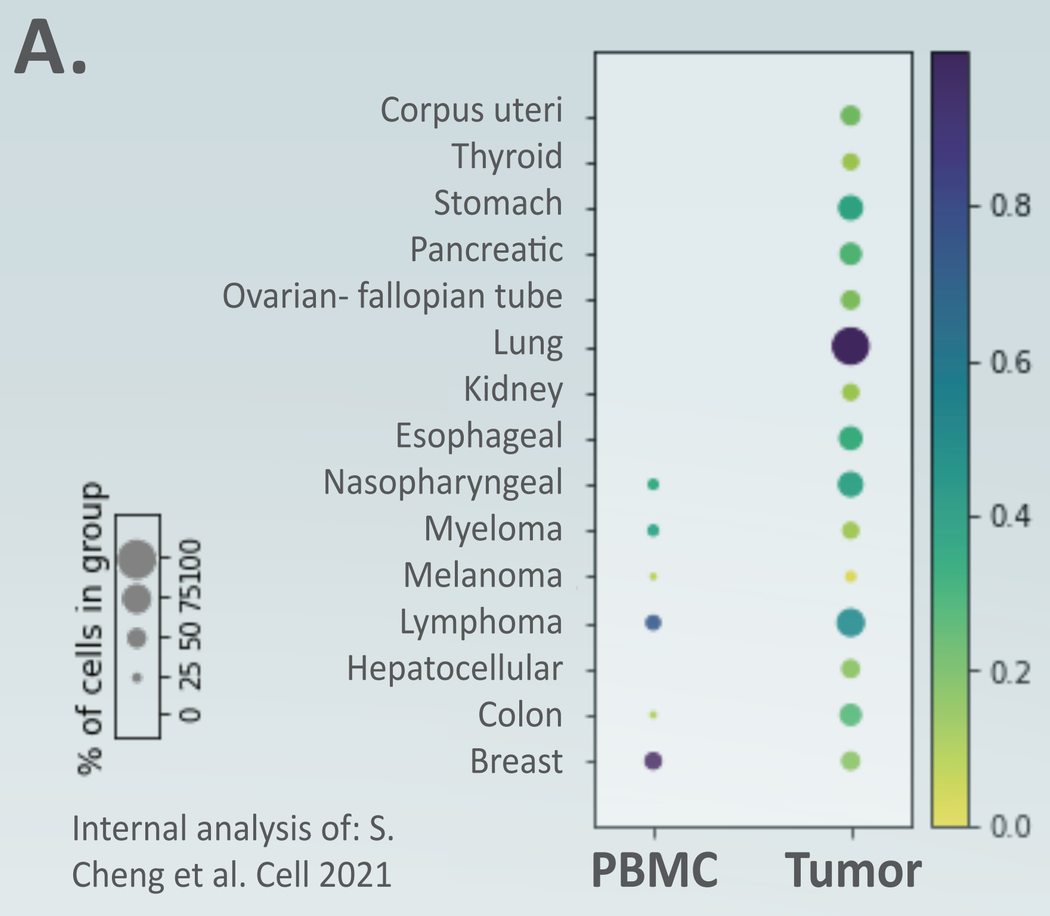
Corpus uteri
Thyroid
Stomach
Pancreatic
Ovarian- fallopian tube
Lung
Kidney
Esophageal
Nasopharyngeal
Myeloma
Melanoma
Lymphoma
Hepatocellular
Colon
Breast
% of cells in group
PBMC
Tumor
Internal analysis of: S. Cheng et al. Cell 2021

B.

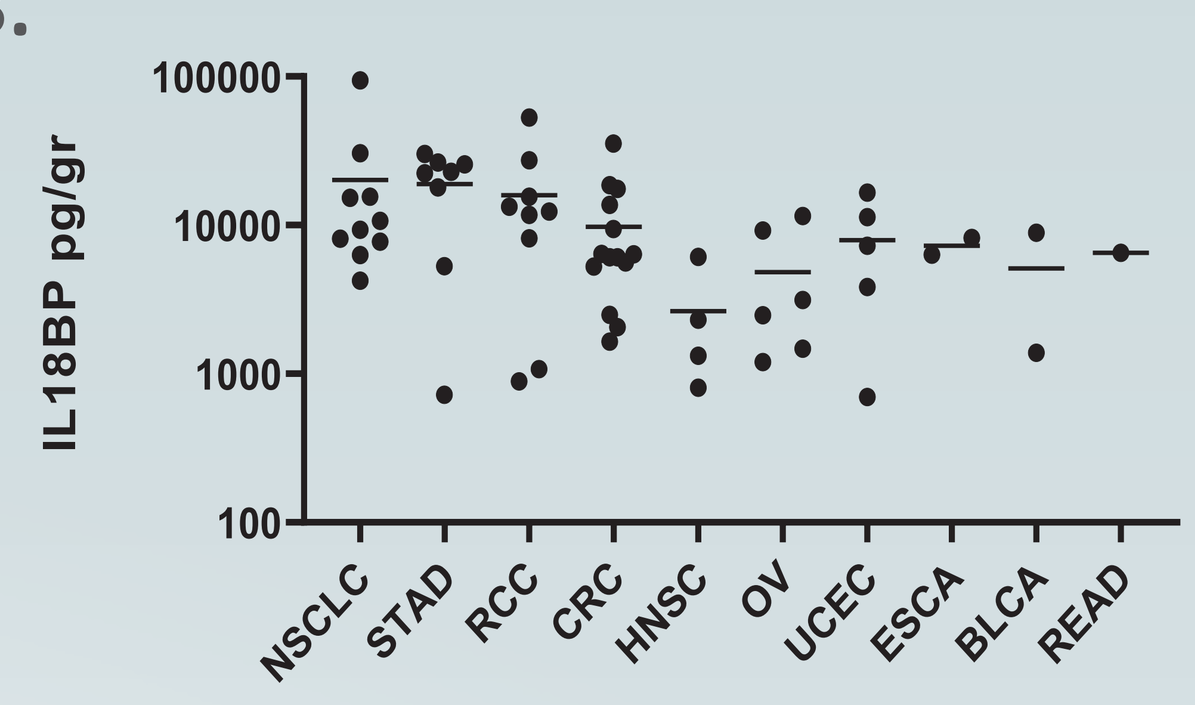
IL18BP pg/gr
NSCLC
STAD
RCC
CRC
HNSC
OV
UCEC
ESCA
BLCA
READ

A. scRNA analyses of tumor-infiltrating myeloid cells, across indications showing that IL-18BP is up-regulated in myeloid population in the TME compared to the periphery (PBMCs). **B.** IL-18BP protein levels in TDS across indications.

COM503 a fully human, high affinity anti-IL18BP antibody restores human TIL and NK cell activity

A.

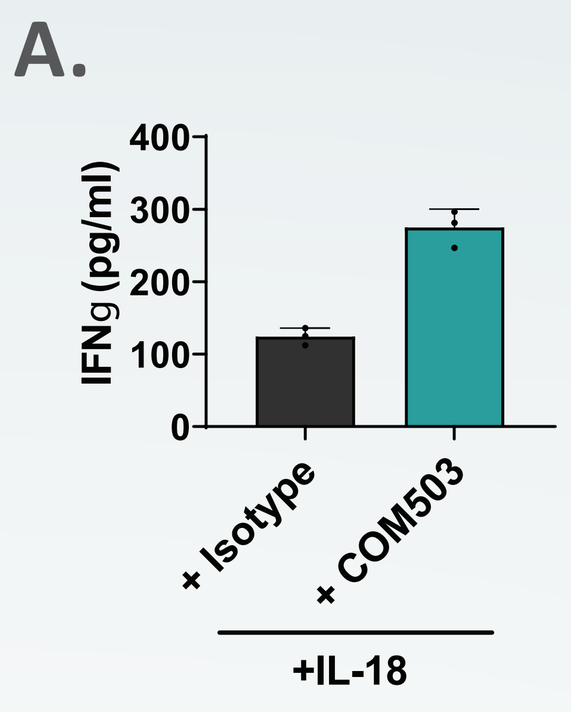
IFNg (pg/ml)
+ Isotype
+ COM503
+IL-18

B.

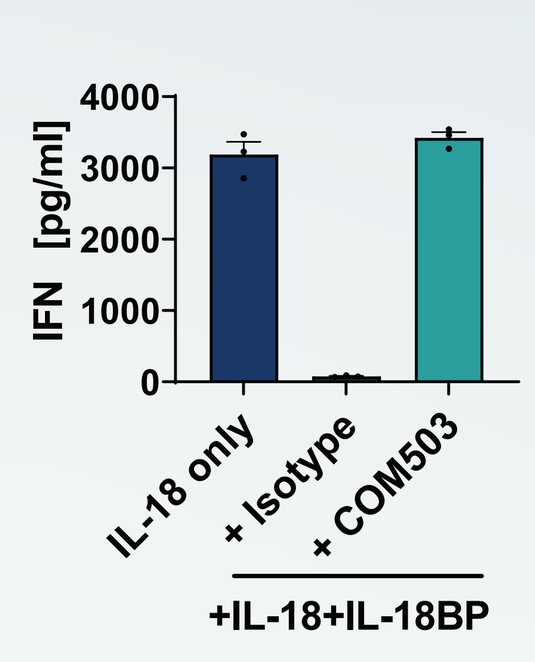
IFN [pg/ml]
IL-18 only
+ Isotype
+ COM503
+IL-18+IL-18BP

C.

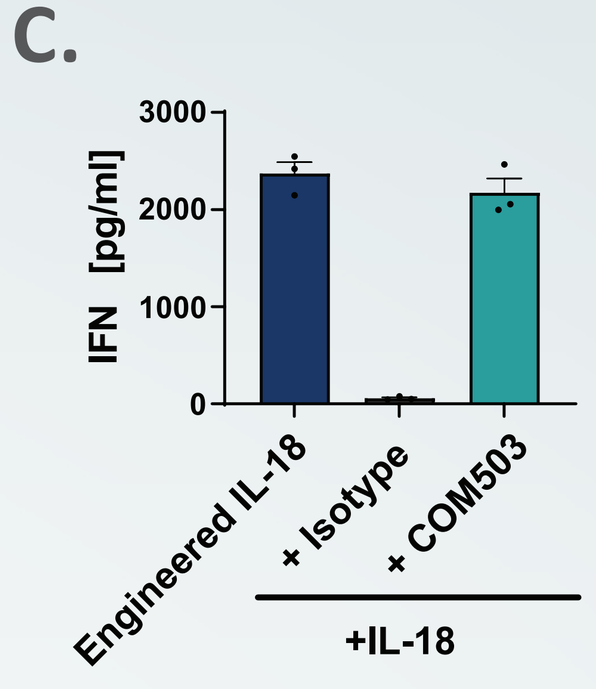
IFN [pg/ml]
Engineered IL-18
+ Isotype
+ COM503
+IL-18

D.

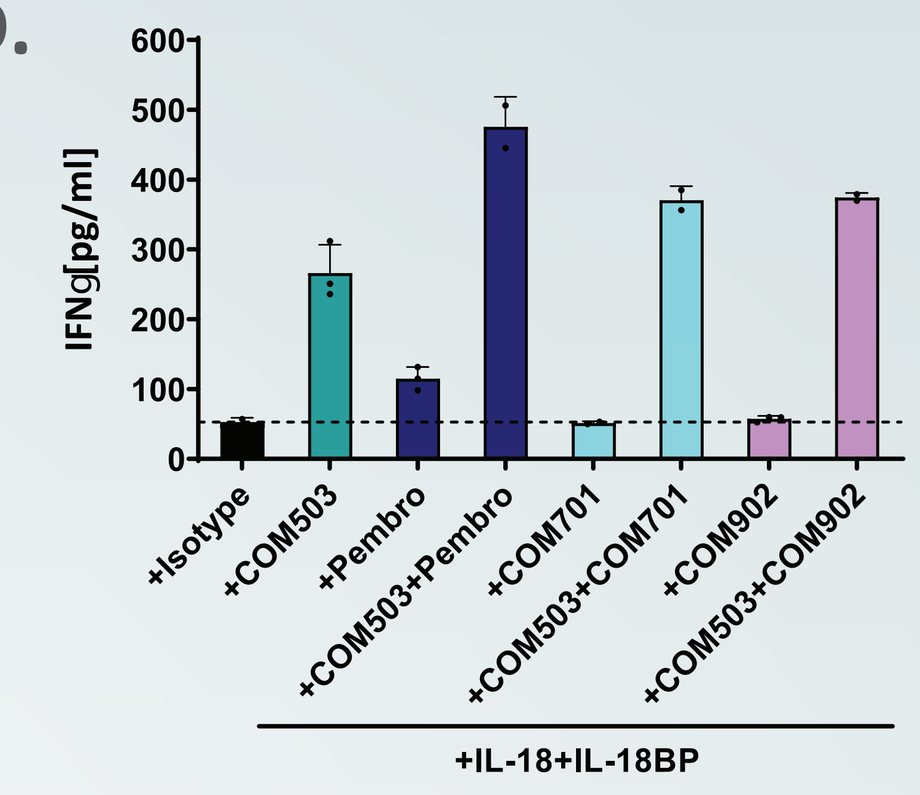
IFNg[pg/ml]
+Isotype
+COM503
+Pembro
+COM503+Pembro
+COM701
+COM503+COM701
+COM902
+COM503+COM902
+IL-18+IL-18BP

A. COM503 increased IFNg release by stimulated human CD8+ TILs in TILs- tumor cells co-culture assay in the presence of endogenous IL-18BP and rIL-18. **B.** COM503 displaces pre-complexed IL-18-IL-18BP to increase IFNg release by NK cells in the presence of IL-12. **C.** COM503 increased IFNg release by PBMCs in the presence of endogenous IL-18BP rIL-18 and IL-12. COM503 effect was similar to engineered IL-18 that does not bind IL-18BP. **D.** COM503 displaces pre-complexed IL-18-IL-18BP to increase IFNg release by by CD8+ CMV-specific T cells in CD8+ CMV-tumor cells co-culture assay as mono and in combination with COM701 (aPVRIG) /COM902 (aTIGIT) /Pembrolizumab (aPD1) Ab.

Anti-mouse IL-18BP antibody demonstrates monotherapy activity across murine syngeneic tumor models

A. E0771

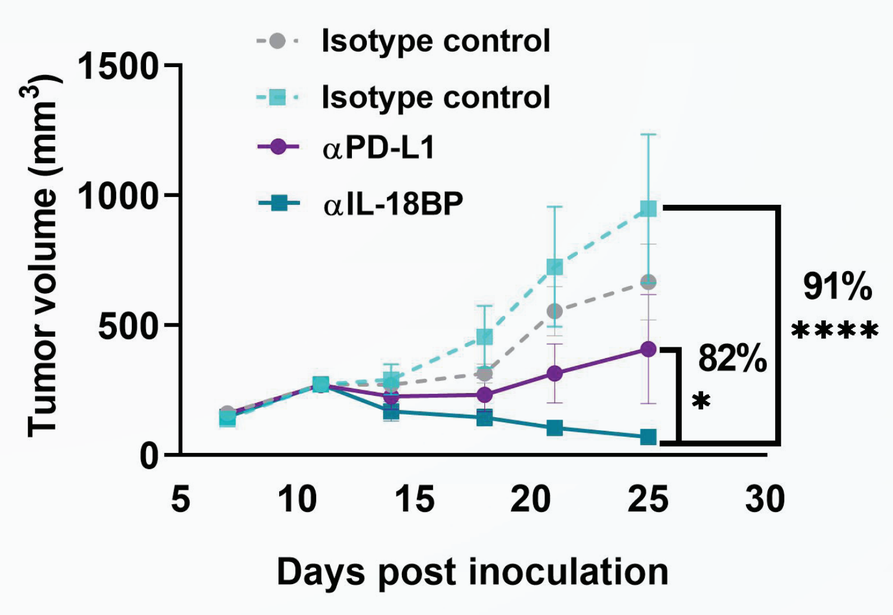
Isotype control
Isotype control
αPD-L1
αIL-18BP
91%

82%
*
Tumor volume (mm³)
Days post inoculation

B. MC38ova

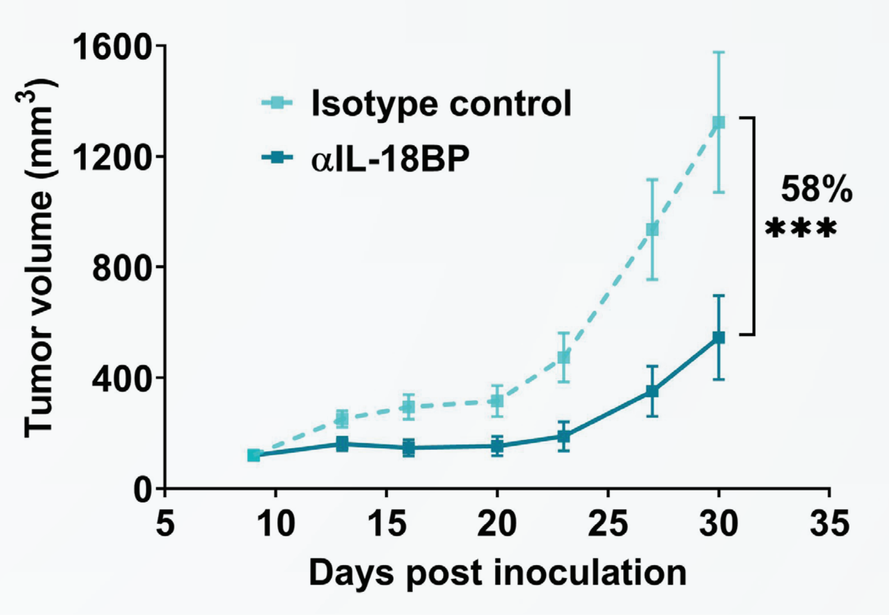
Isotype control
αIL-18BP
58%

Tumor volume (mm³)
Days post inoculation

C. B16F10-hmgp100

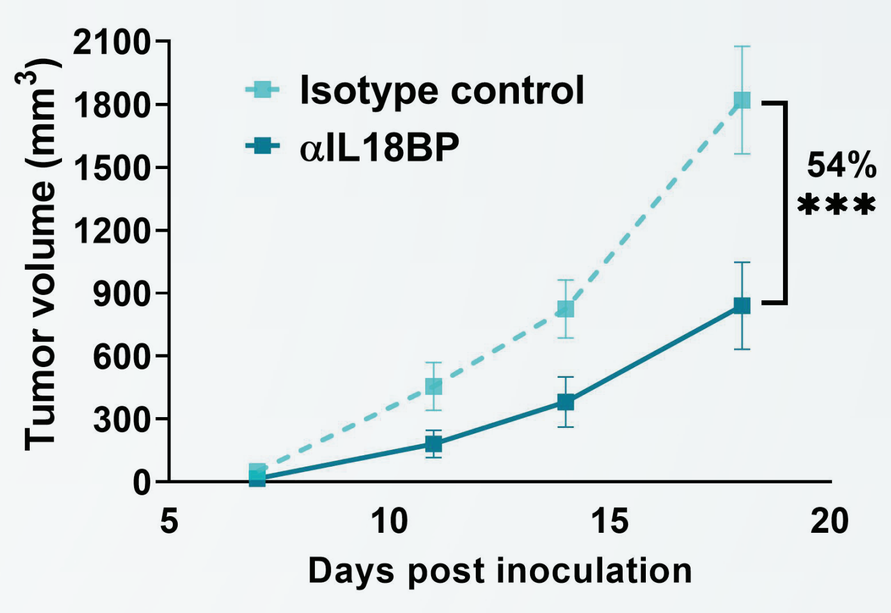
Isotype control
αIL18BP
54%

Tumor volume (mm³)
Days post inoculation

A-C. Anti-mouse IL-18BP Ab inhibits tumor growth in E0771 orthotopic mouse breast tumor model (A) MC38ova mouse CRC tumor model (B) and B16F10-hmgp100 mouse melanoma tumor model (C). Tumor volumes are represented as the Mean volume + SEM. Treatment initiated at tumor volume of $120mm^3$ and $250mm^3$ for MC38ova and E0771 respectively, on day 4 for B16gp100, and was injected twice a week for 3 weeks.

Anti-mouse IL-18BP Ab demonstrates combo activity with anti-PD-L1, modulate the TME & induces immune memory in E0771

A.

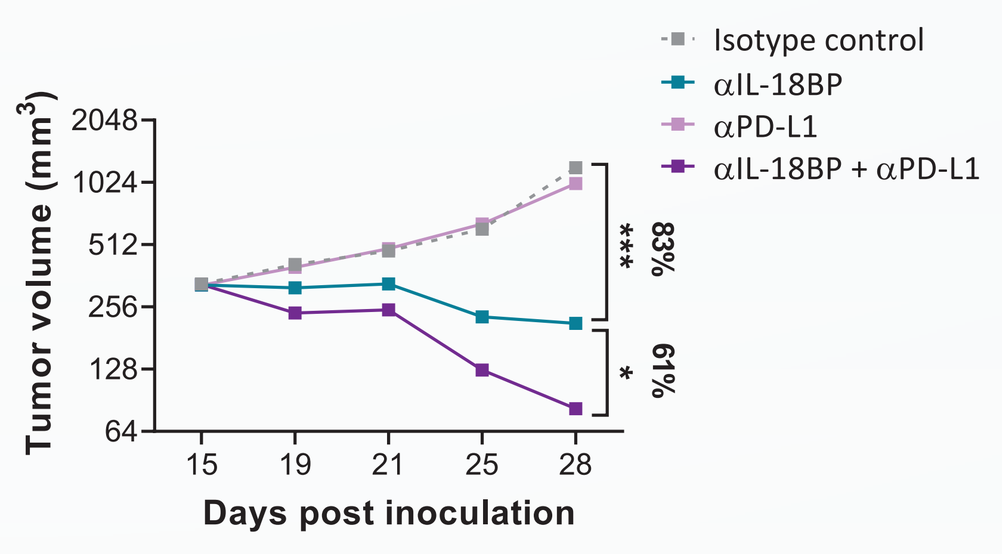
Isotype control
αIL-18BP
αPD-L1
αIL-18BP + αPD-L1
Tumor volume (mm³)
Days post inoculation
83%

61%
*

B.

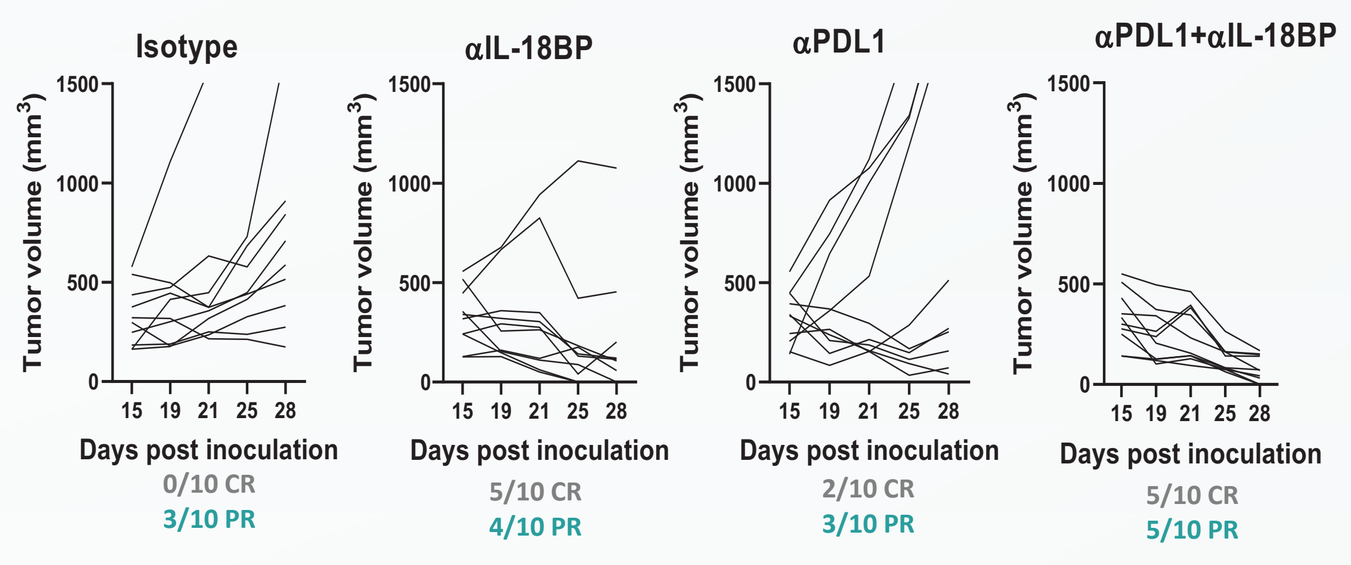
Isotype
αIL-18BP
αPDL1
αPDL1+αIL-18BP
Tumor volume (mm³)
Days post inoculation
0/10 CR
3/10 PR
5/10 CR
4/10 PR
2/10 CR
3/10 PR
5/10 CR
5/10 PR

C.

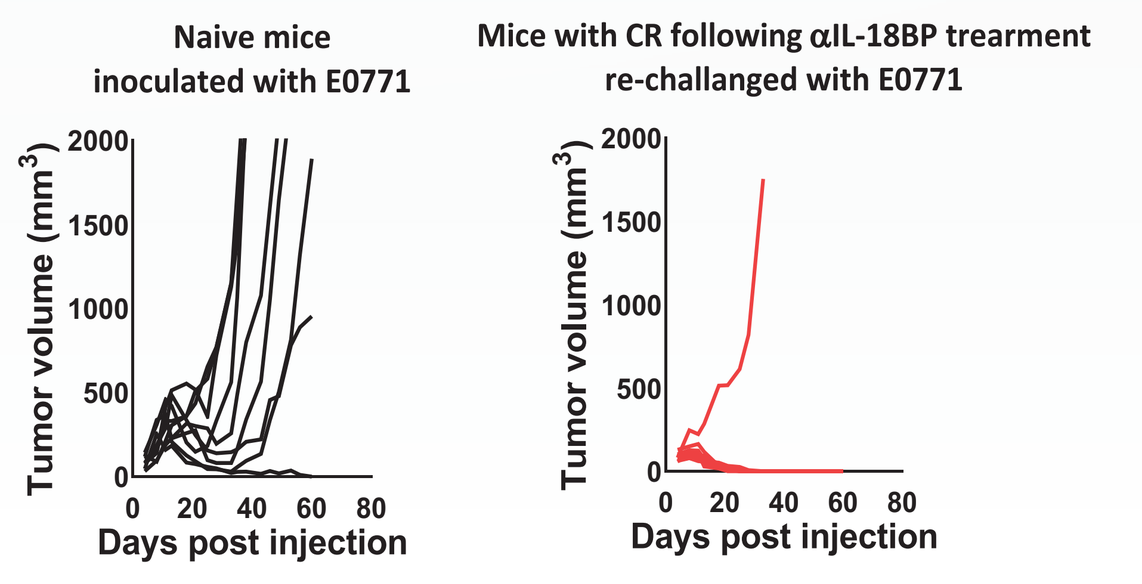
Naive mice inoculated with E0771
Mice with CR following αIL-18BP treatment re-challanged with E0771
Tumor volume (mm³)
Days post injection

D.

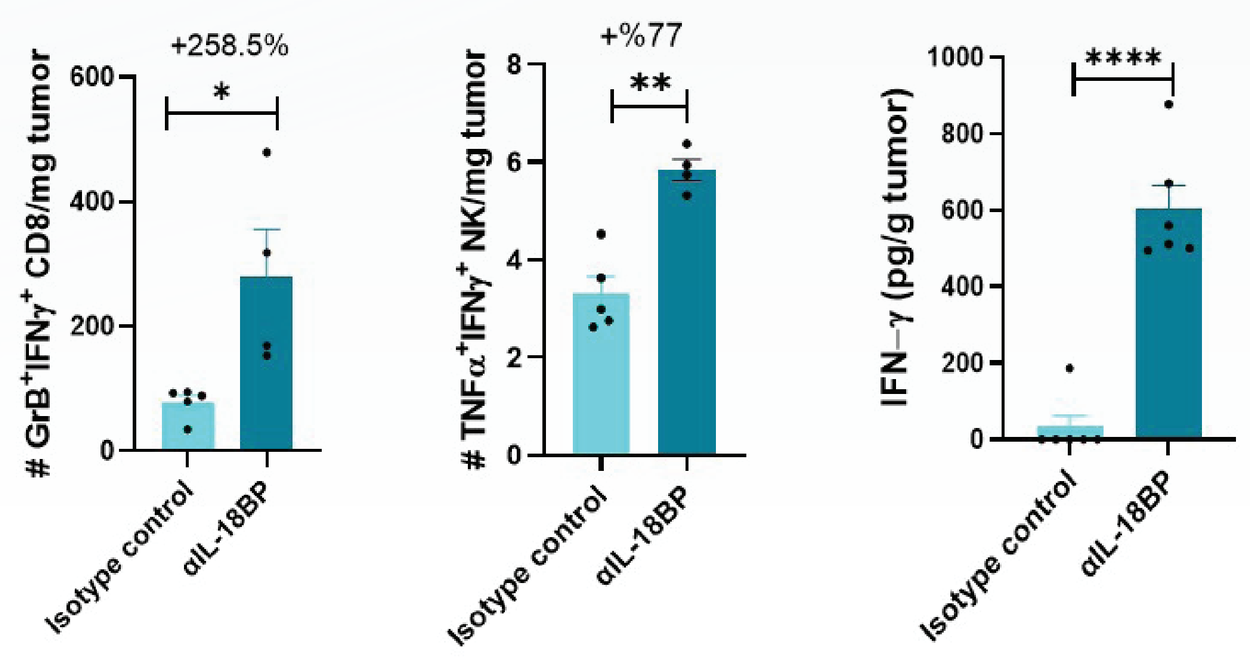
+258.5%
*
GrB⁺IFNγ⁺ CD8/mg tumor
+%77
**
TNFα⁺IFNγ⁺ NK/mg tumor

IFN−γ (pg/g tumor)
Isotype control
αIL-18BP

A. Anti-mouse IL-18BP Ab in combination with anti PD-L1 Ab inhibits tumor growth in E0771 mouse tumor model. **B.** Individual tumors measurements for each mouse are depicted **C.** Groups of 5-10 C57BL/6 tumor- naïve age-matched mice were orthotopically inoculated with E0771, followed by a treatment either with anti-IL-18BP Ab or isotype control. After two months, tumor-free and naïve aged-matched mice were orthotopically re-inoculated with E0771. Individual tumors measurements for each mouse are depicted. **D.** Anti-mouse IL-18BP Ab triggers the release of IFNg and increase activation of T and NK cells in the TME. CR- complete response. PR- partial response

Anti-mouse IL-18BP Ab modulates tumor microenvironment without affecting the periphery

A.

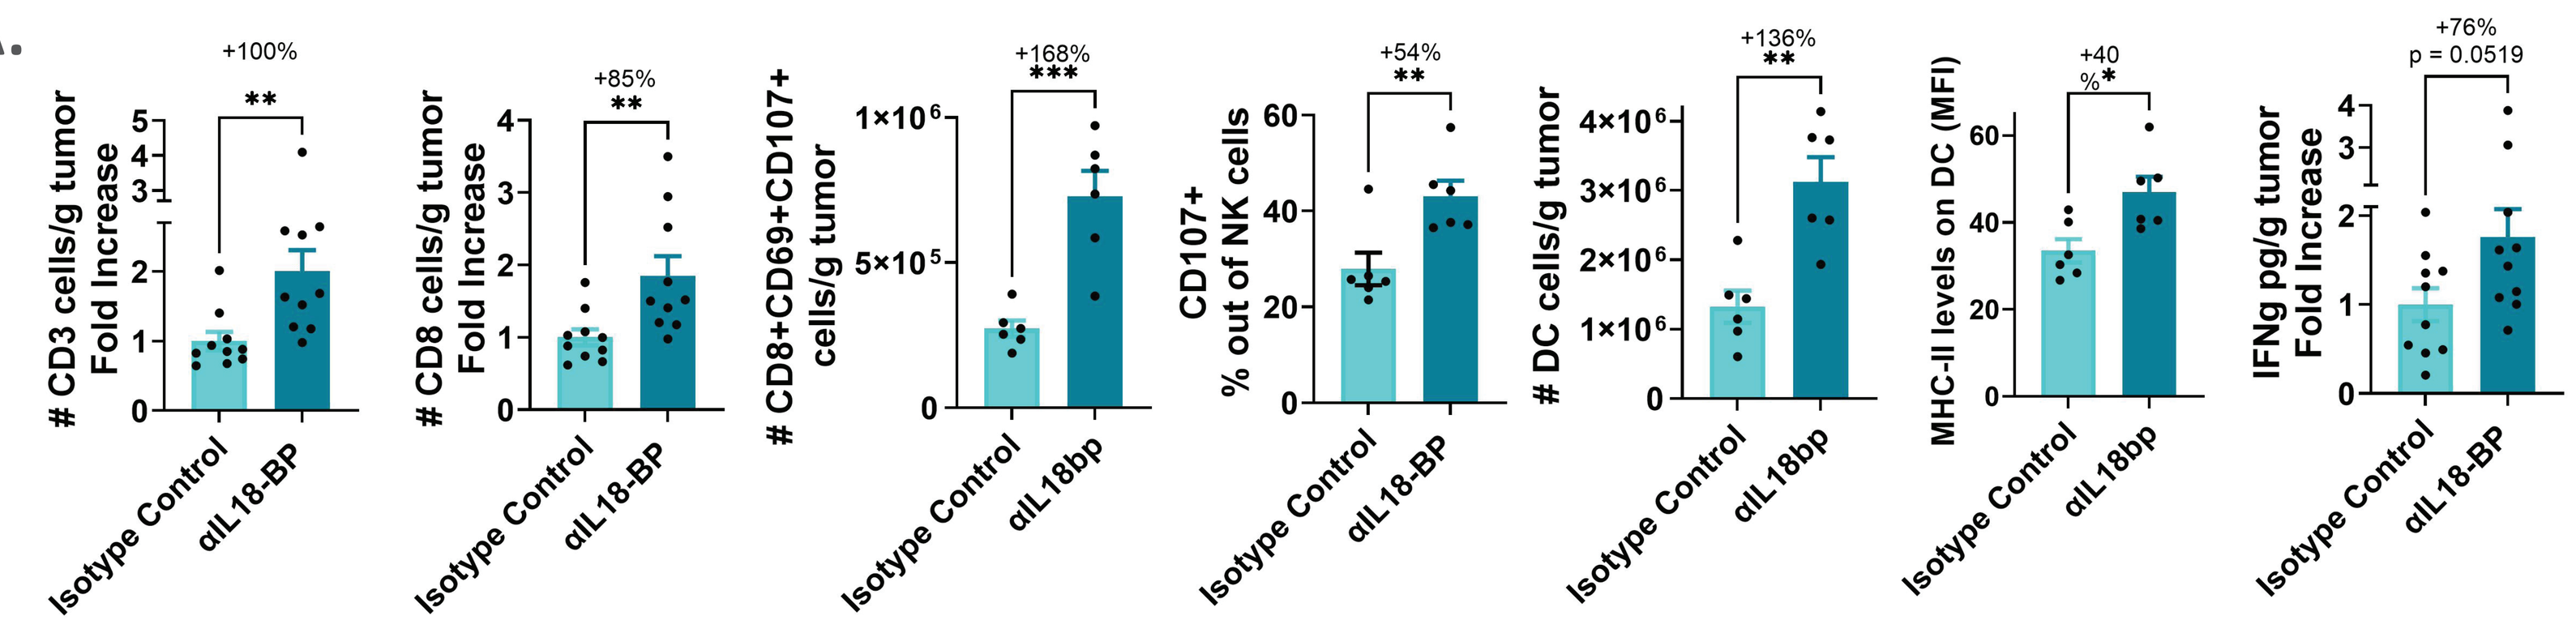
+100%
**
CD3 cells/g tumor Fold Increase
+85%
**
CD8 cells/g tumor Fold Increase
+168%

CD8+CD69+CD107+ cells/g tumor
+54%
**
CD107+ % out of NK cells
+136%
**
DC cells/g tumor
+40 %*
MHC-II levels on DC (MFI)
+76%
p = 0.0519
IFNg pg/g tumor Fold Increase
Isotype Control
αIL18-BP
αIL18bp

B.

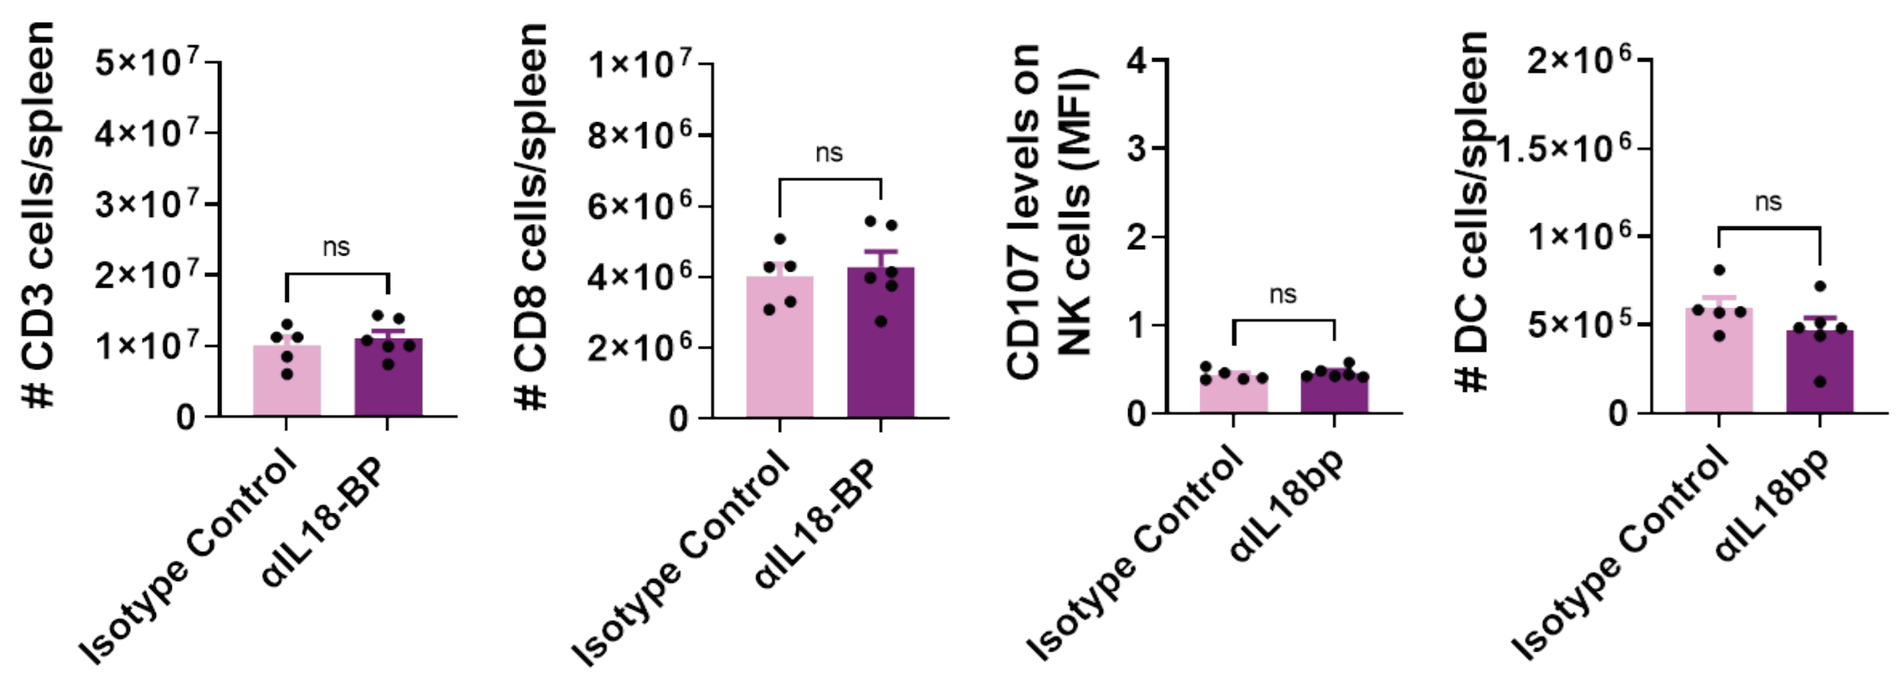
CD3 cells/spleen
CD8 cells/spleen
CD107 levels on NK cells (MFI)
DC cells/spleen
ns
Isotype Control
αIL18-BP
αIL18bp

C.

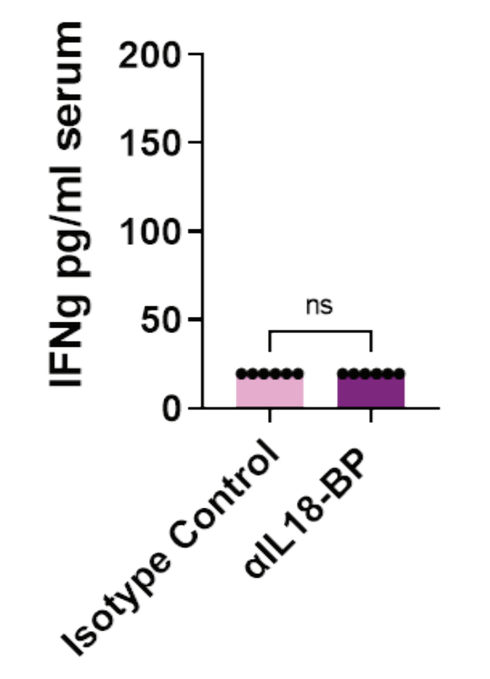
IFNg pg/ml serum
ns
Isotype Control
αIL18-BP

A-C. C57BL/6 mice were subcutaneously injected with MC38ovadim and were treated with anti-mouse IL-18BP Ab. Tumors, spleens and serum were harvested, and immune composition and cytokine concentrations were determined in the tumor microenvironment (A), in the spleen (B) and in the serum (C).

Anti-IL-18BP Ab is expected to have a better therapeutic window compared to recombinant cytokines

A.

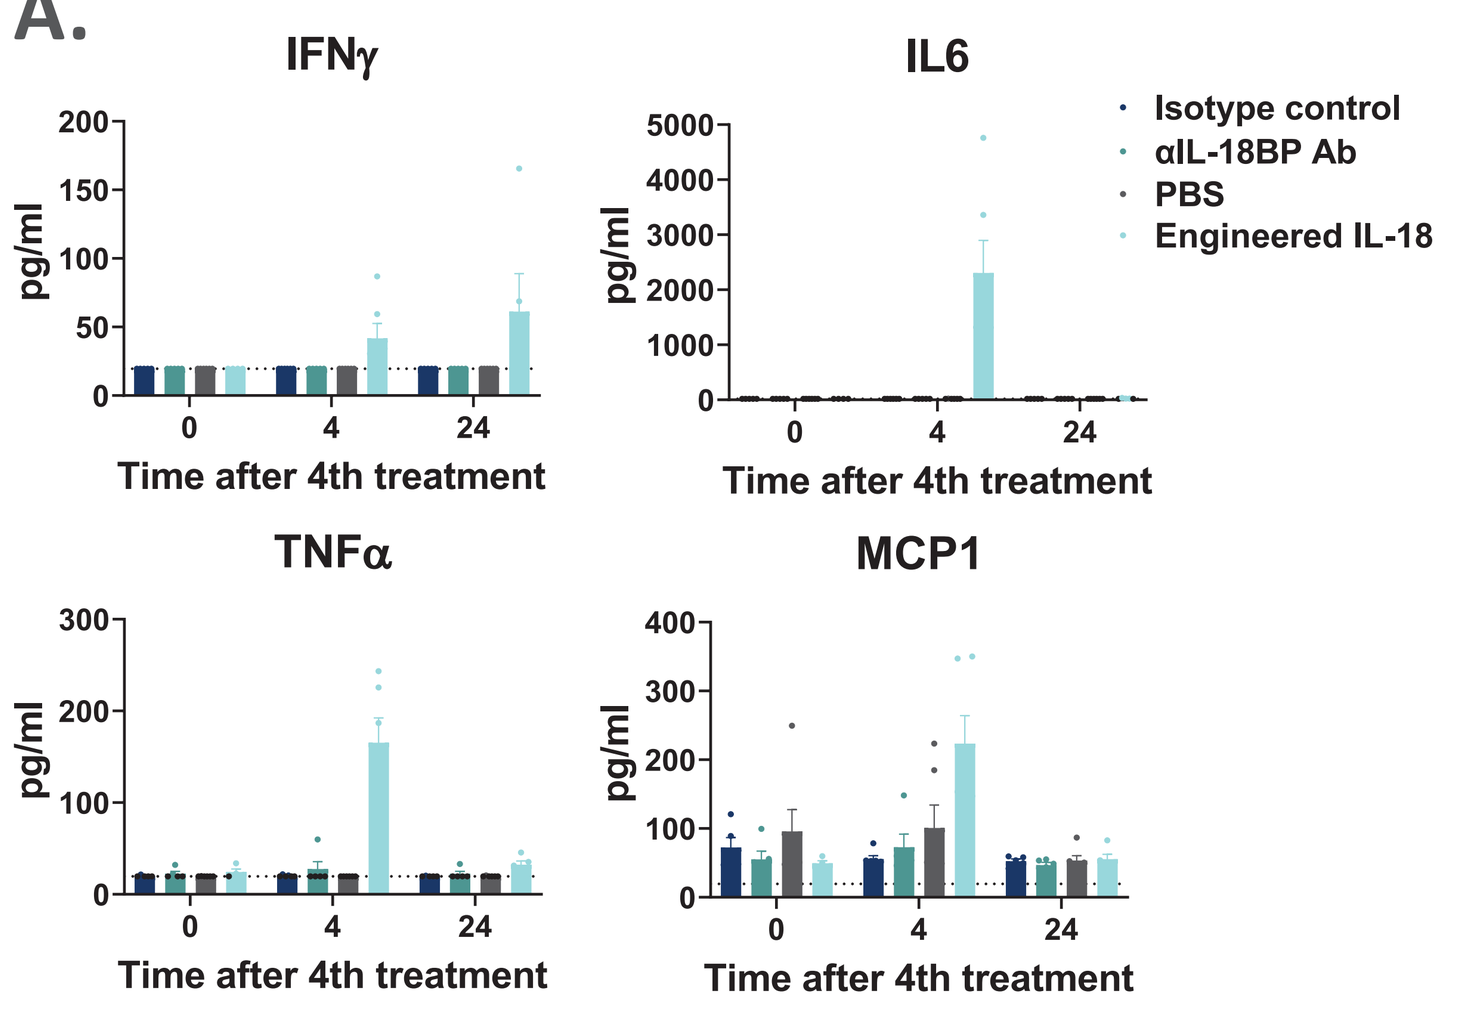
IFNγ
IL6
TNFα
MCP1
pg/ml
Time after 4th treatment
Isotype control
αIL-18BP Ab
PBS
Engineered IL-18

B.

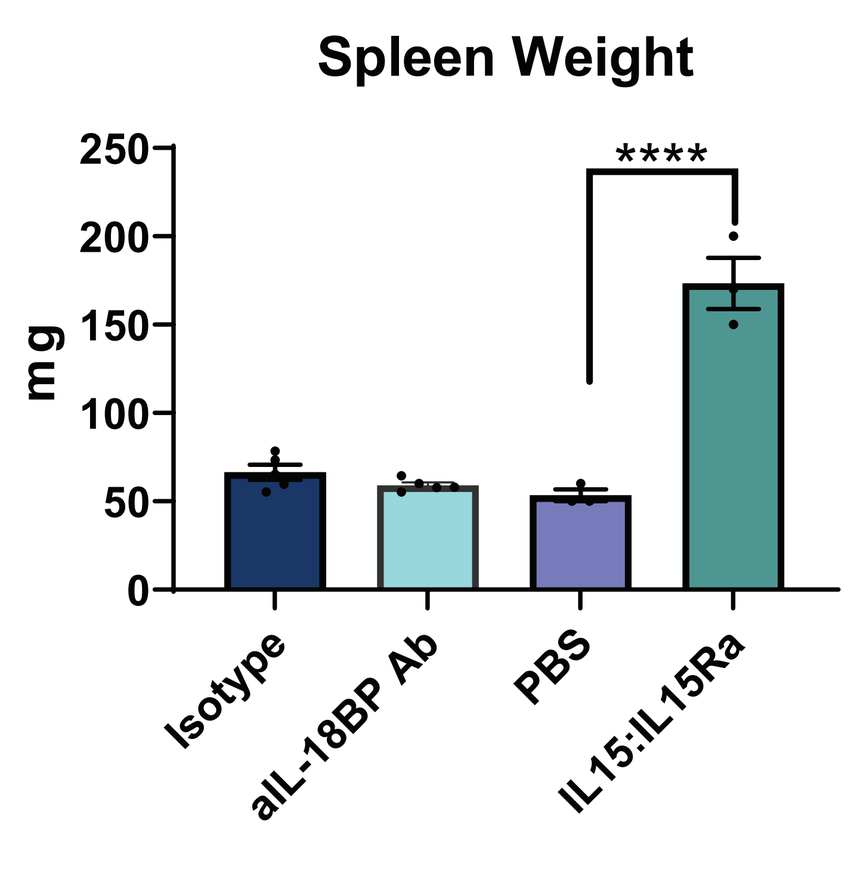
Spleen Weight
mg

Isotype
aIL-18BP Ab
PBS
IL15:IL15Ra

C57BL/6 mice were subcutaneously injected with MC38ova cells and treated with anti-mouse IL-18BP Ab, isotype control, PBS, or engineered IL-18 (engineered IL-18 does not bind IL-18BP) twice weekly. **A.** Serum was analyzed 4 and 24hr after the 4th treatment for presence of indicated molecules - IFNg, TNFa, IL6 and MCP1. **B.** Administration anti-mIL-18BP Ab to mice did not result in splenomegaly in contrast to rIL-15:IL-15Ra complex.

CONCLUSION

• IL-18 is upregulated in the TME and is mostly bound by IL-18BP • COM503, a potential first-in-class, high-affinity anti-IL-18BP Ab, induces human T and NK cell responses in-vitro • In mouse, anti-IL-18BP Ab induces potent anti-tumor responses and pronounced TME-constrained immune activation, this in contrast to systemically administered therapeutic cytokines, which generate a non-localized inflammatory response • Taken together, blocking IL-18BP is a promising novel approach to harness cytokine biology for the treatment of cancer